Characterization of Vascular Alterations in Human Alzheimer's Disease Patients in Comparison to the APP_{SL} and 5xFAD Mouse Models

Vera Niederkofler*[1], Magdalena Temmel*[1], Karin Lanegger[2] , Joerg Neddens[1], Ewald Auer, [1] Johannes Attems[3], Birgit Hutter-Paier[1]
[1]QPS Neuropharmacology, Grambach, Austria; [2]FH Joanneum Graz, Austria; [3]Newcastle University, Newcastle, United Kingdom; *equal contribution

QPS® helps you navigate

ABSTRACT

Converging evidence suggests a link between vascular dysfunction and Alzheimer's disease (AD). This study aimed at investigating vascular changes in AD patients as well as in two transgenic AD mouse models showing amyloid beta (Aβ) pathology - APP_{SL} and 5xFAD. To this end, we quantified immunofluorescently labeled sections from human AD patients at different Braak stages and brain sections from the two transgenic mouse lines across different time points. Most of the parameters analyzed in the cortex and the hippocampus showed a parallel progression in both species. Our data highlight the validity of the quantitative histological approach used here. Furthermore, they demonstrate that the APP_{SL} and the 5xFAD mouse models are valuable tools to study Aβ as well as vascular-related alterations in AD.

MATERIALS AND METHODS

Tissue processing and imaging

Systematic random sets of 6-10 µm thick brain sections were immunofluorescently labeled for Aβ (6E10, Convance) and collagen IV (Abcam), imaged and quantitatively evaluated using image analysis software (Axio.Imager Z1 microscope, ImageProPlus).

Human brain tissue	Mouse brain tissue	
AD patients (Braak I/II, II/IV, V/VI)	• APP_{SL} • 5xFAD	6, 9, 12 months
Non-AD subjects	Non-transgenic (nTg)	

Measurement of cerebral amyloid angiopathy (CAA)

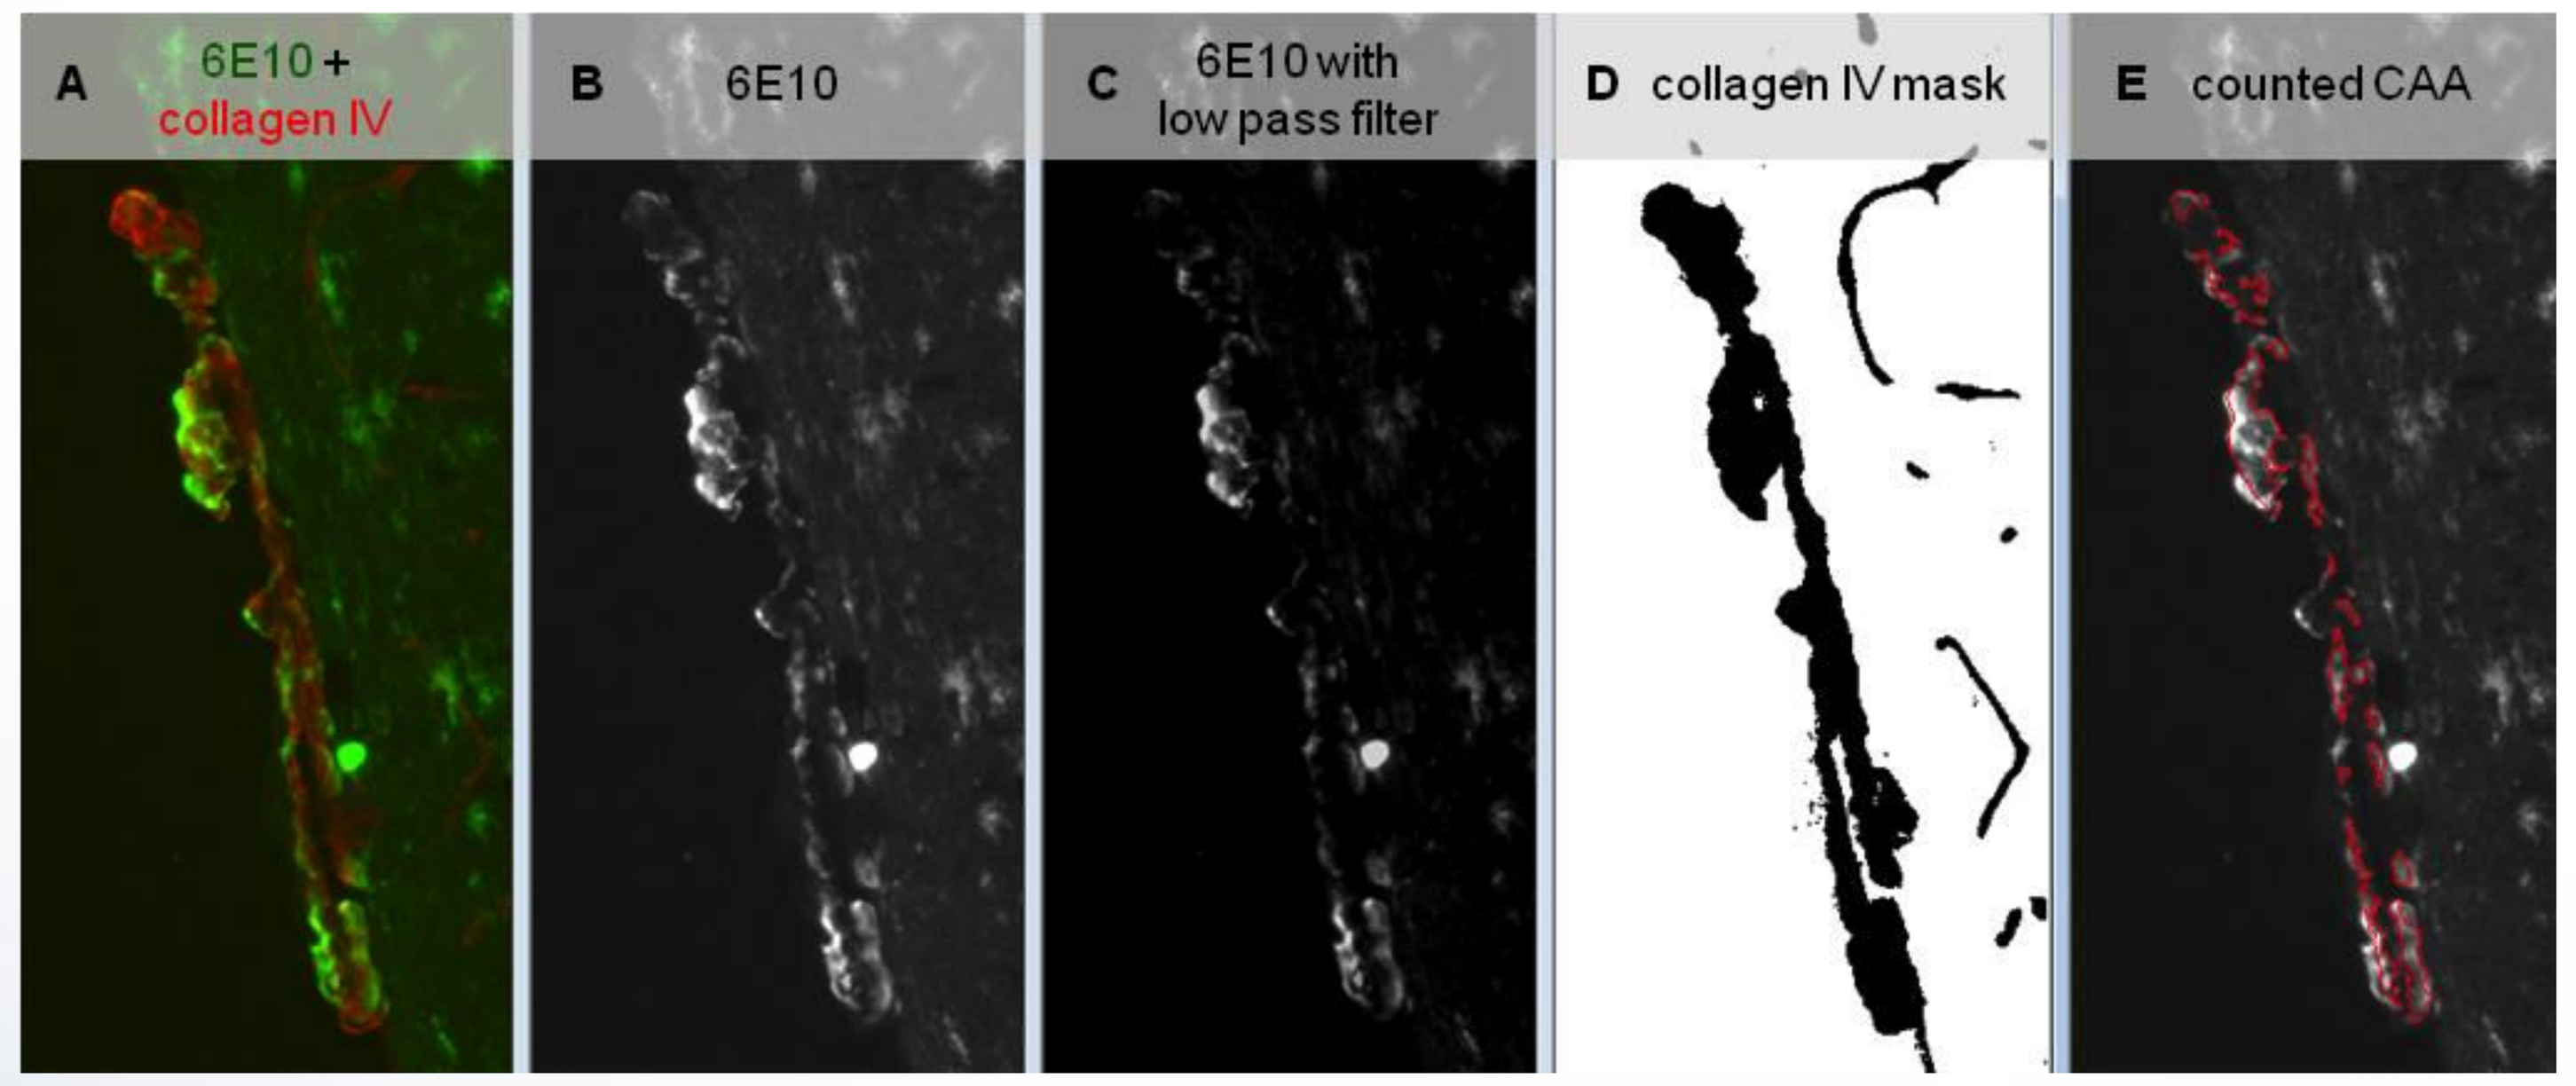
A 6E10 + collagen IV
B 6E10
C 6E10 with low pass filter
D collagen IV mask
E counted CAA

