Co-expression of Virally Induced P301L Tau and Transgenic β-Amyloid in APP Mice as Model of Alzheimer's Disease to Closer Mimic Human Pathology

Ewald Auer[1], Meritxell Aguiló[1,2], Vera Niederkofler[1], Nicole Taub[1], Joerg Neddens[1], Robert Zimmermann[2], Birgit Hutter-Paier[1]

[1]QPS Austria GmbH, Parkring 12, 8074 Grambach, Austria [2] Institute of Molecular Biosciences, University of Graz, Graz, Austria

QPS® helps you navigate

BACKGROUND

Major histopathological features of Alzheimer´s disease (AD) are: (1) Neurofibrillary tangles (NFT) of hyperphosphorylated Tau aggregates, and (2) senile plaques of insoluble β- amyloid (Aβ).

To generate a mouse model that combines these two hallmarks we virally induced expression of P301L human Tau in the hippocampus of APP_{SL} transgenic mice by stereotactic injection. Human P301L Tau increases NFT and is the main cause of *Frontotemporal Dementia with Parkinsonism-17* (FTDP-17) (*Hutton et al., 1998, Spillantini et al., 1998).*

We investigated spatiotemporal patterns of histopathology with a focus on potential interaction between both markers that are associated with human AD pathology.

MATERIALS AND METHODS

Recombinant adeno-associated virus serotype 9 (AAV9) with human P301L Tau gene under the control of CMV promoter was injected into the hippocampus of 3 months old male APP_{SL} mice (AP: -1.8; ML: +-1.4; DV: -1.4).

- immunohistological and biochemical analyses were performed using:
- human Tau (Tau 13, Enzo Life Sciences)
- hyperphosphorylated Tau:
 AT100 (Pierce Biotechnology), AT180 (Thermo Scientific), Ser262 (MBL), PHF-13 (Cell Signaling), EPR2603 (abcam)
- aggregated beta amyloid (LOC, abcam)
- APP (6E10, BioLegend)

