Analysis of Spatio-Temporal Features of Histopathology in Murine AD Models and Human AD Samples

Christina Höller[1, (2)], Vera Niederkofler[1], Jörg Neddens[1], Ewald Auer[1], Heinrich Römer[2], Johannes Attems[3], Birgit Hutter-Paier[1]
[1]QPS-Austria, Parkring 12, 8074 Grambach, Austria [2]Karl-Franzens University, Institute of Zoology, 8010 Graz, Austria [3]Institute for Ageing and Health, Newcastle University, UK

QPS® helps you navigate

BACKGROUND & AIMS

Alzheimer‘s disease (AD) is a degenerative brain disease characterized by two pathological hallmarks: amyloid plaques and neurofibrillary tangels (NFTs). The latter consits of hyperphosphorylated protein tau. This protein is, under physiological conditions, responsible for the axonal microtubule stabilization. Hyperphosphorylated tau protein may aggregate and further lead to NFTs and the loss of the stabilizing function. The hyperphosphorylation takes place at certain tau protein residues like serine, threonin and tyrosine.
This study was done to investigate tau pathologies in transgenic APP_{SL} mice overexpressing the human APP and in human brains. The aim of this study is to test if there is an increase of tau with age or Braak stage, respectively. Furthermore, it will be examined if APP_{SL} mice show any tau pathologies at all in the absence of human tau mutations. If murine tau protein aggregates in transgenic mice and can be evaluated the same way as in human samples, APP_{SL} mice could be a potential model of human AD histopathology.

MATERIALS AND METHODS

Indirect immunofluorescence was used to visualize hyperphosphorylated tau for three specific phospho-sites (pSer199, pSer396, pSer422) on human and mouse tissue, using:

- human demented brain samples (Braak I/II, III/IV, V/VI) and healthy controls, 5 brain areas

- Transgenic APP_{SL} mouse brains (6, 9 and 12 months) and age-matched non-transgenic controls

- Polyclonal Rabbit Anti-Tau pSer199
- Monoclonal Mouse Anti-Tau pSer396 (clone PHF13)
- Monoclonal Mouse Anti-Tau pSer422 (clone 5.6.11)

RESULTS

pSer199, pSer396 and pSer422 increase with disease progression in human brains

pTau labeling in front. cortex across Braak stages | pTau labeling in HC across Braak stages