Fig. 1: Exemplary measurement process of CAA in the cortex of a 12 month old APP_{SL} mouse. (A) Composite image of 6E10 + collagen IV channels. **(B)** Single Aβ (6E10) labeling. **(C)** Modified human amyloid labeling with the low pass filter correction. **(D)** Inverted collagen IV mask. **(E)** Ultimate count of CAA (red outline).

RESULTS

Aβ accumulates in brain tissue of humans and both AD mouse models – APP_{SL} and 5xFAD

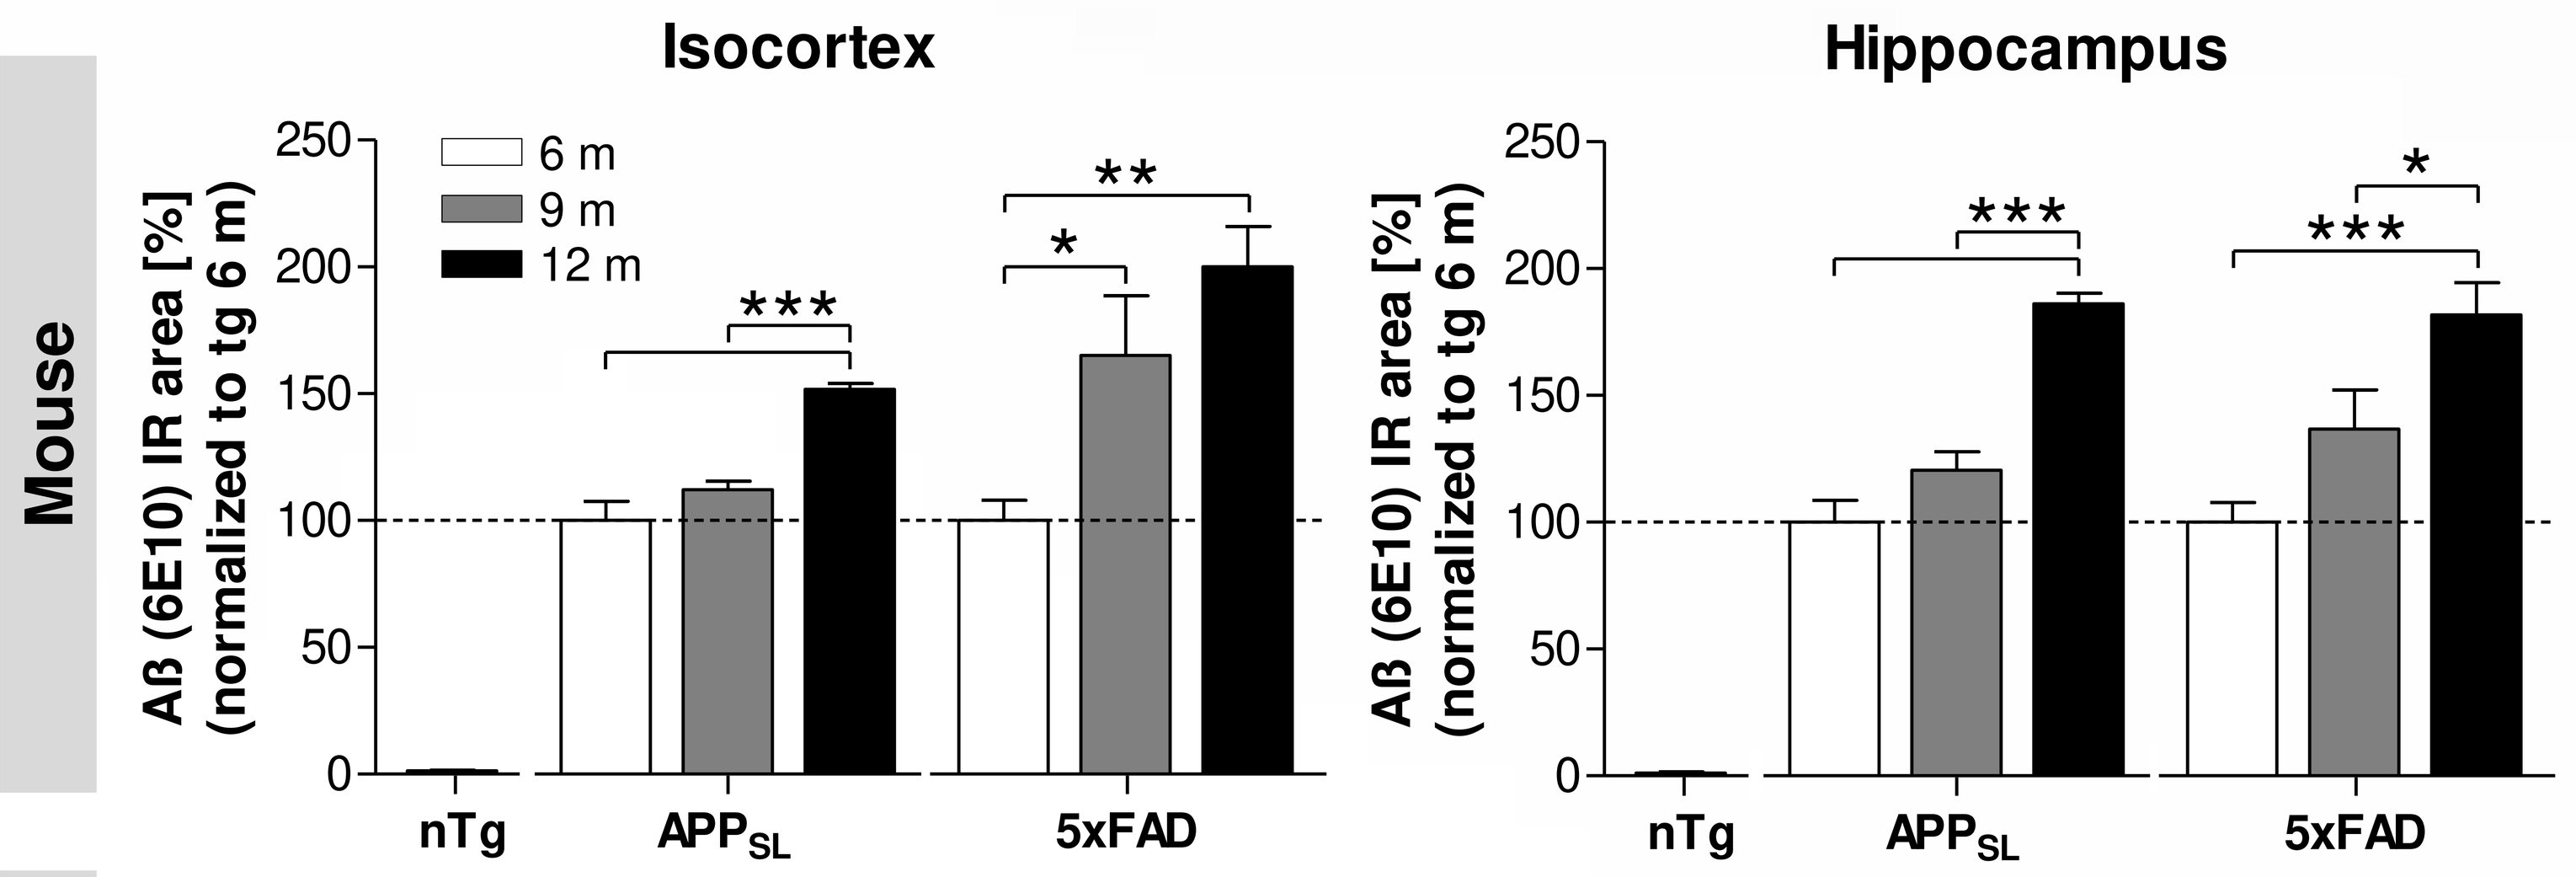
Mouse
Isocortex
Hippocampus
Aß (6E10) IR area [%] (normalized to tg 6 m)
6 m
9 m
12 m
nTg
APPSL
5xFAD