Meet QPS at AAIC 2016

Booth #207

RESULTS

Stable expression of human P301L Tau in hippocampus

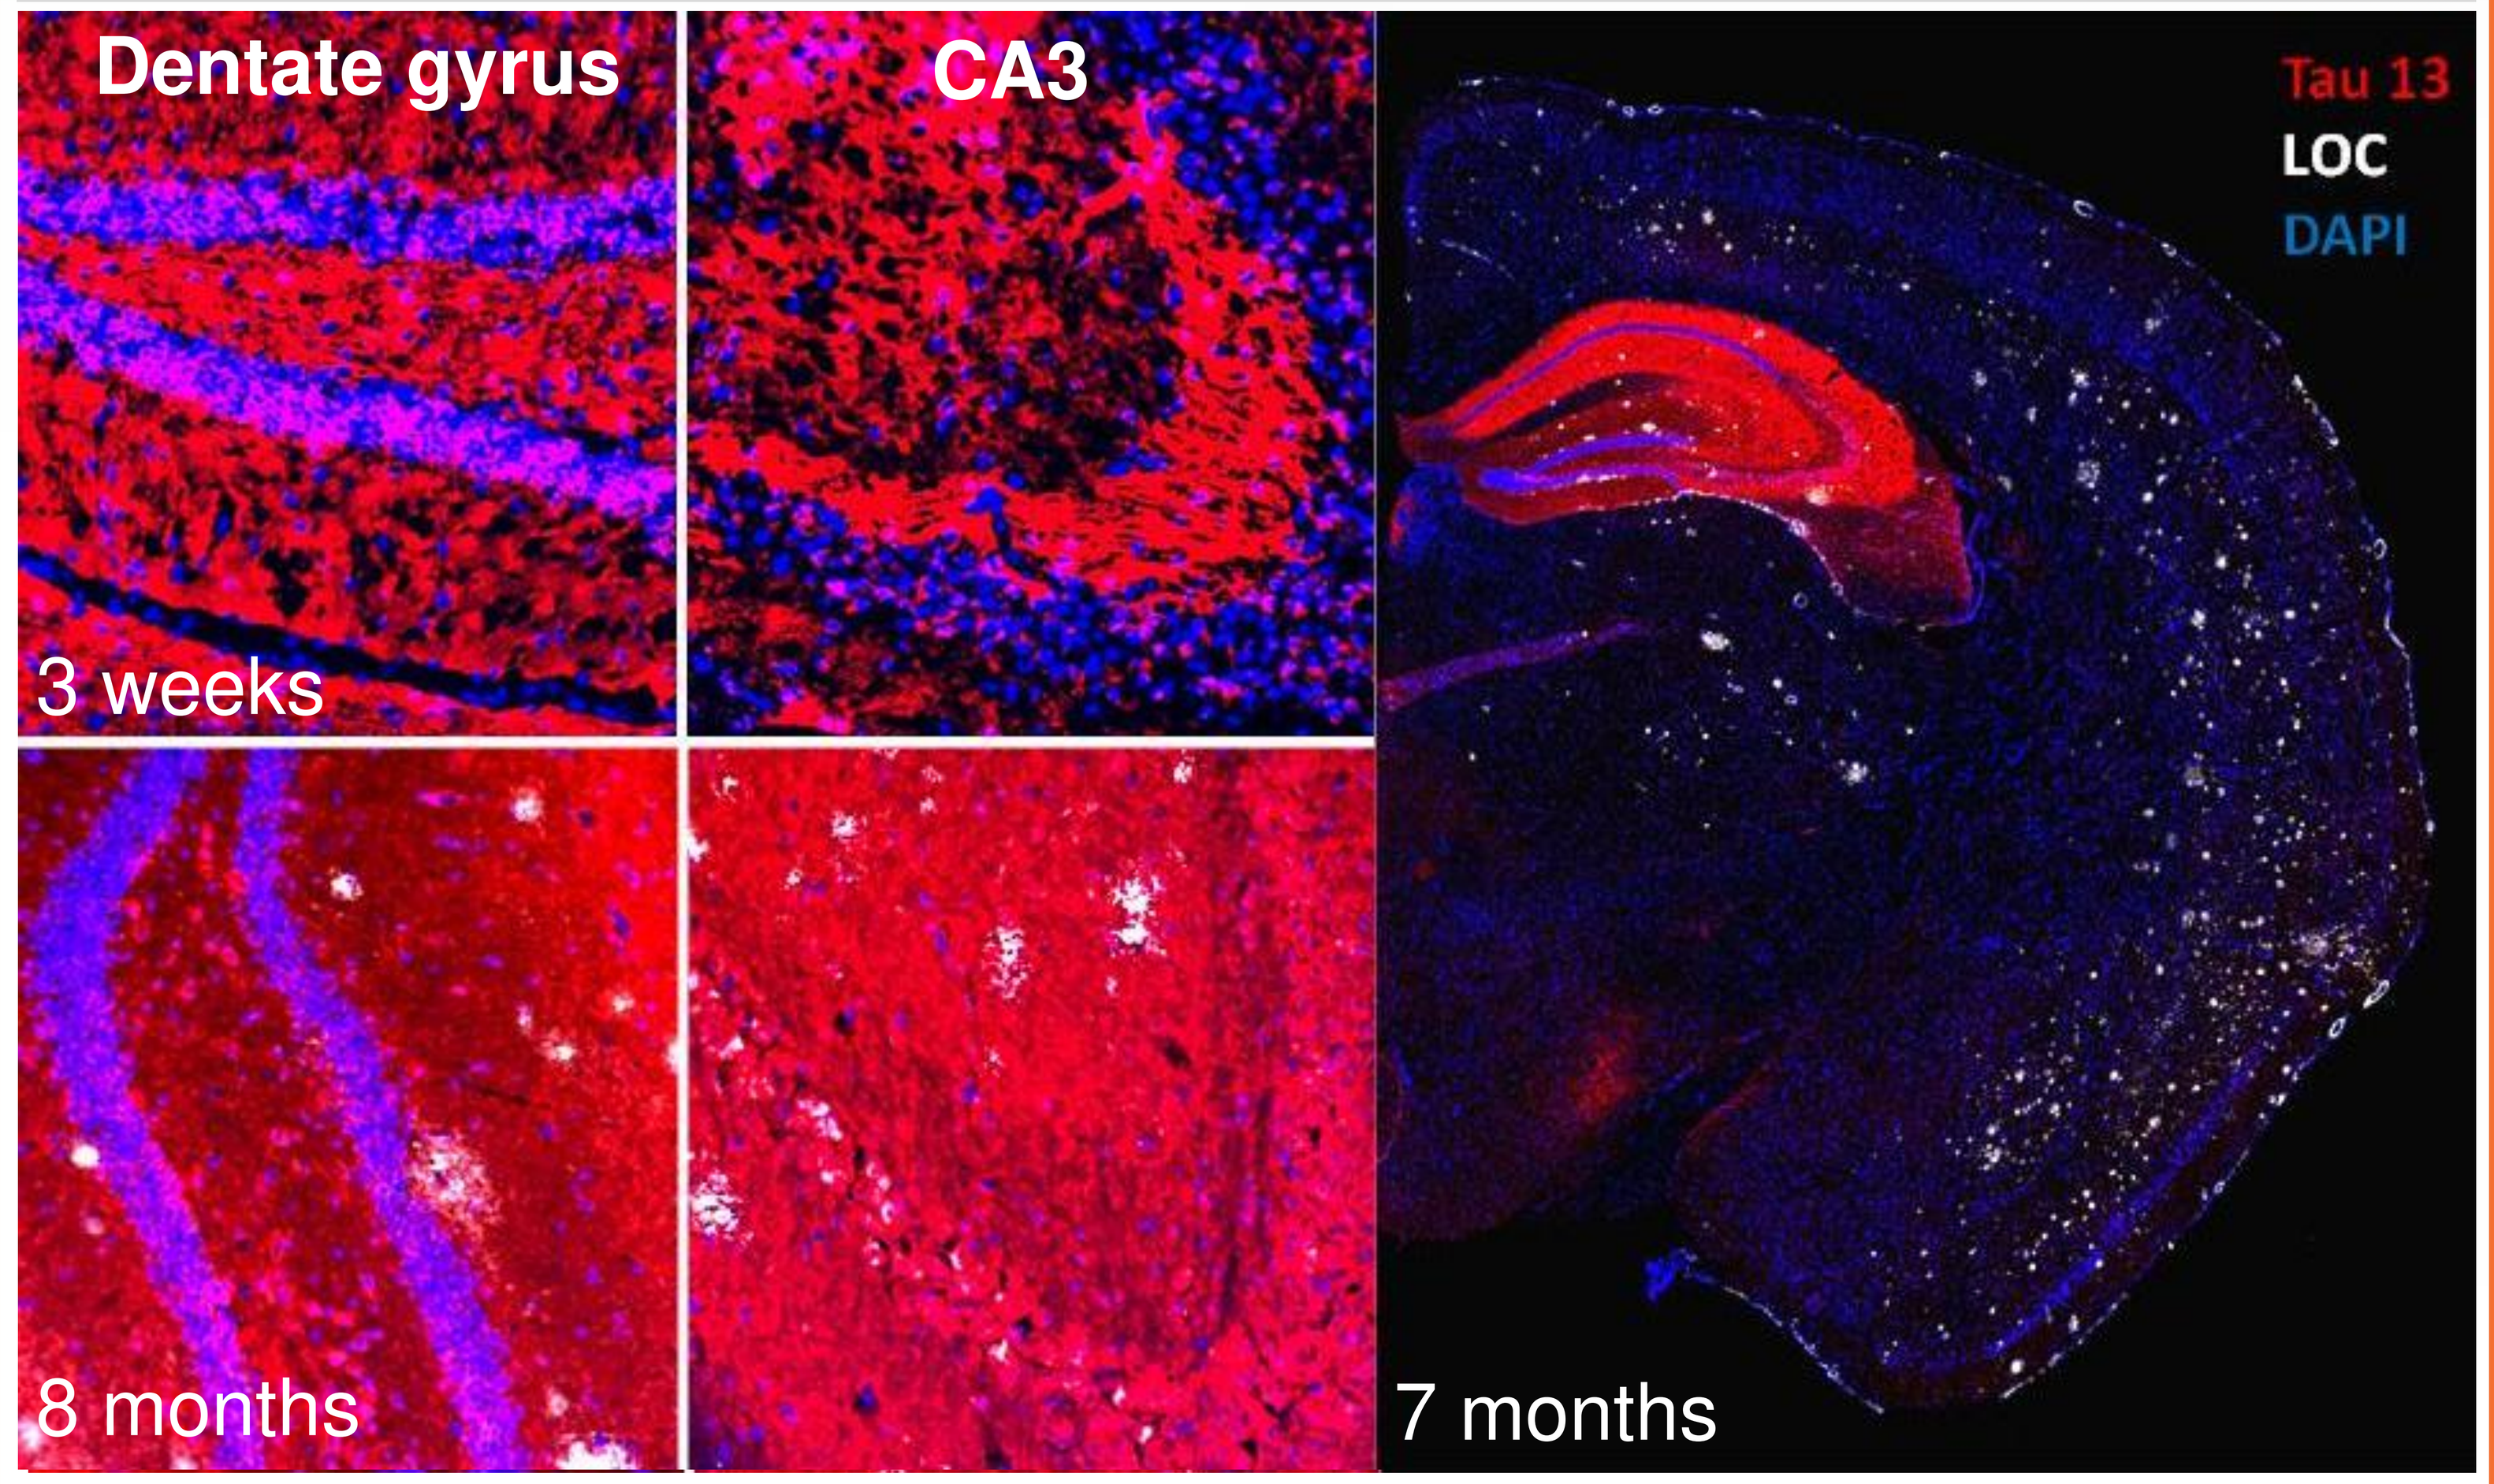
Dentate gyrus
CA3
3 weeks
8 months
Tau 13
LOC
DAPI
7 months