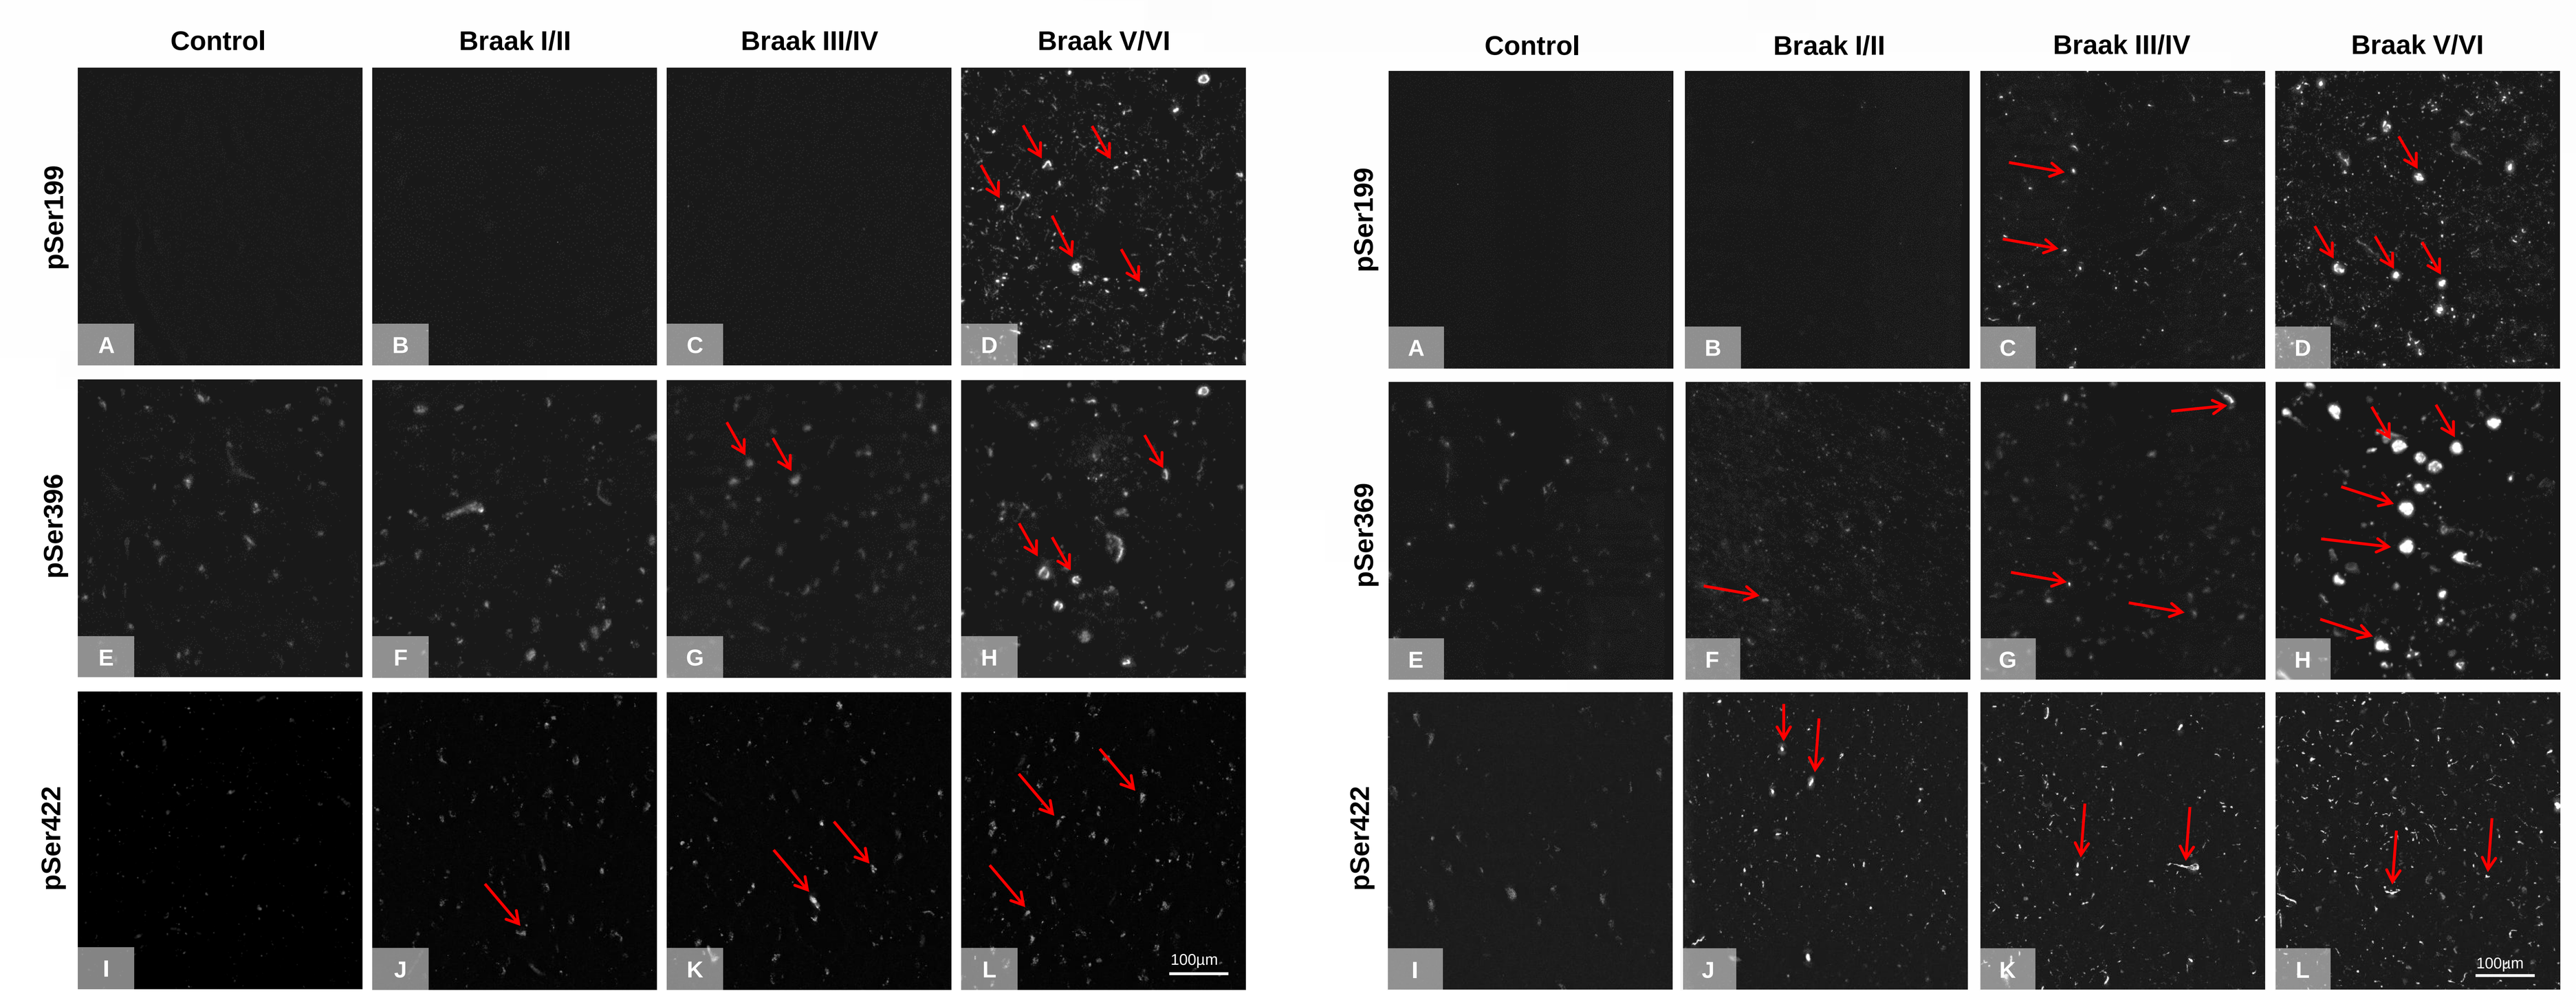
Control
Braak I/II
Braak III/IV
Braak V/VI
pSer199
pSer396
pSer422
pSer369
100µm

Figure 1. pSer199, pSer396 and pSer422 labeling of human frontal cortex *(left)* and hippocampus *(right)* at different Braak stages (I/II, III/IV and V/VI) and the control. An increase of pSer199, pSer396 and pSer422 tau was observed with Braak stages for frontal cortex and hippocampus. Evaluations showed that the strongest tau pathologies appeared in the hippocampus. Furhtermore,pSer199 showed the highest tau pathology at Braak V/VI and seems therefore to be a late indicator for AD in human.