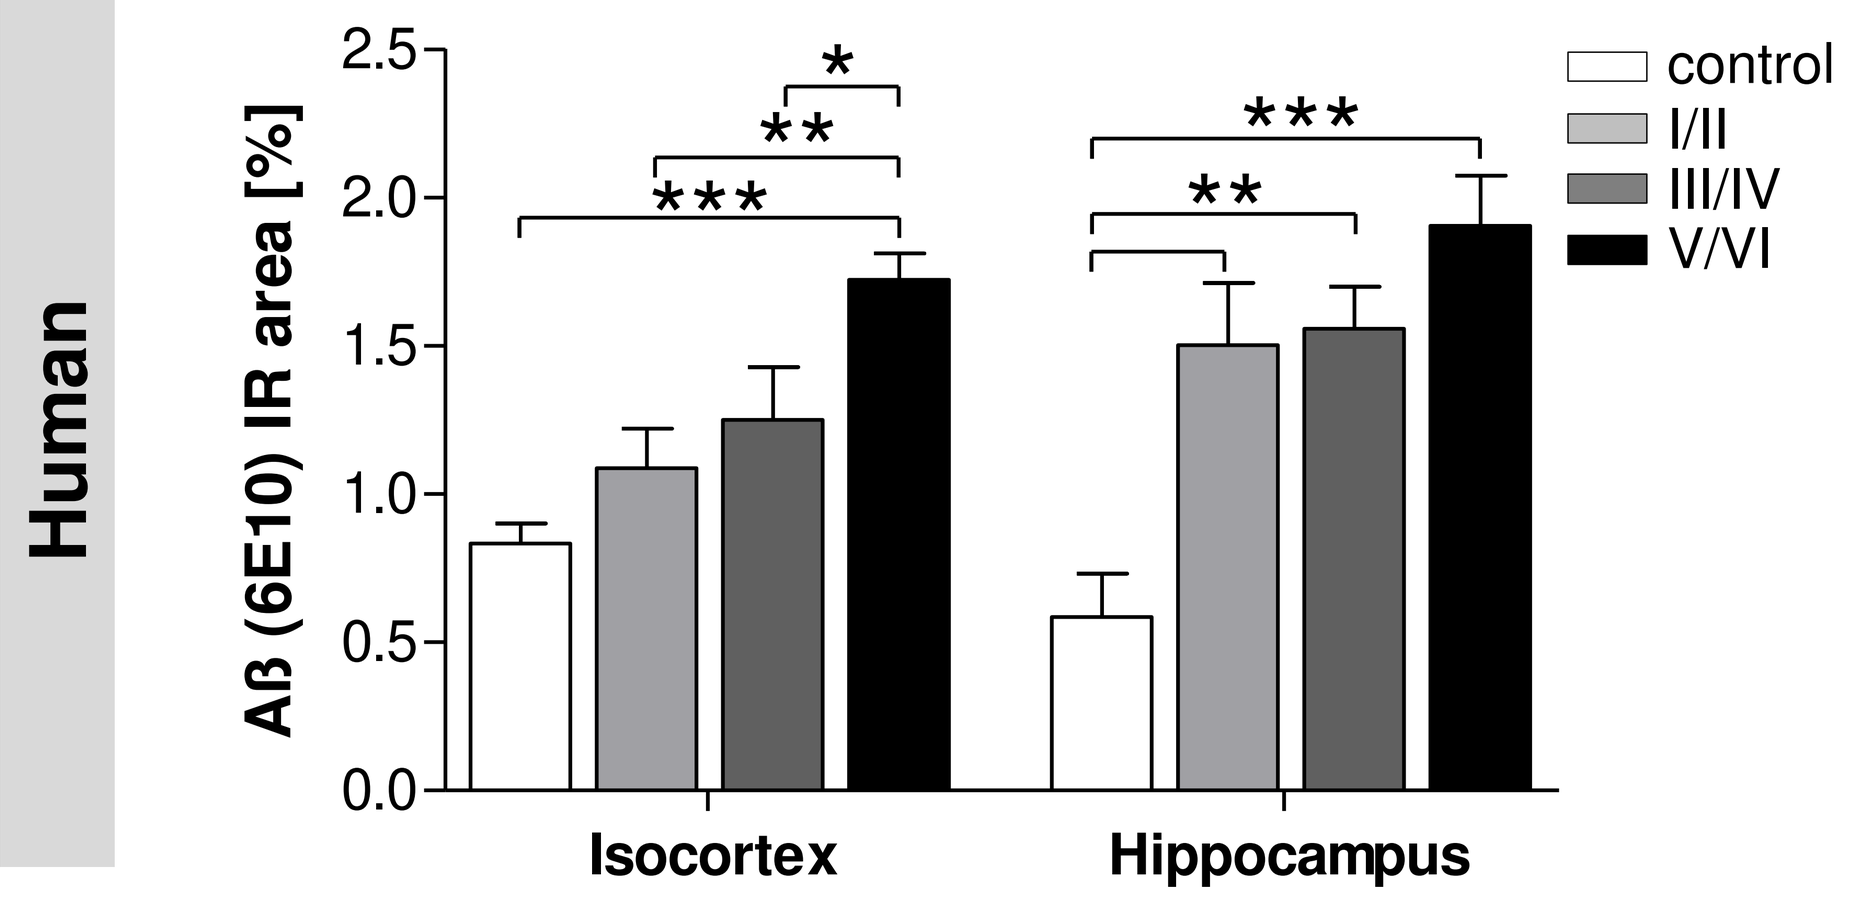
Human
Aß (6E10) IR area [%]
control
I/II
III/IV
V/VI
Isocortex
Hippocampus

Fig. 2: Histological quantification of Aβ accumulation in mouse and human brain tissue. One-way ANOVA; Newman-Keuls post hoc test; *p≤0.05, **p≤0.01, ***p≤0.001. m = months.