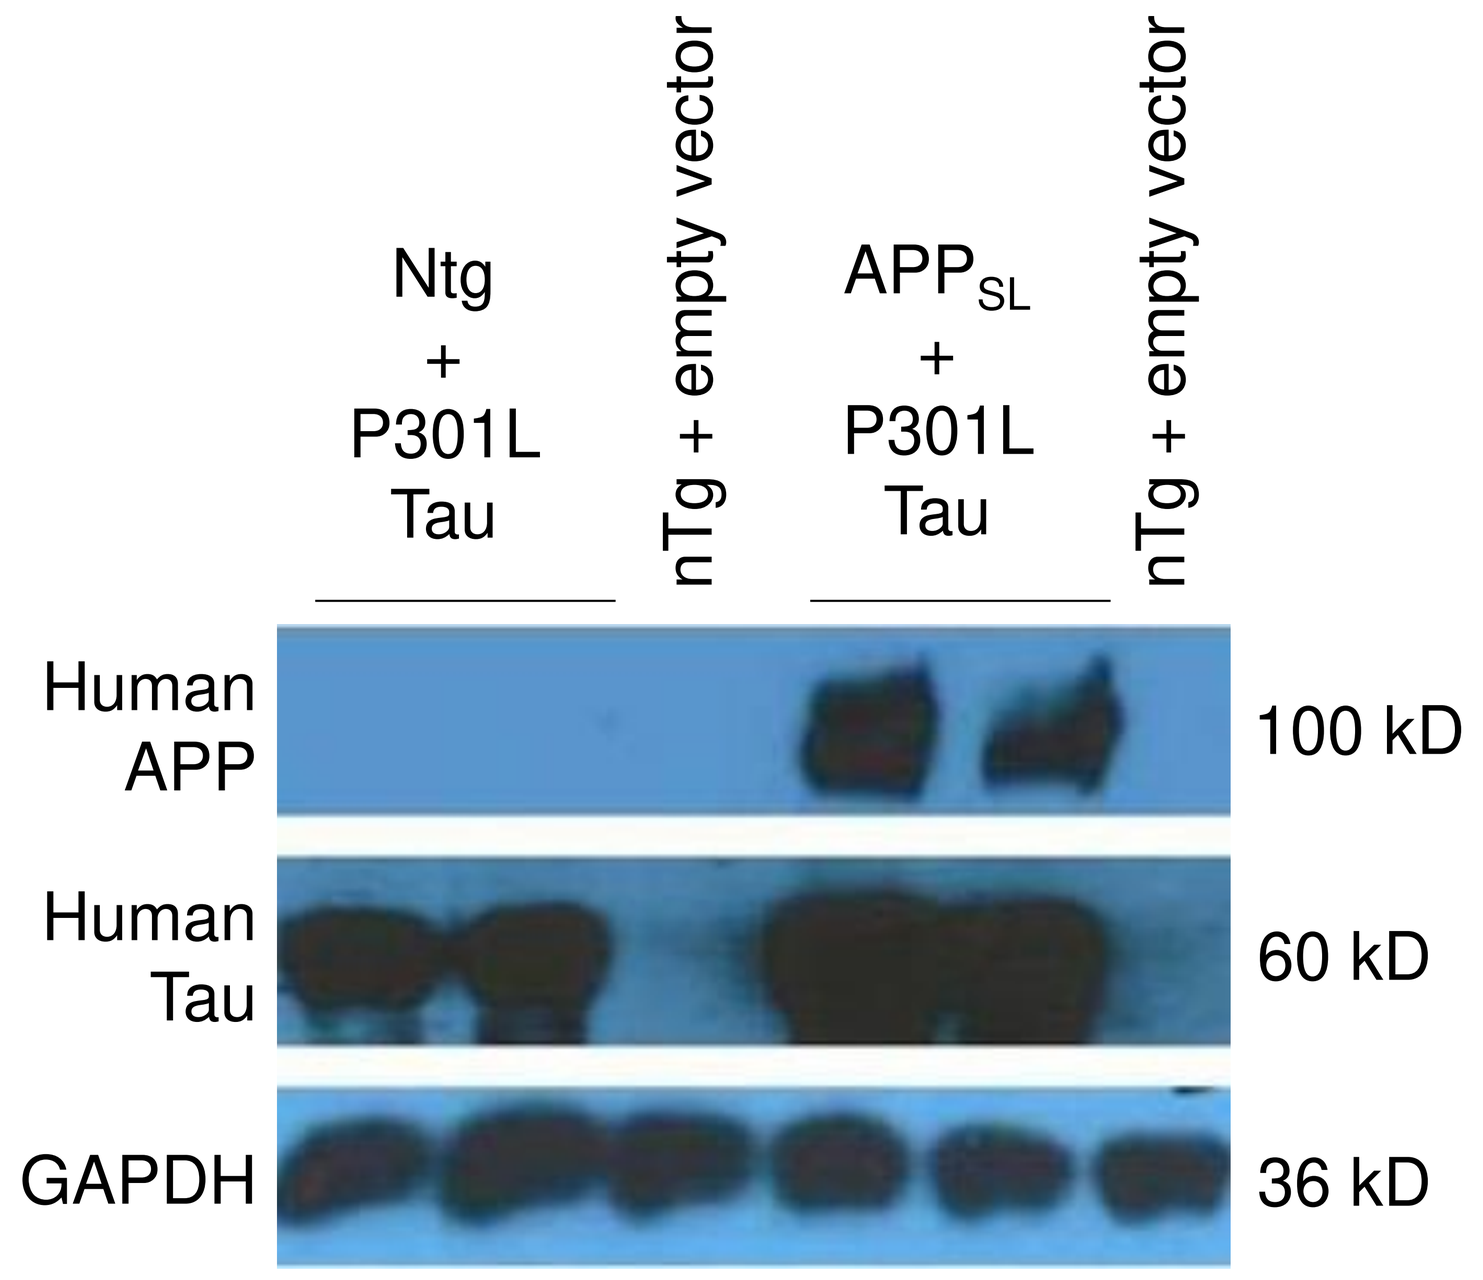
Ntg + P301L Tau
nTg + empty vector
APP_SL + P301L Tau
nTg + empty vector
Human APP
Human Tau
GAPDH
100 kD
60 kD
36 kD