Example of pSer422 labeling in cortex and HC

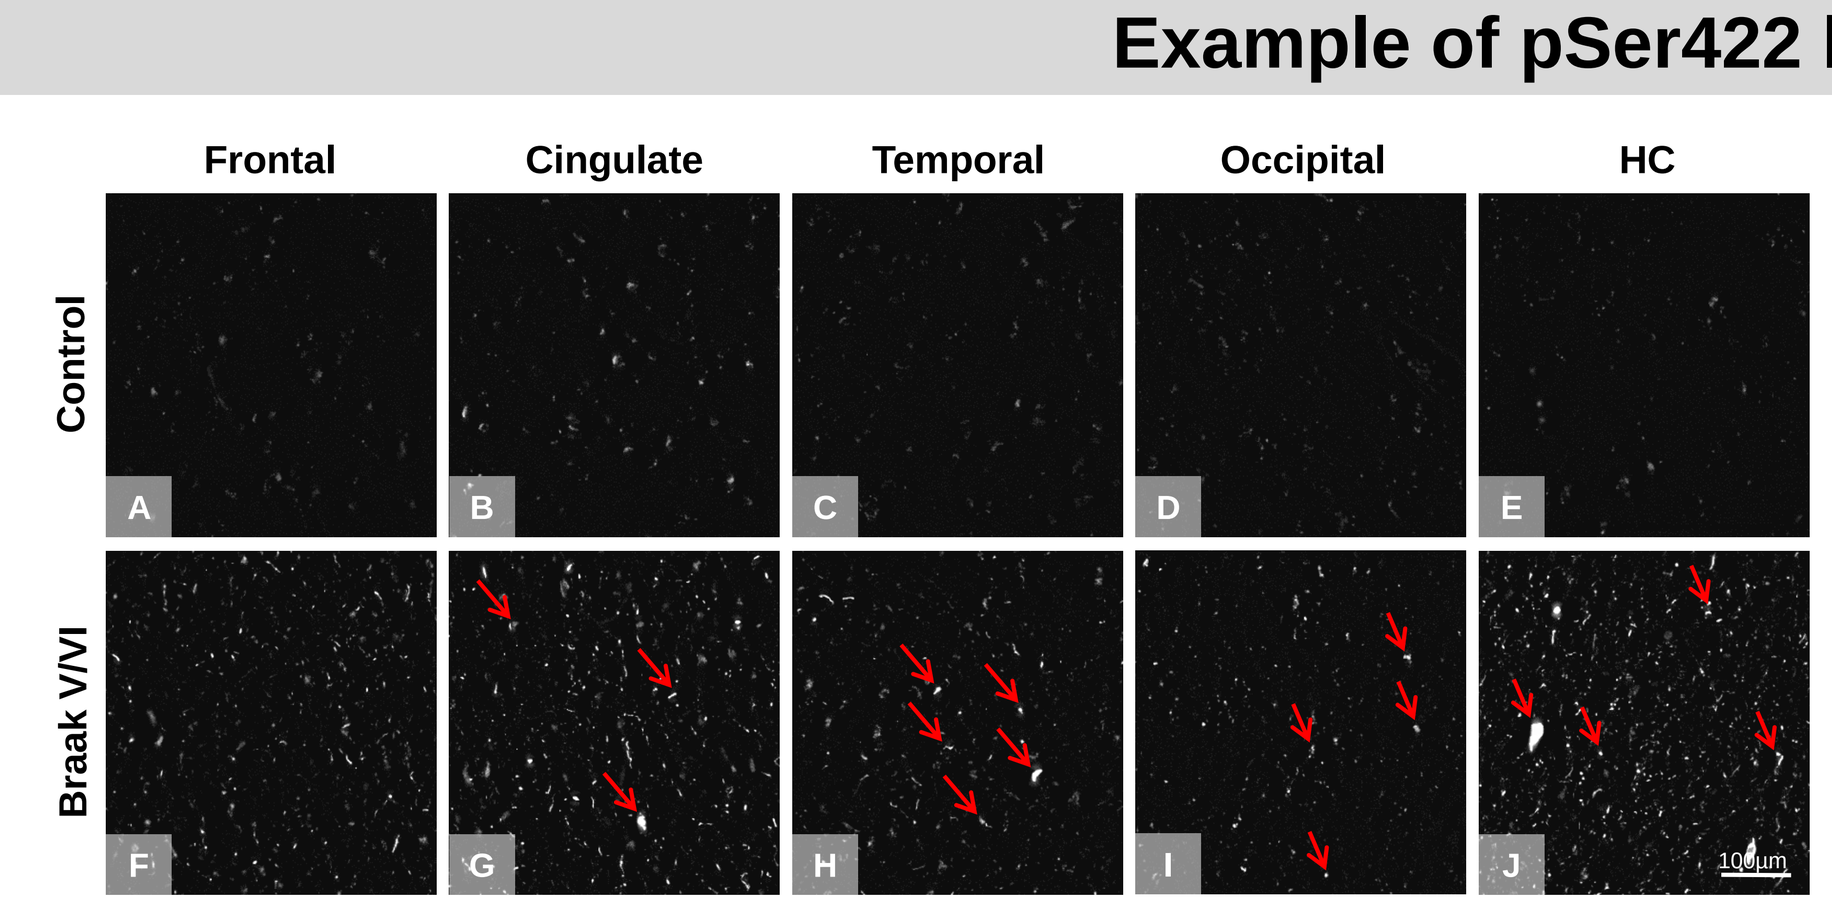
Frontal
Cingulate
Temporal
Occipital
HC
Control
Braak V/VI

Figure 2: pSer422 labeling of five different human brain regions at Braak V/VI and the control. The cingulate, frontal, occipital and temporal cortex, as well as the hippocampus showed an increase of pSer422 tau signal of Braak V/VI compared to the control. pSer422 tau labeled hippocampal tissue (J) showed the highest amount and density of hyperphosphorylated tau compared to cortical regions.