CAA builds up with age in APP_{SL} and 5xFAD mice

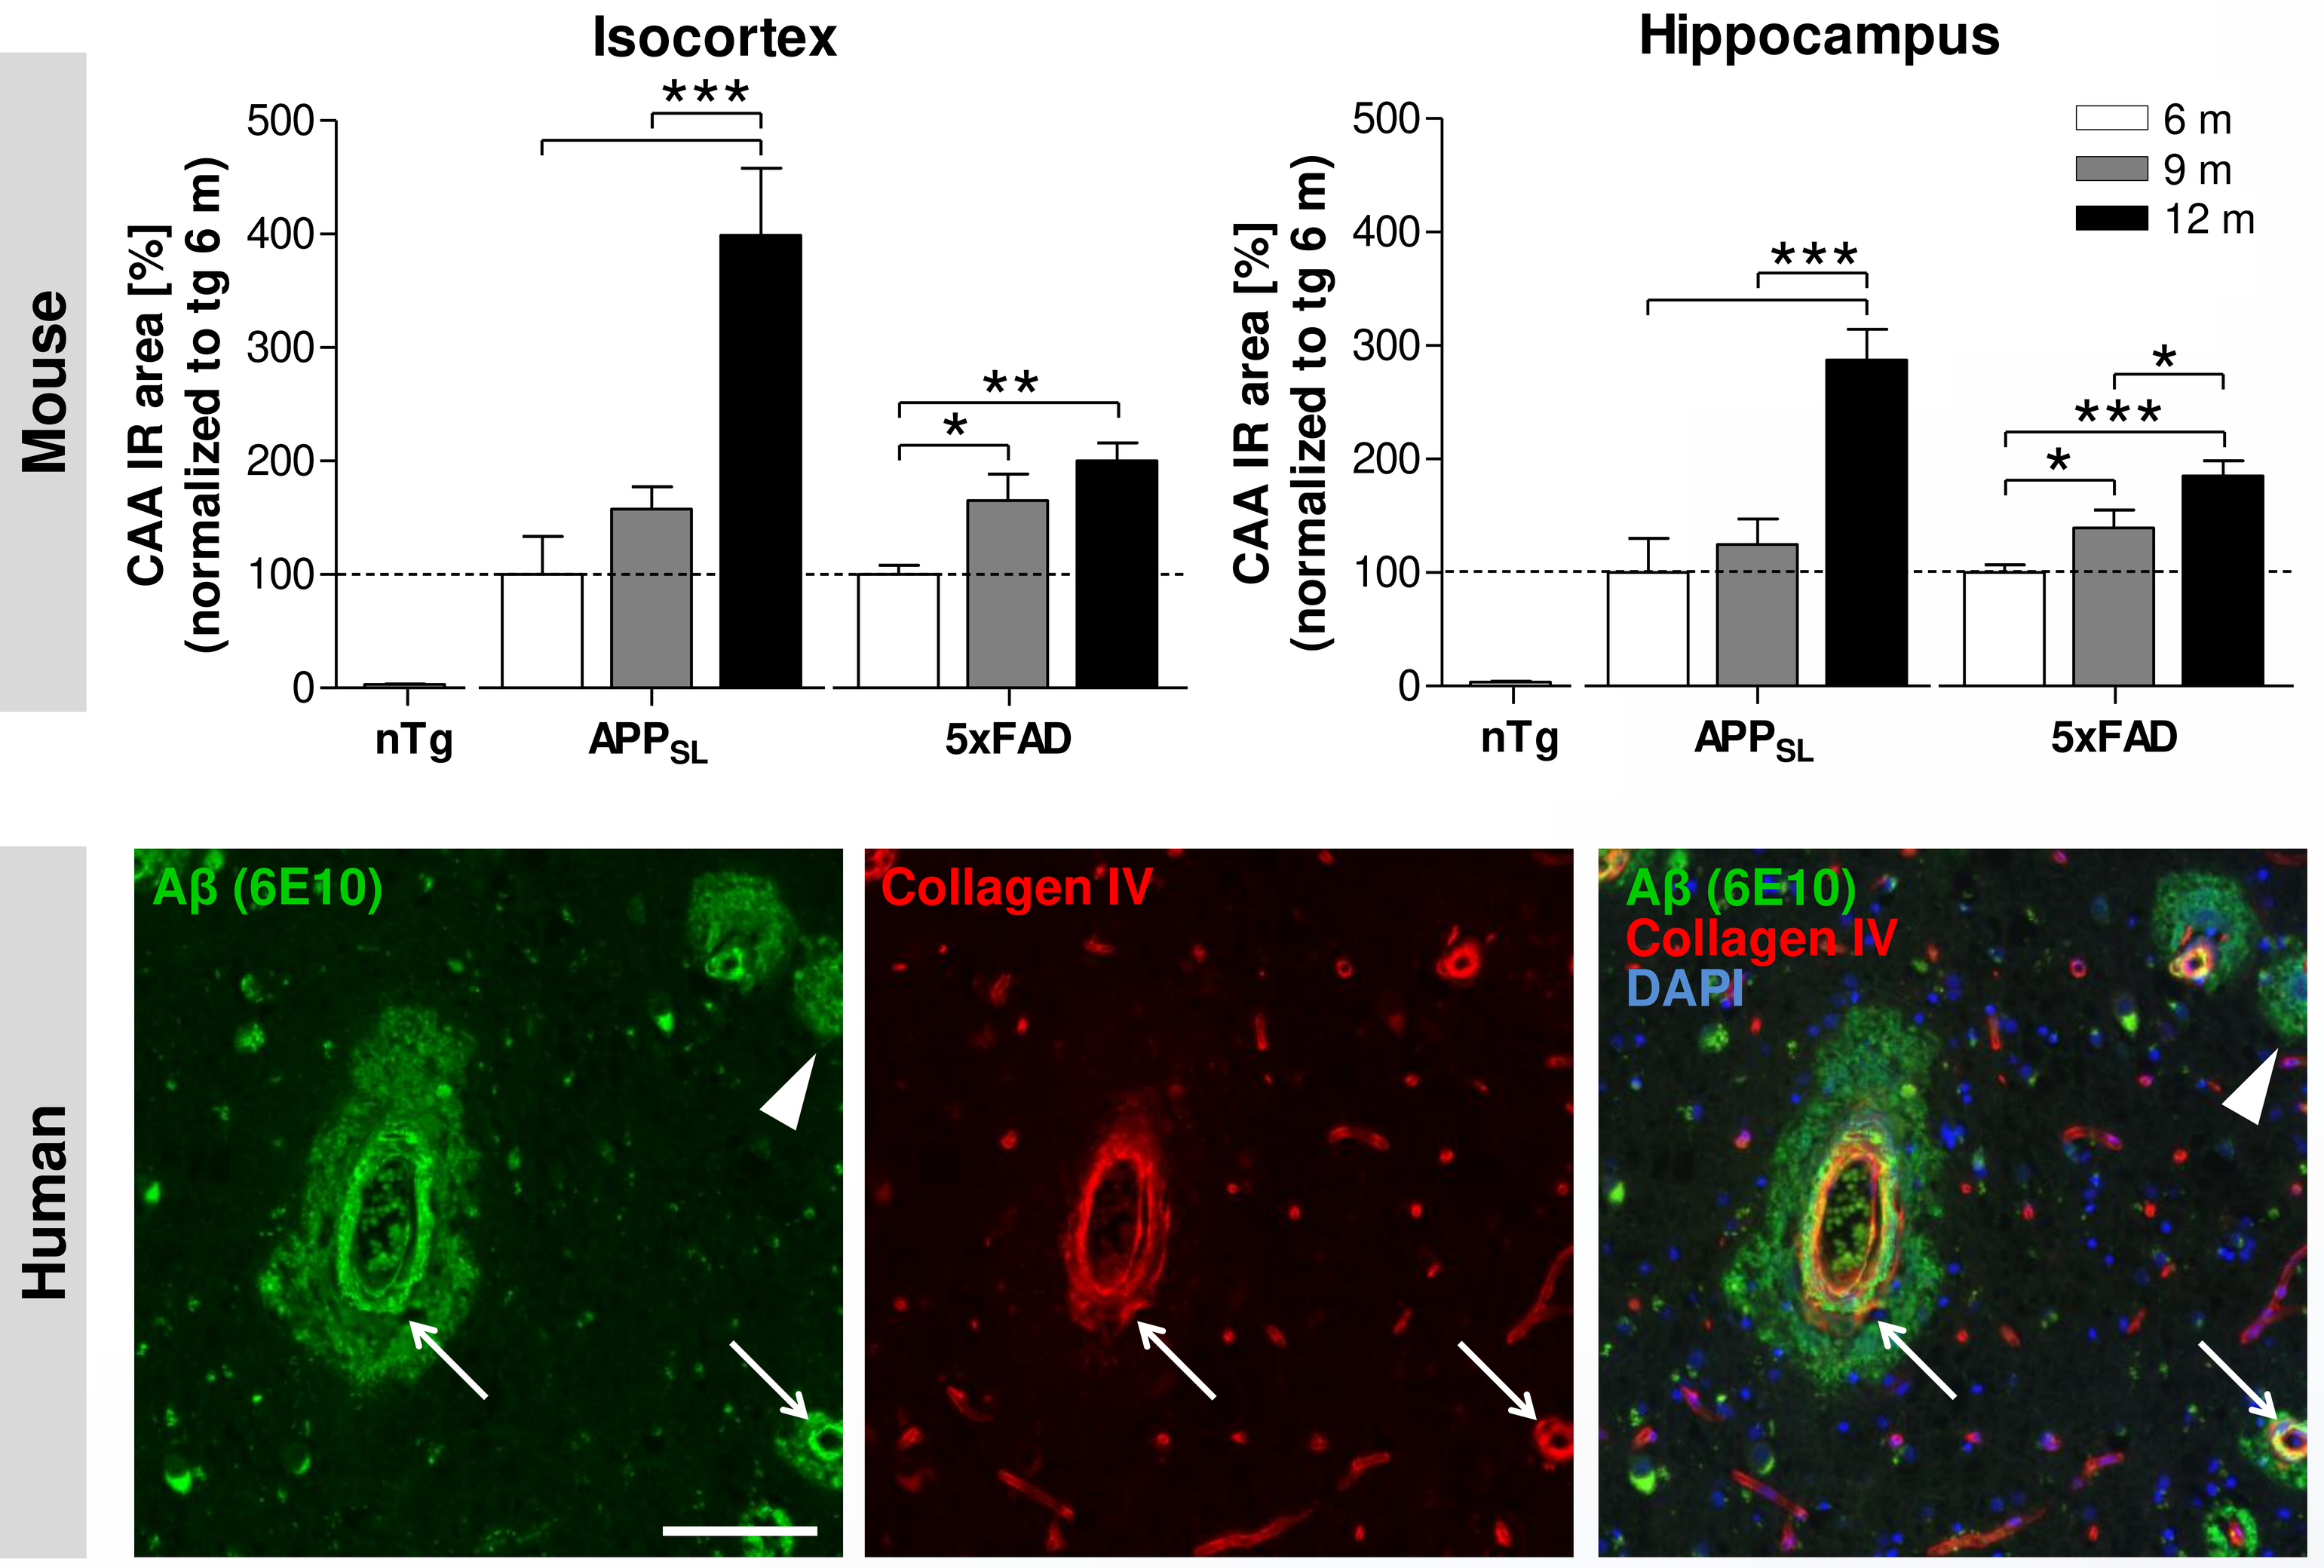
Isocortex
Hippocampus
Mouse
CAA IR area [%] (normalized to tg 6 m)
6 m
9 m
12 m
nTg
APPSL
5xFAD
Human
Aβ (6E10)
Collagen IV
DAPI