Figure 1. Concomitant occurrence of P301L human tau and amyloid plaques.

(Top) Induced P301L Tau was already widely detected 3 weeks after injection and remained stable up to 8 months after surgery. The image of an entire hemi-brain shows that Tau expression is restricted to the hippocampus. Tau expression was confirmed 3 weeks, and 2, 3, 4, 7 and 8 months after injection.

(Left) High levels of virally expressed human Tau are detected by Tau 13 in Western blot, whereas empty vector yields no signal. Co-expression of transgenic human APP and human Tau is confirmed in APP_{SL} mice.

Rapid spread of virally induced pathological Tau

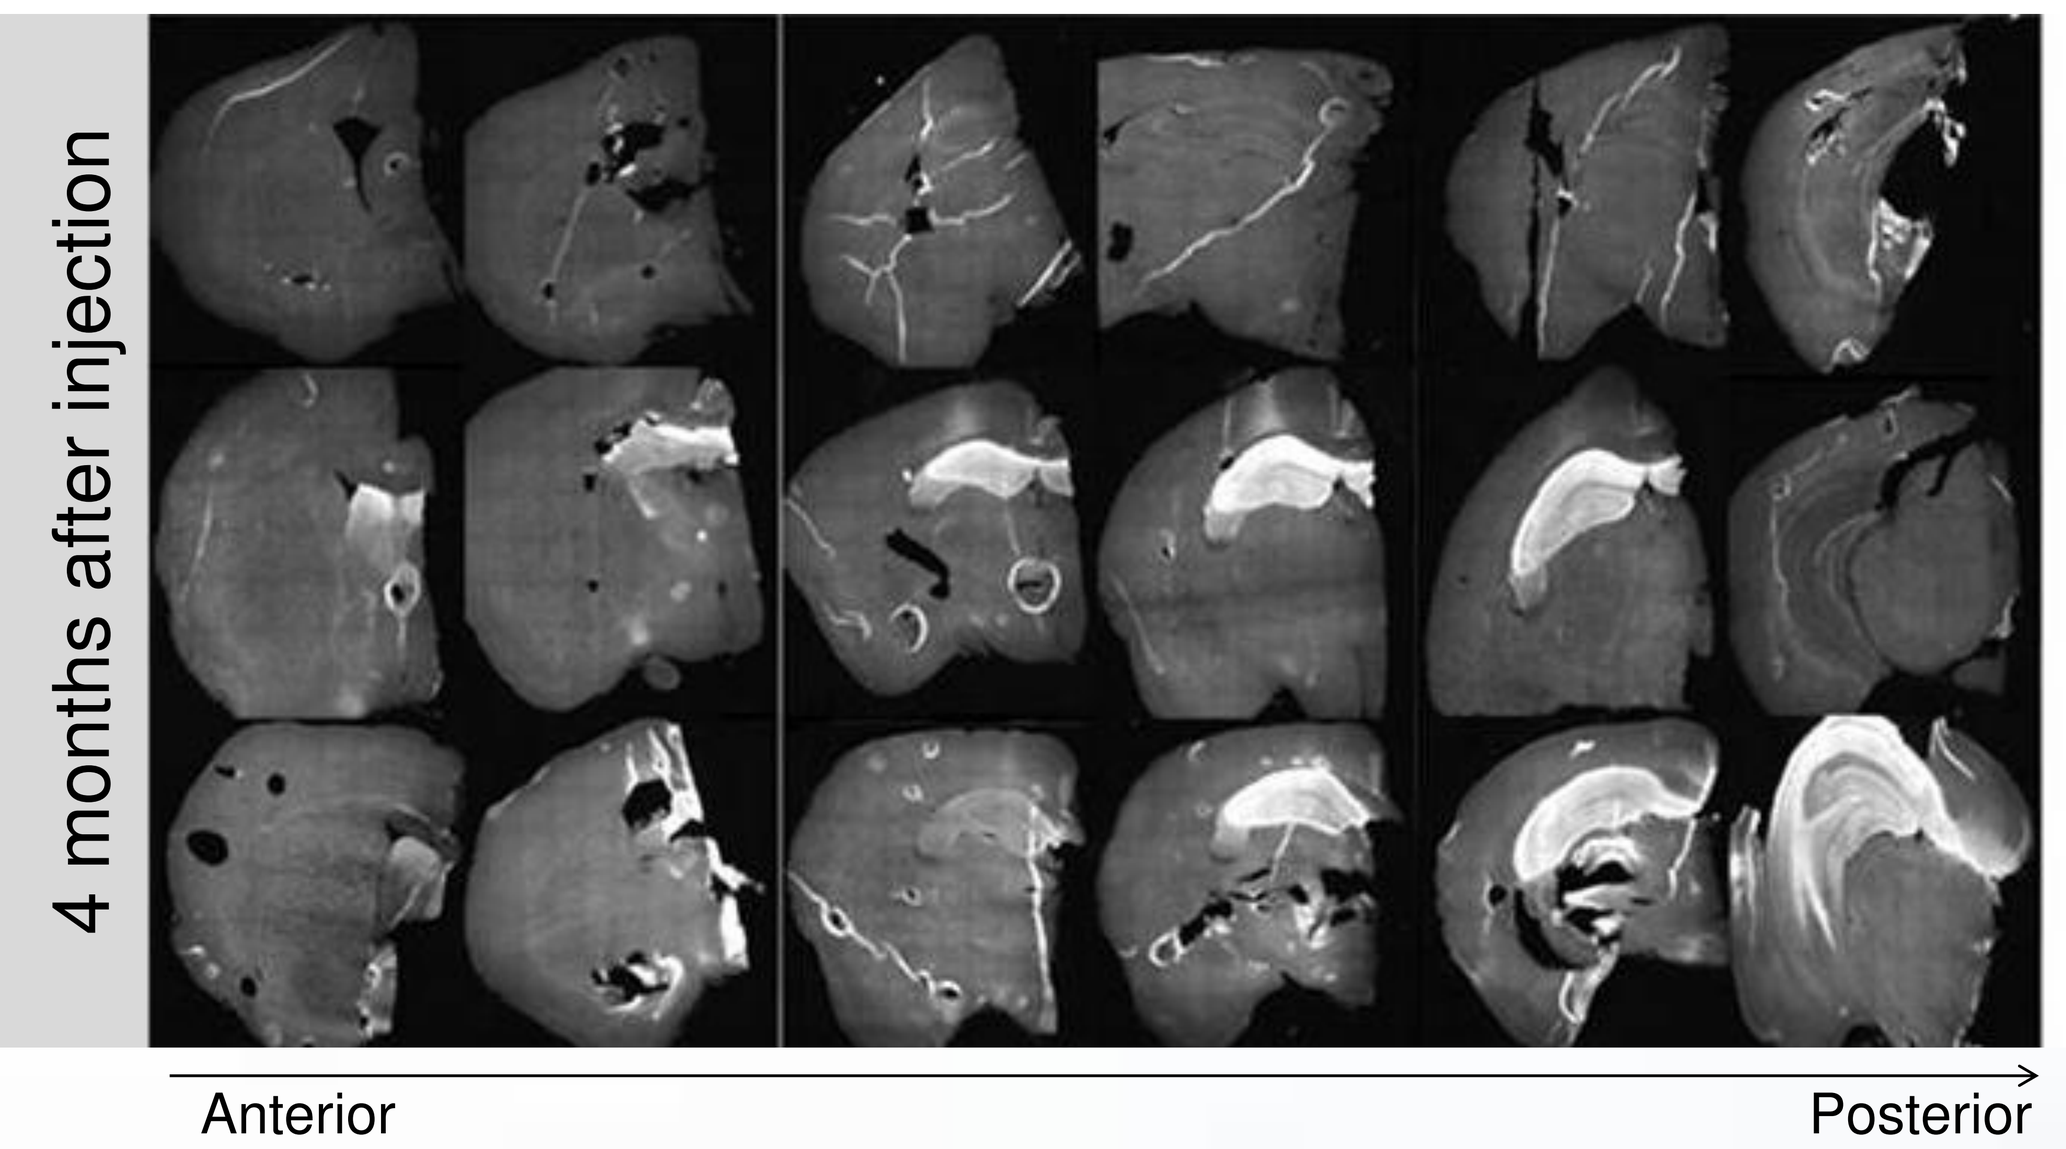
4 months after injection
Ntg + empty vector
Ntg + P301L Tau
APP_SL + P301L Tau
Anterior
Posterior

Figure 2. Human P301L Tau expression.
Although focally injection in the CA1, human Tau expression was evident throughout the entire hippocampal formation within 3 weeks.