Quantification of pTau in human cortical areas and hippocampus

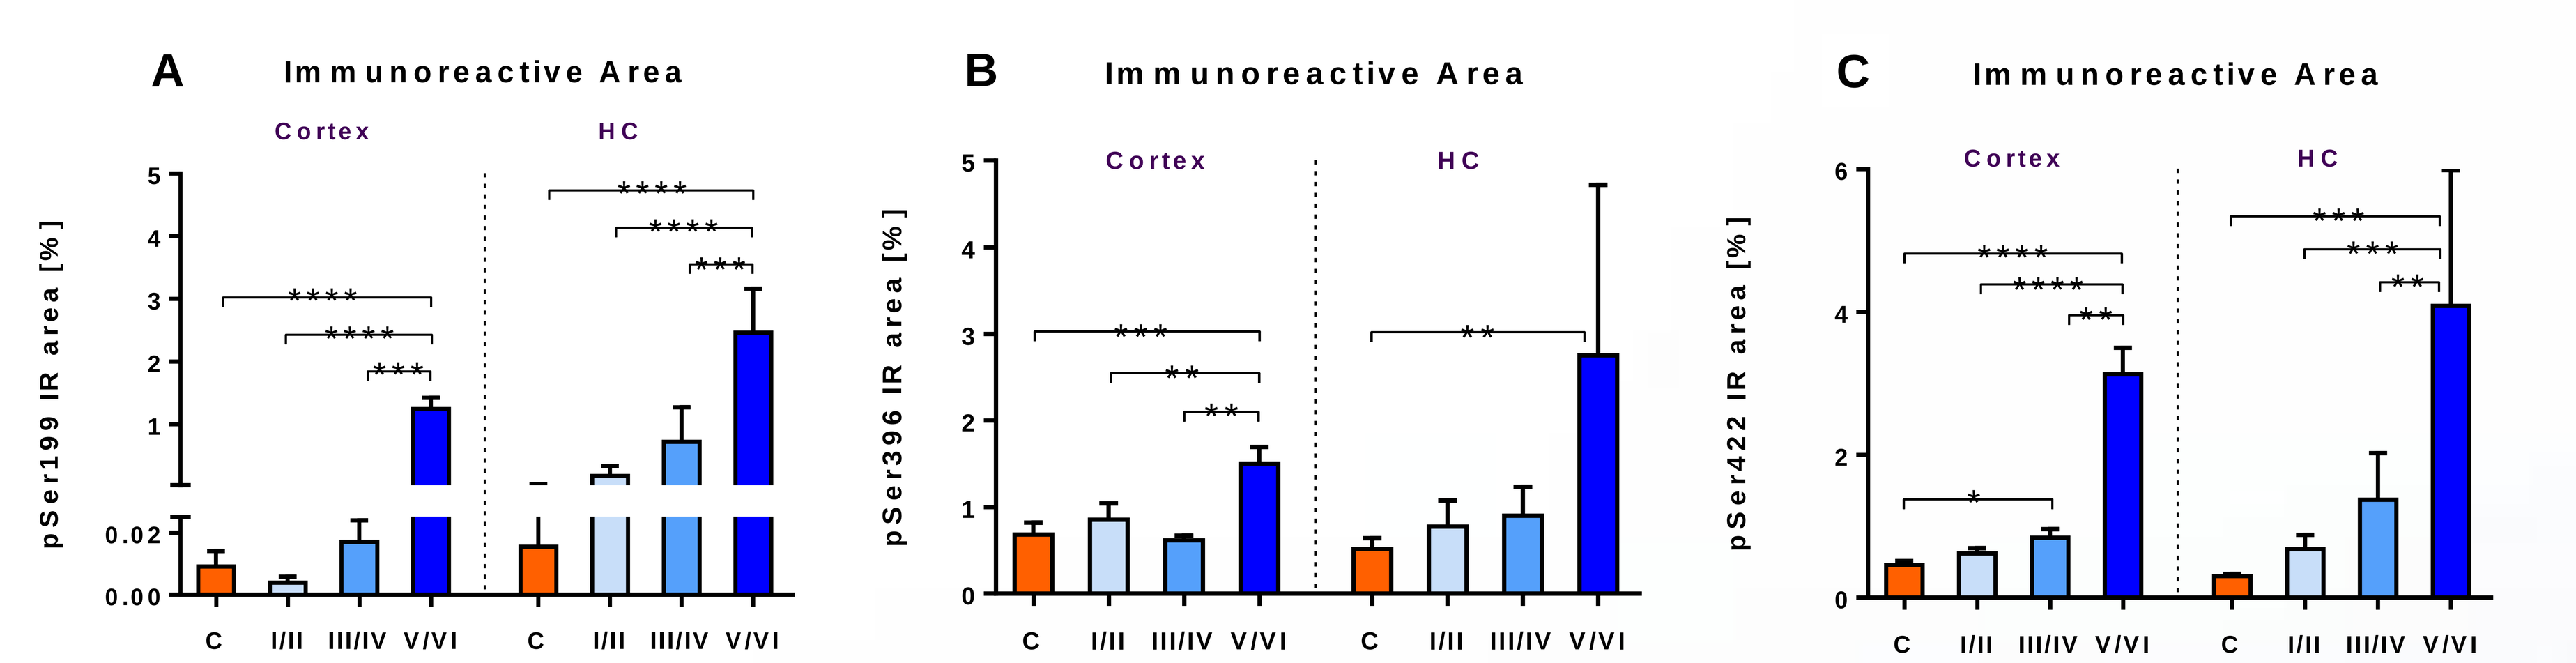
A Immunoreactive Area
B Immunoreactive Area
C Immunoreactive Area
Cortex
HC
pSer199 IR area [%]
pSer396 IR area [%]
pSer422 IR area [%]
C I/II III/IV V/VI

Figure 3: IR area of pSer199 *(A)*, pSer396 *(B)* and pSer422 *(C)* tau in the total cortex and hippocampus of Braak stages (I/II, III/IV, V/VI; blue) and the control (C; orange). Tau pathologies labeled with pSer199, pSer396 and pSer422 showed an increase with Braak stages for cortical areas as well as for the hippocampus.