Fig. 3: Quantification of CAA progression in APP_{SL} and 5xFAD mice over age. Images show an example of CAA in the cortex of a human AD patient (Braak V/VI).
Scale bar: 100 µm; arrows: Aβ in blood vessels (CAA); arrowhead : parenchymal Aβ.
One-way ANOVA; Newman-Keuls post hoc test; *p≤0.05, **p≤0.01, ***p≤0.001,

CAA load per blood vessel increases over age in both AD mouse models

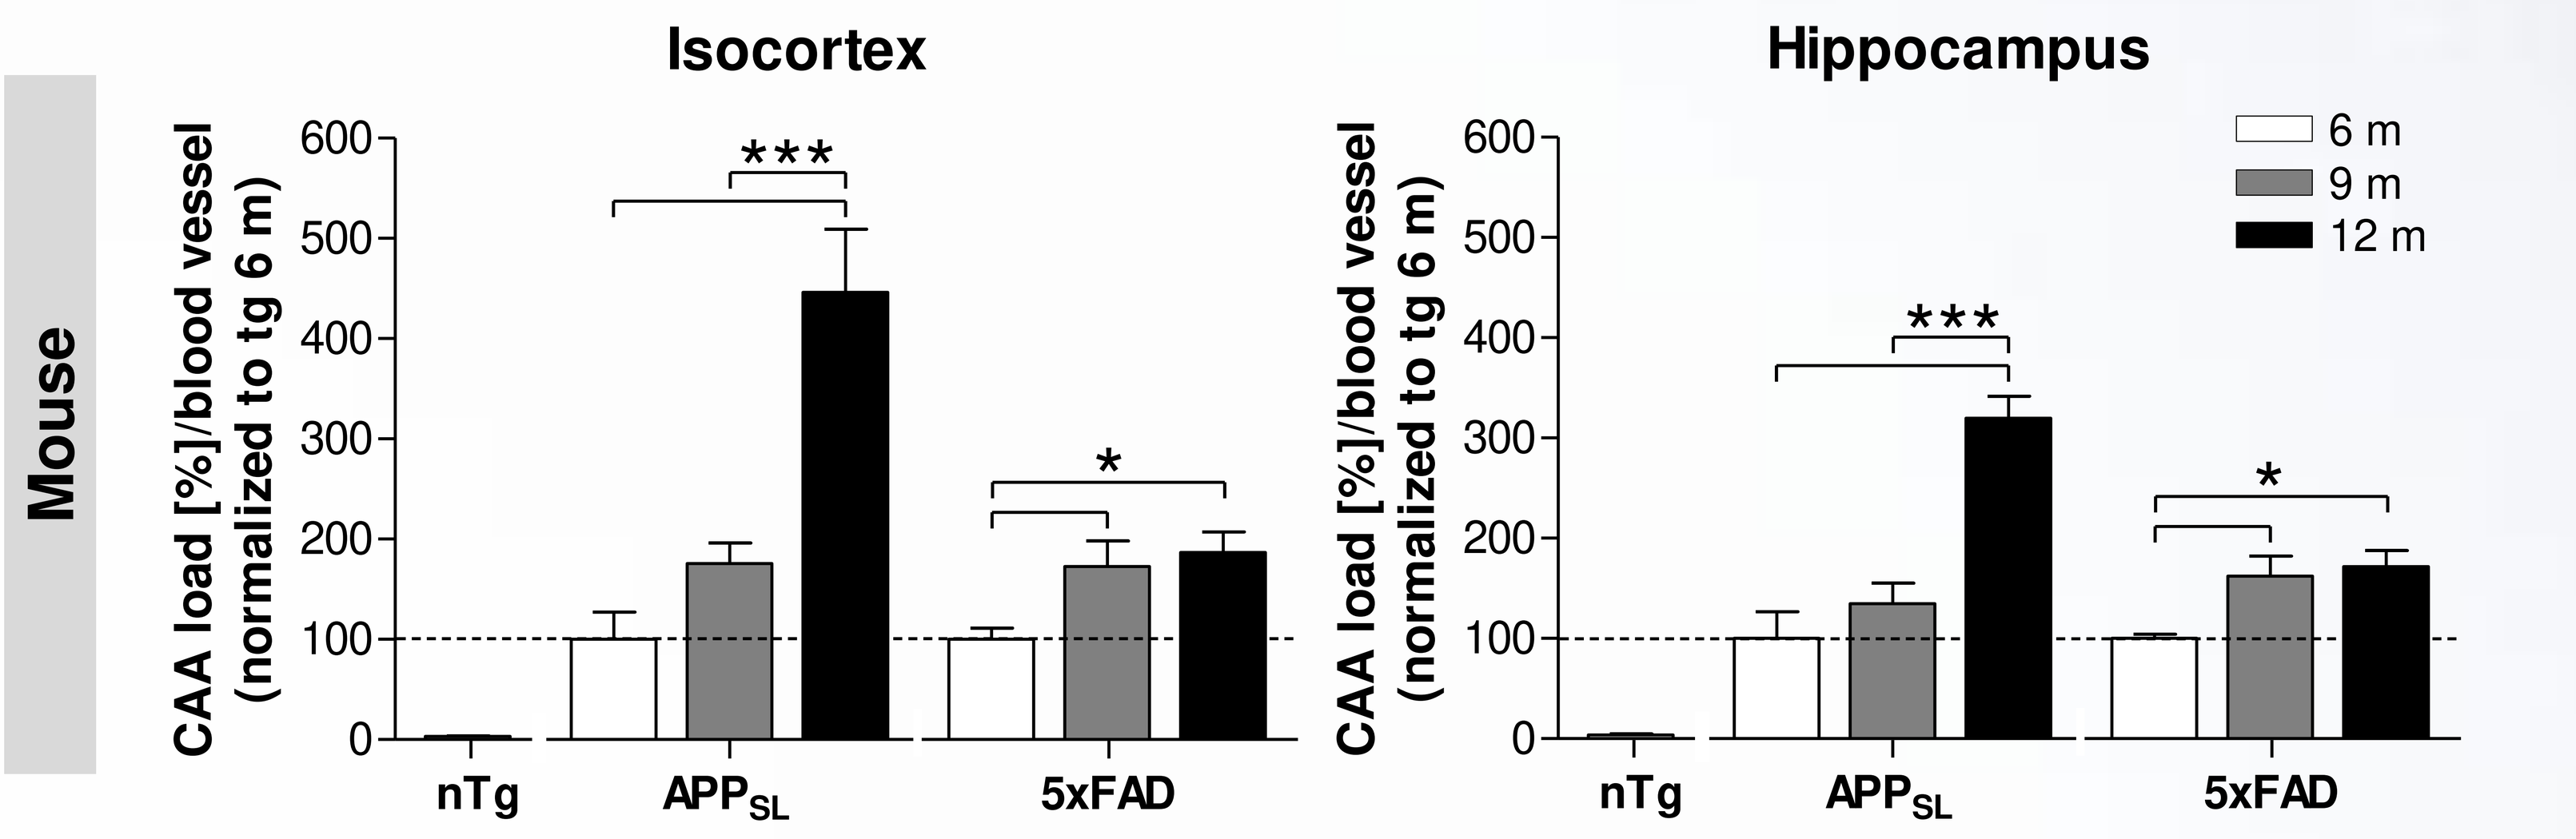
Isocortex
Hippocampus
Mouse
CAA load [%]/blood vessel (normalized to tg 6 m)
6 m
9 m
12 m
nTg
APPSL
5xFAD