P301L Tau is aberrantly phosphorylated at several residues

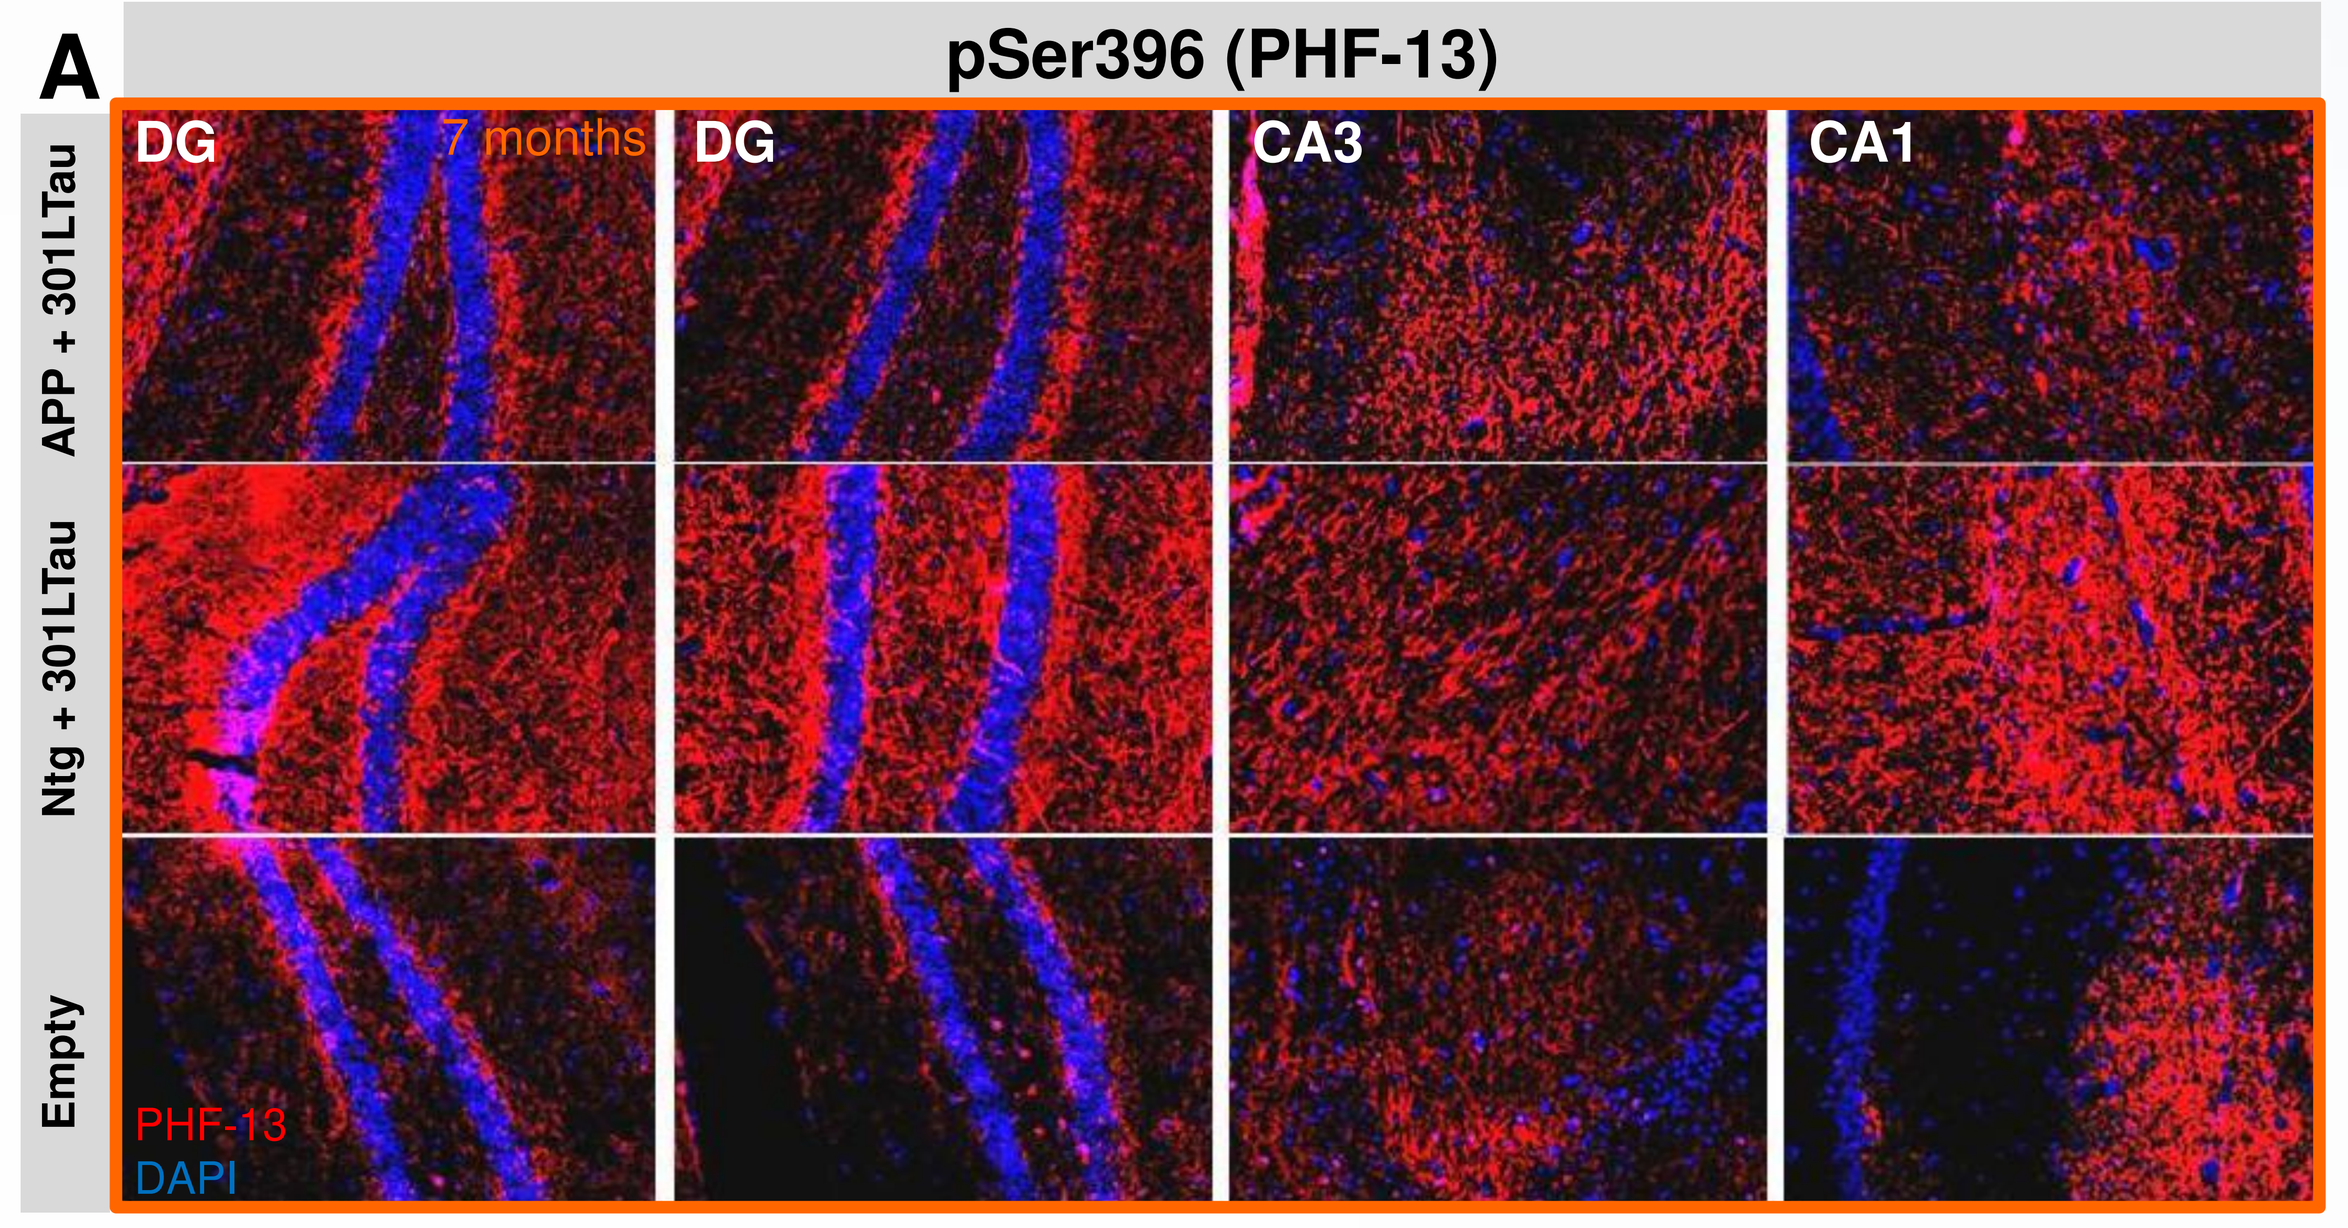
A
pSer396 (PHF-13)
DG
7 months
DG
CA3
CA1
APP + 301LTau
Ntg + 301LTau
Empty
PHF-13
DAPI