Hyperphosphorylated tau residues in APP_{SL} brains

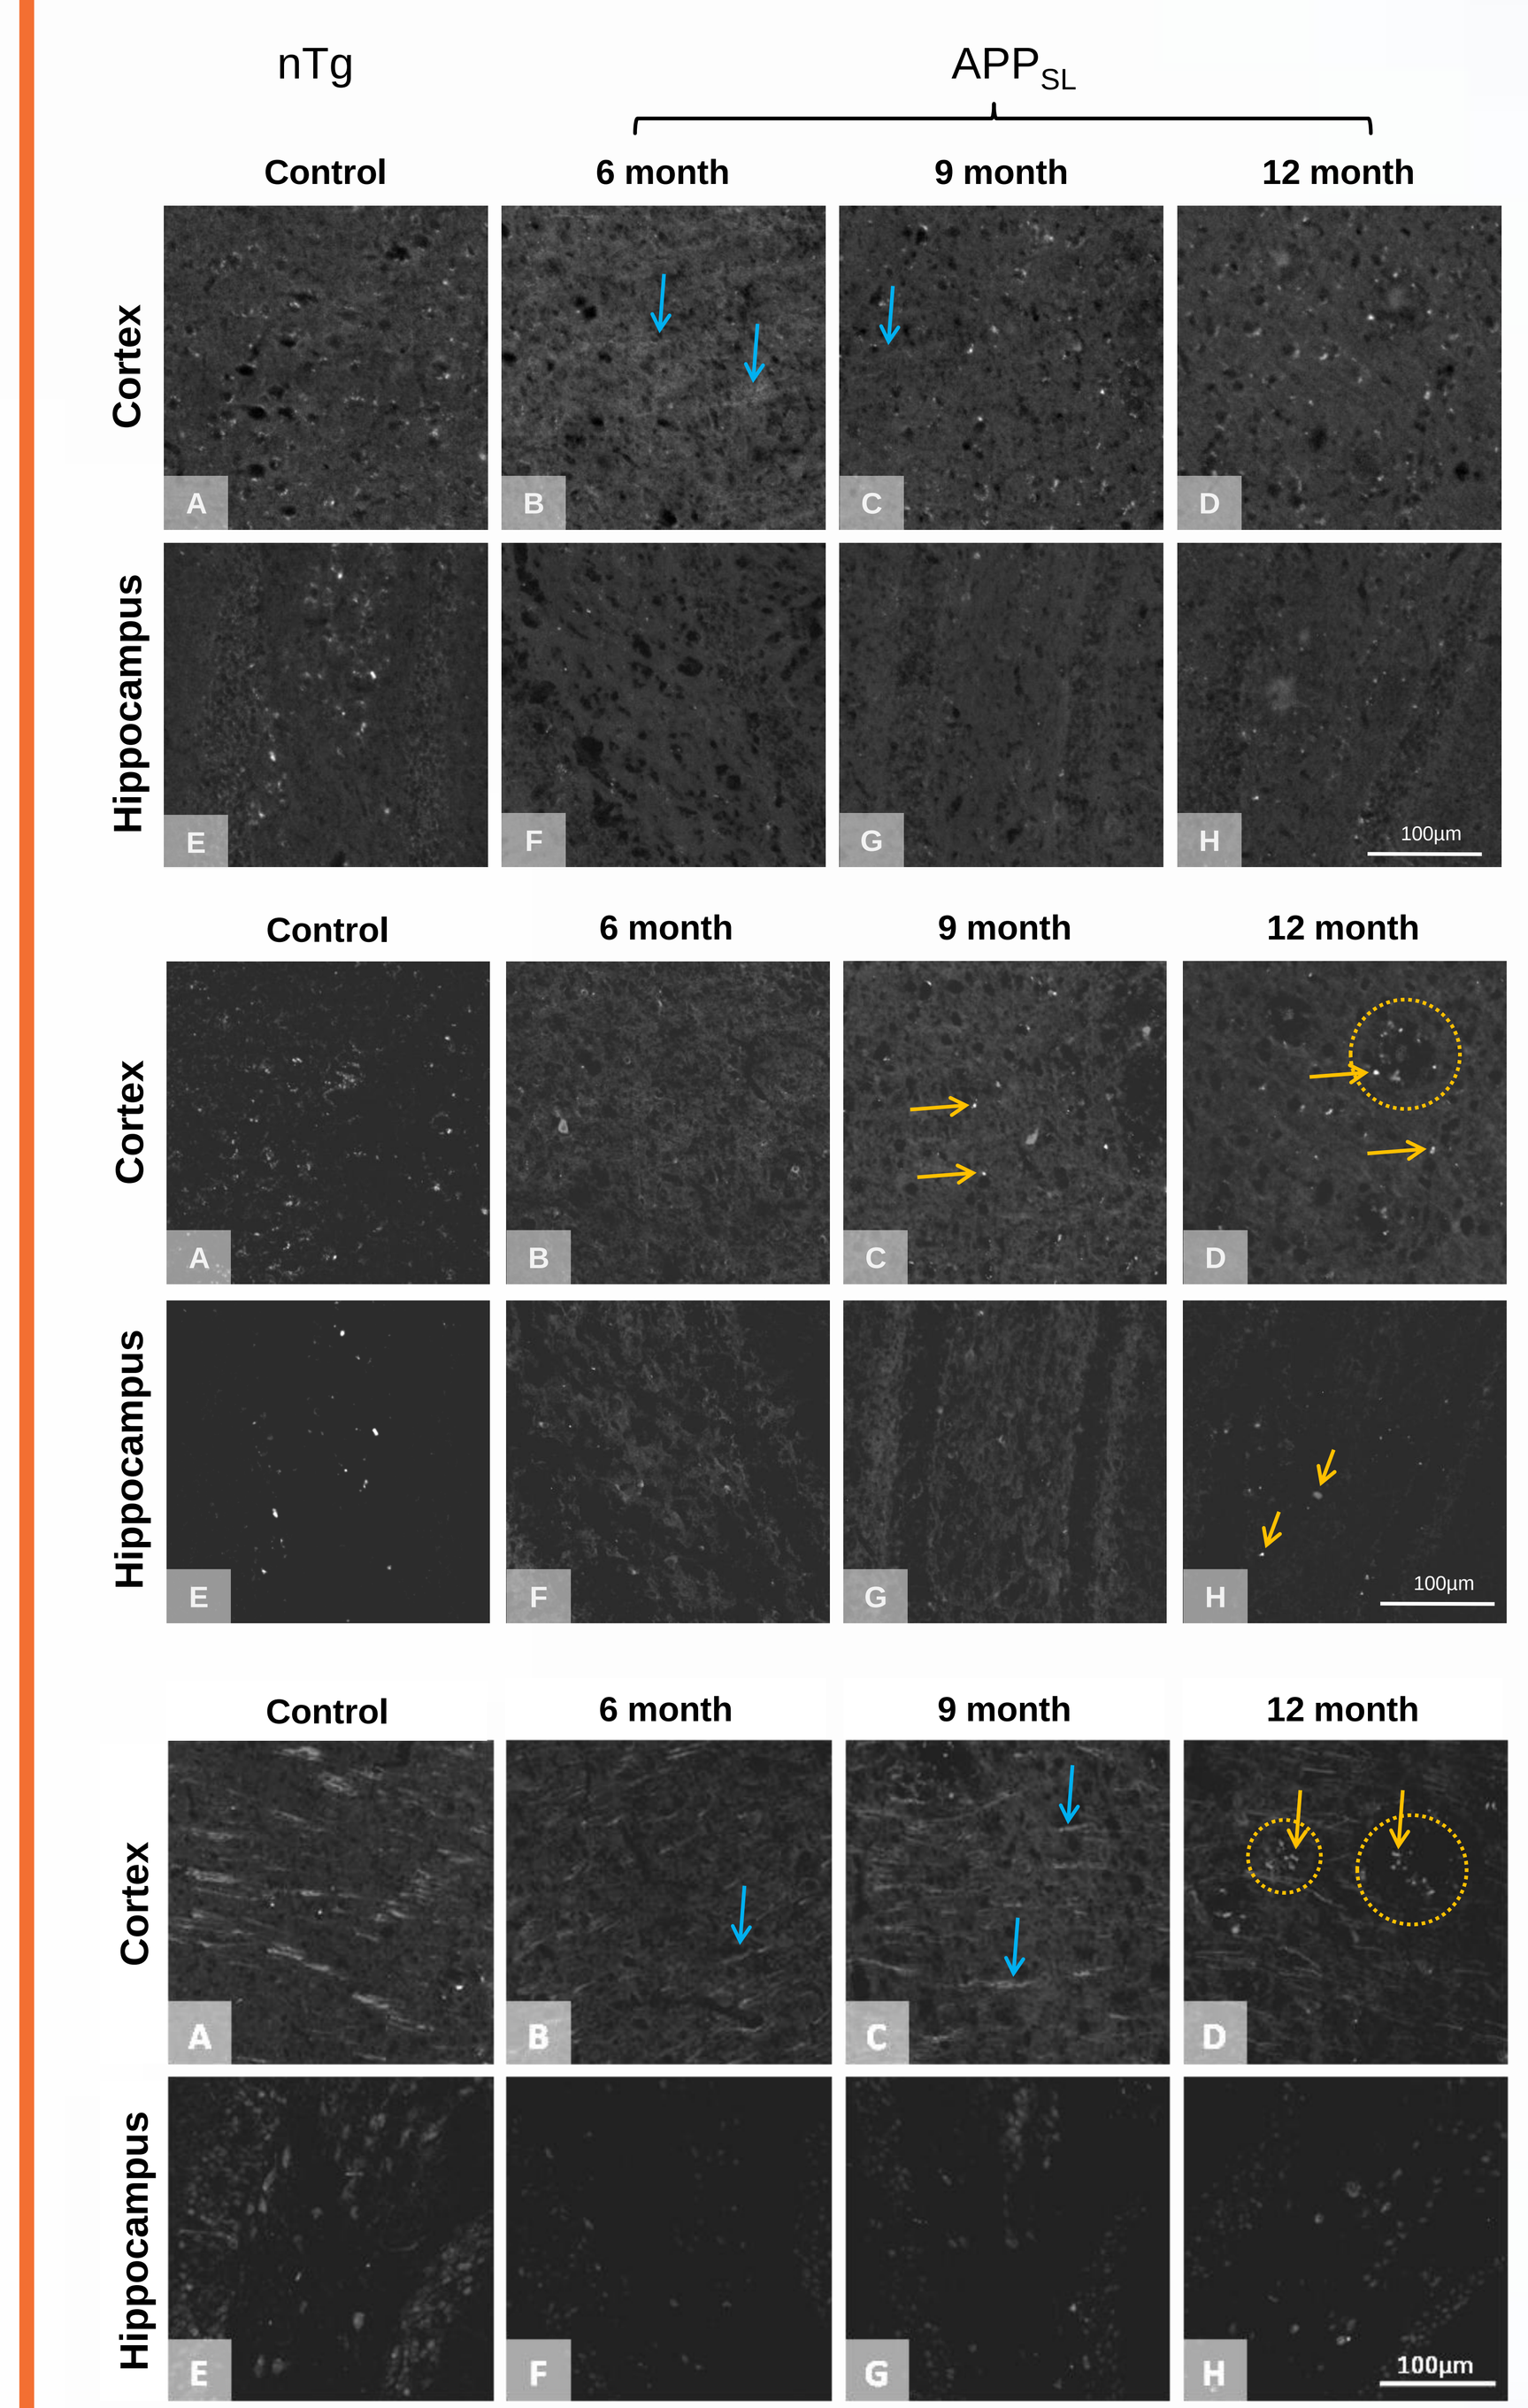
nTg
APPSL
Control
6 month
9 month
12 month
Cortex
Hippocampus
100µm