Fig. 4: Quantification of CAA per blood vessel. One-way ANOVA; Newman-Keuls post hoc test; *p≤0.05, ***p≤0.001.

Collagen IV labeling intensity is associated with disease progression

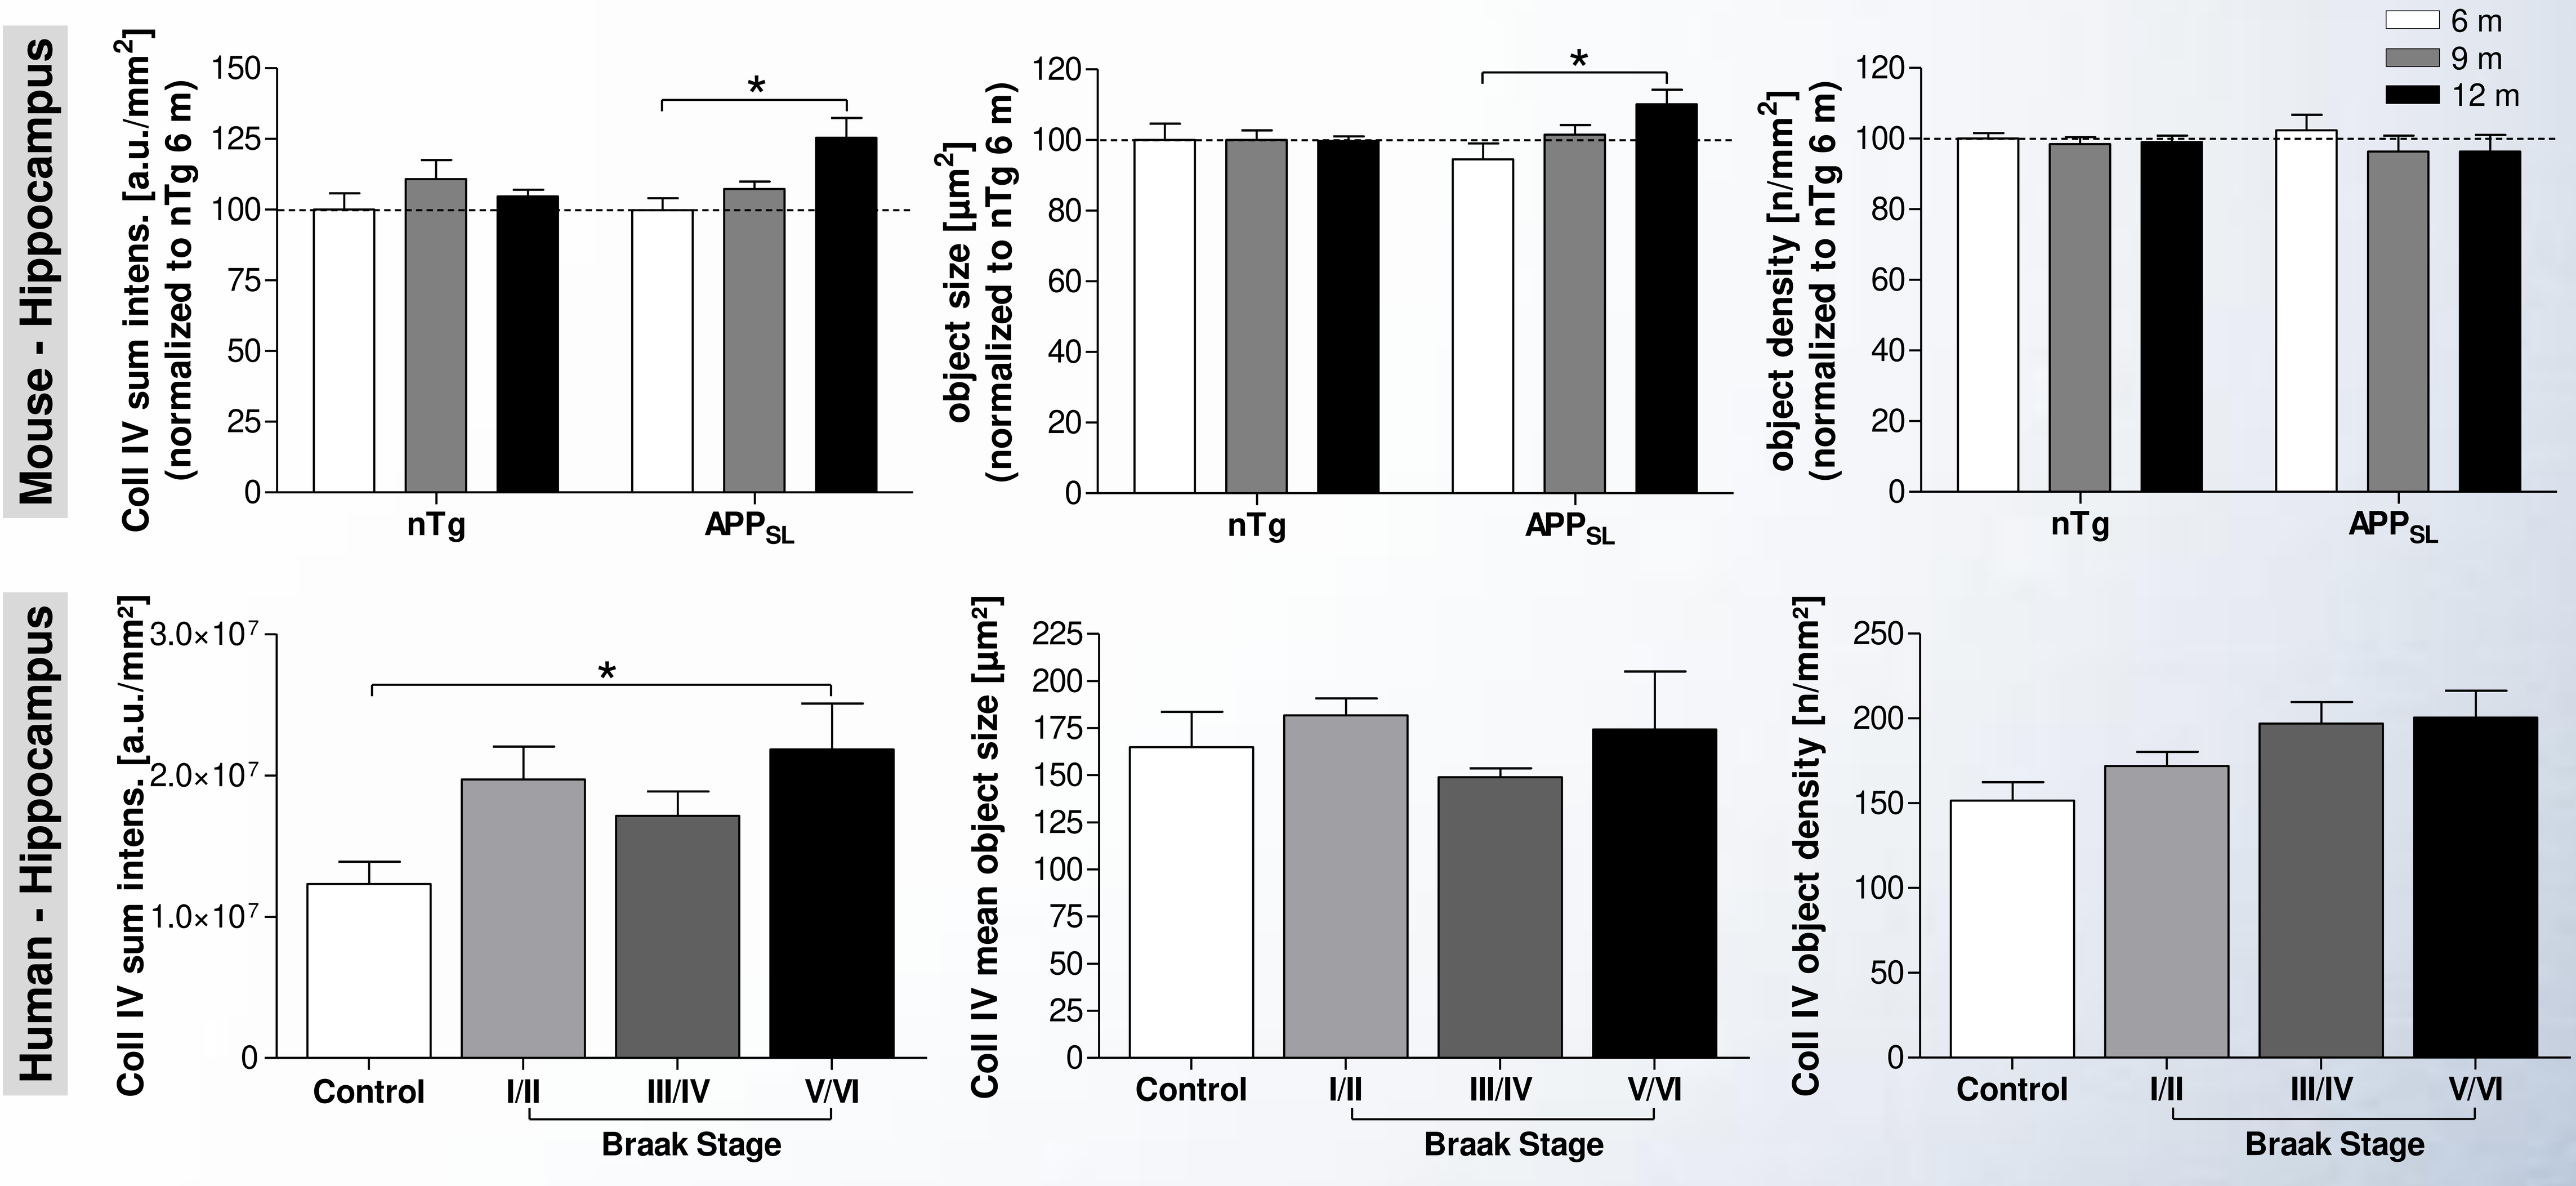
Mouse - Hippocampus
Coll IV sum intens. [a.u./mm²] (normalized to nTg 6 m)
object size [µm²] (normalized to nTg 6 m)
object density [n/mm²] (normalized to nTg 6 m)
6 m
9 m
12 m
nTg
APPSL
Human - Hippocampus
Coll IV sum intens. [a.u./mm²]
Coll IV mean object size [µm²]
Coll IV object density [n/mm²]
Control
I/II
III/IV
V/VI
Braak Stage

Fig. 5: Quantification of collagen IV in mouse (top row) and human (bottom row) hippocampi.
Mouse: Two-way ANOVA; Bonferroni post hoc test. Human: One-way ANOVA ; Newman-Keuls post hoc test. *p≤0.05, **p≤0.01, ***p≤0.001.

CONCLUSION

- **Aβ accumulation can be detected in mouse and human using the same approach.**
- **CAA increases significantly in APP_{SL} as well as in 5xFAD mice with age.**
- **Collagen IV levels are associated with disease progression in human and analyzed mouse models.**

Meet us at Booth #207

AAIC 2016, P3-14-296, Poster: #10816

Contact: Birgit Hutter-Paier | Director Neuropharmacology | QPS Austria GmbH | Parkring 12 | 8074 Grambach | Austria | birgit.hutter-paier@qps.com | www.qpsneuro.com

www.qpsneuro.com