Figure 4: pSer199, pSer396 and pSer422 tau immunolabeled murine cortex and hippocampus. Labeling of pSer199 tau showed a strongly labeled dendritic tau in APP_{SL} mice at the age of 6 months which decreases with age in cortex and HC. pSer396 and pSer422 tau labeled cortical tissue showed an increase of tau pathology with age for cortex and HC, especially high tau signals were found around amyloid plaques (orange encircled). Furtheremore, high amounts of axonal pSer422 tau were detected in the cortex.

Immunoreactive area (IR) in murine hippocampus (HC) and cortex (Ctx)

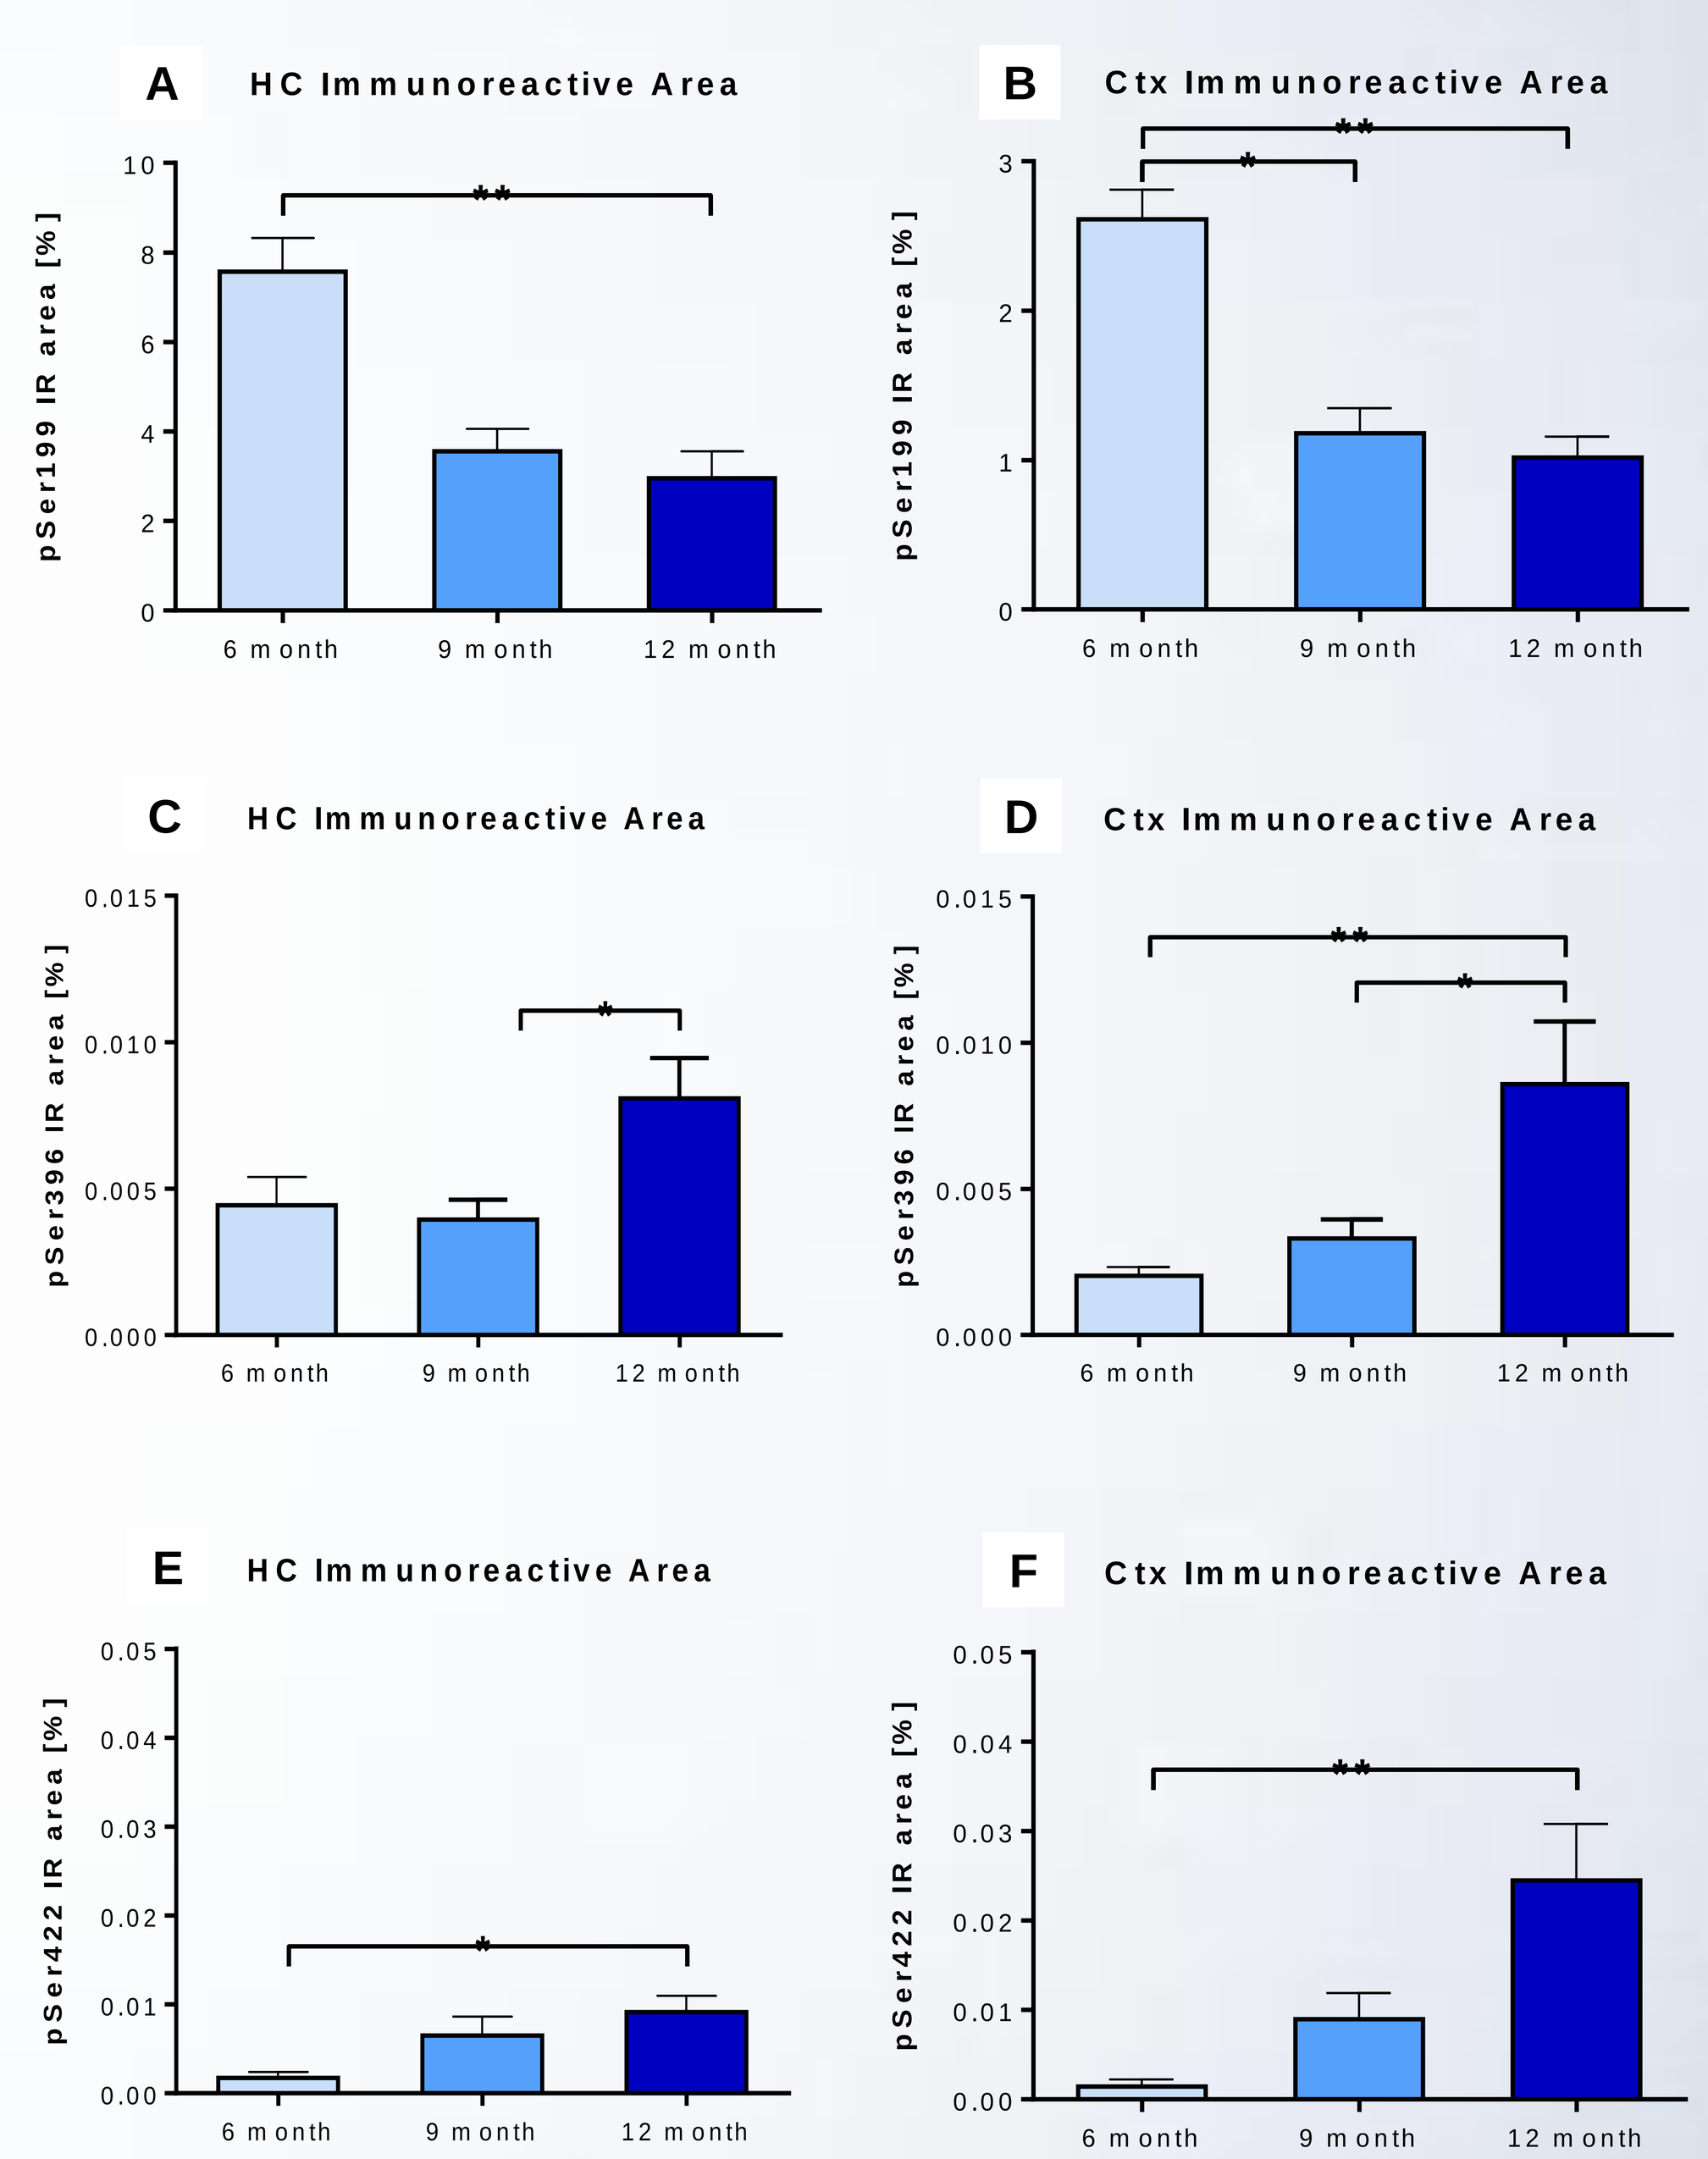
A HC Immunoreactive Area
B Ctx Immunoreactive Area
C HC Immunoreactive Area
D Ctx Immunoreactive Area
E HC Immunoreactive Area
F Ctx Immunoreactive Area
pSer199 IR area [%]
pSer396 IR area [%]
pSer422 IR area [%]
6 month
9 month
12 month

Figure 5: IR areas of pSer199, pSer396 and pSer422 in hippocampus (left) and cortex (right) of APP_{SL} mice. IR of pSer199 tau in murine hippocampus and cortex showed a decrease with age. Quantifications of pSer396 and pSer422 tau showed an significant increase of hyperphosphorylated tau with age for hippocampus and cortex.

CONCLUSION

Data showed a comparable increase of pSer396 tau and pSer422 tau over age and Braak stage for APP_{SL} mice and human samples, respectively. The results of pSer199 tau showed a decrease over age, while in human tissues an increase over Braak stages occurred. The APP_{SL} mouse model is in some read-outs similar to human samples. The APP_{SL} mouse is therefore a useful model for the analyses of tau-related AD pathologies.

Contact us to support your drug development programme!

Contact: Birgit Hutter-Paier I Director Neuropharmacology I QPS Austria GmbH
Parkring 12 I 8074 Grambach I Austria
birgit.hutter-paier@qps.com I www.qpsneuro.com

Meet us at Booth #207

AAIC 2016, P4-10-225, Poster: #10791