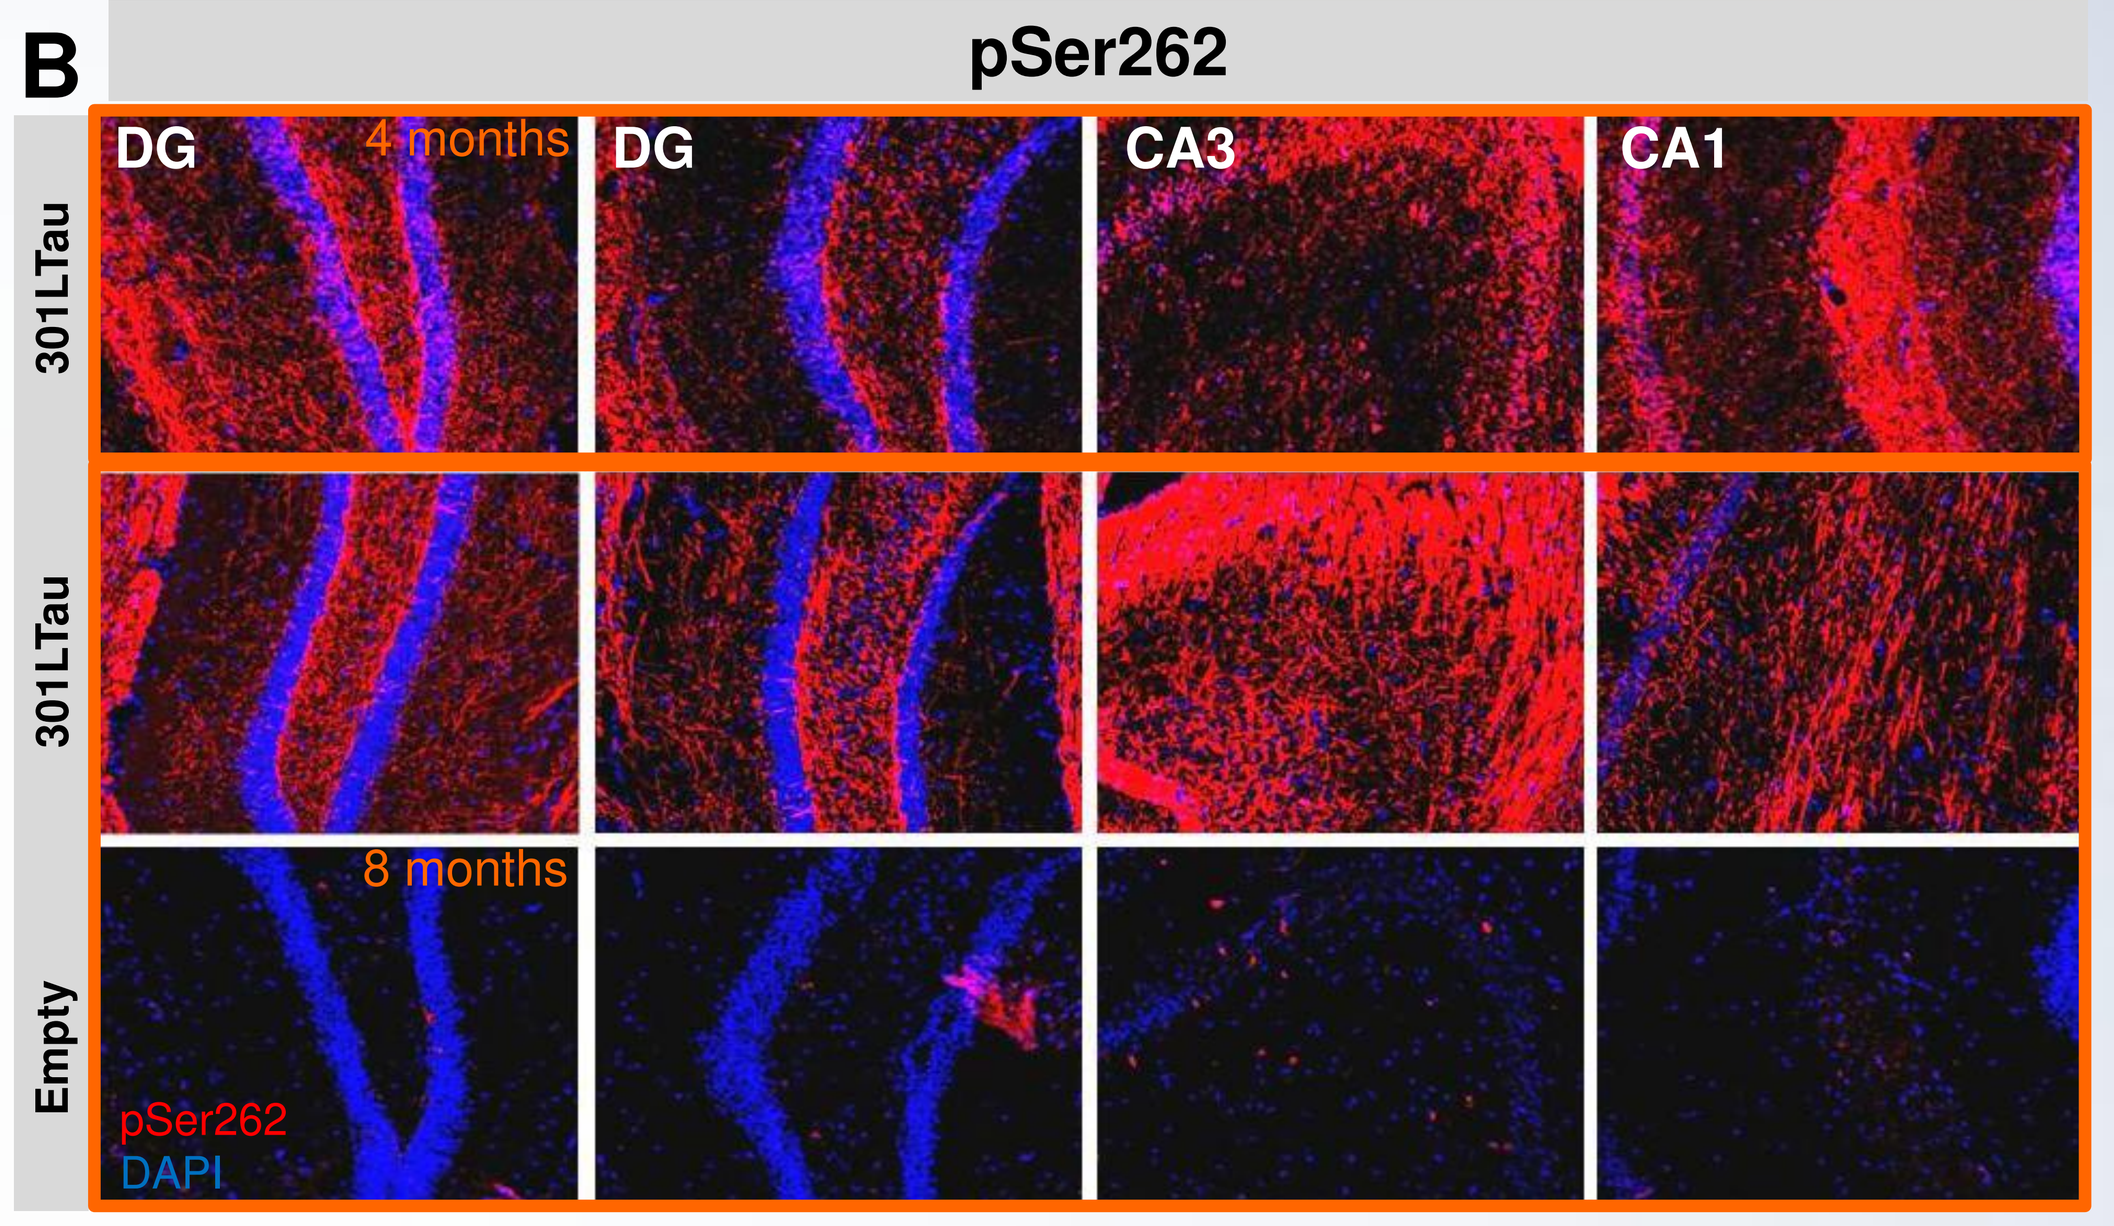
B
pSer262
DG
4 months
DG
CA3
CA1
301LTau
301LTau
Empty
8 months
pSer262
DAPI

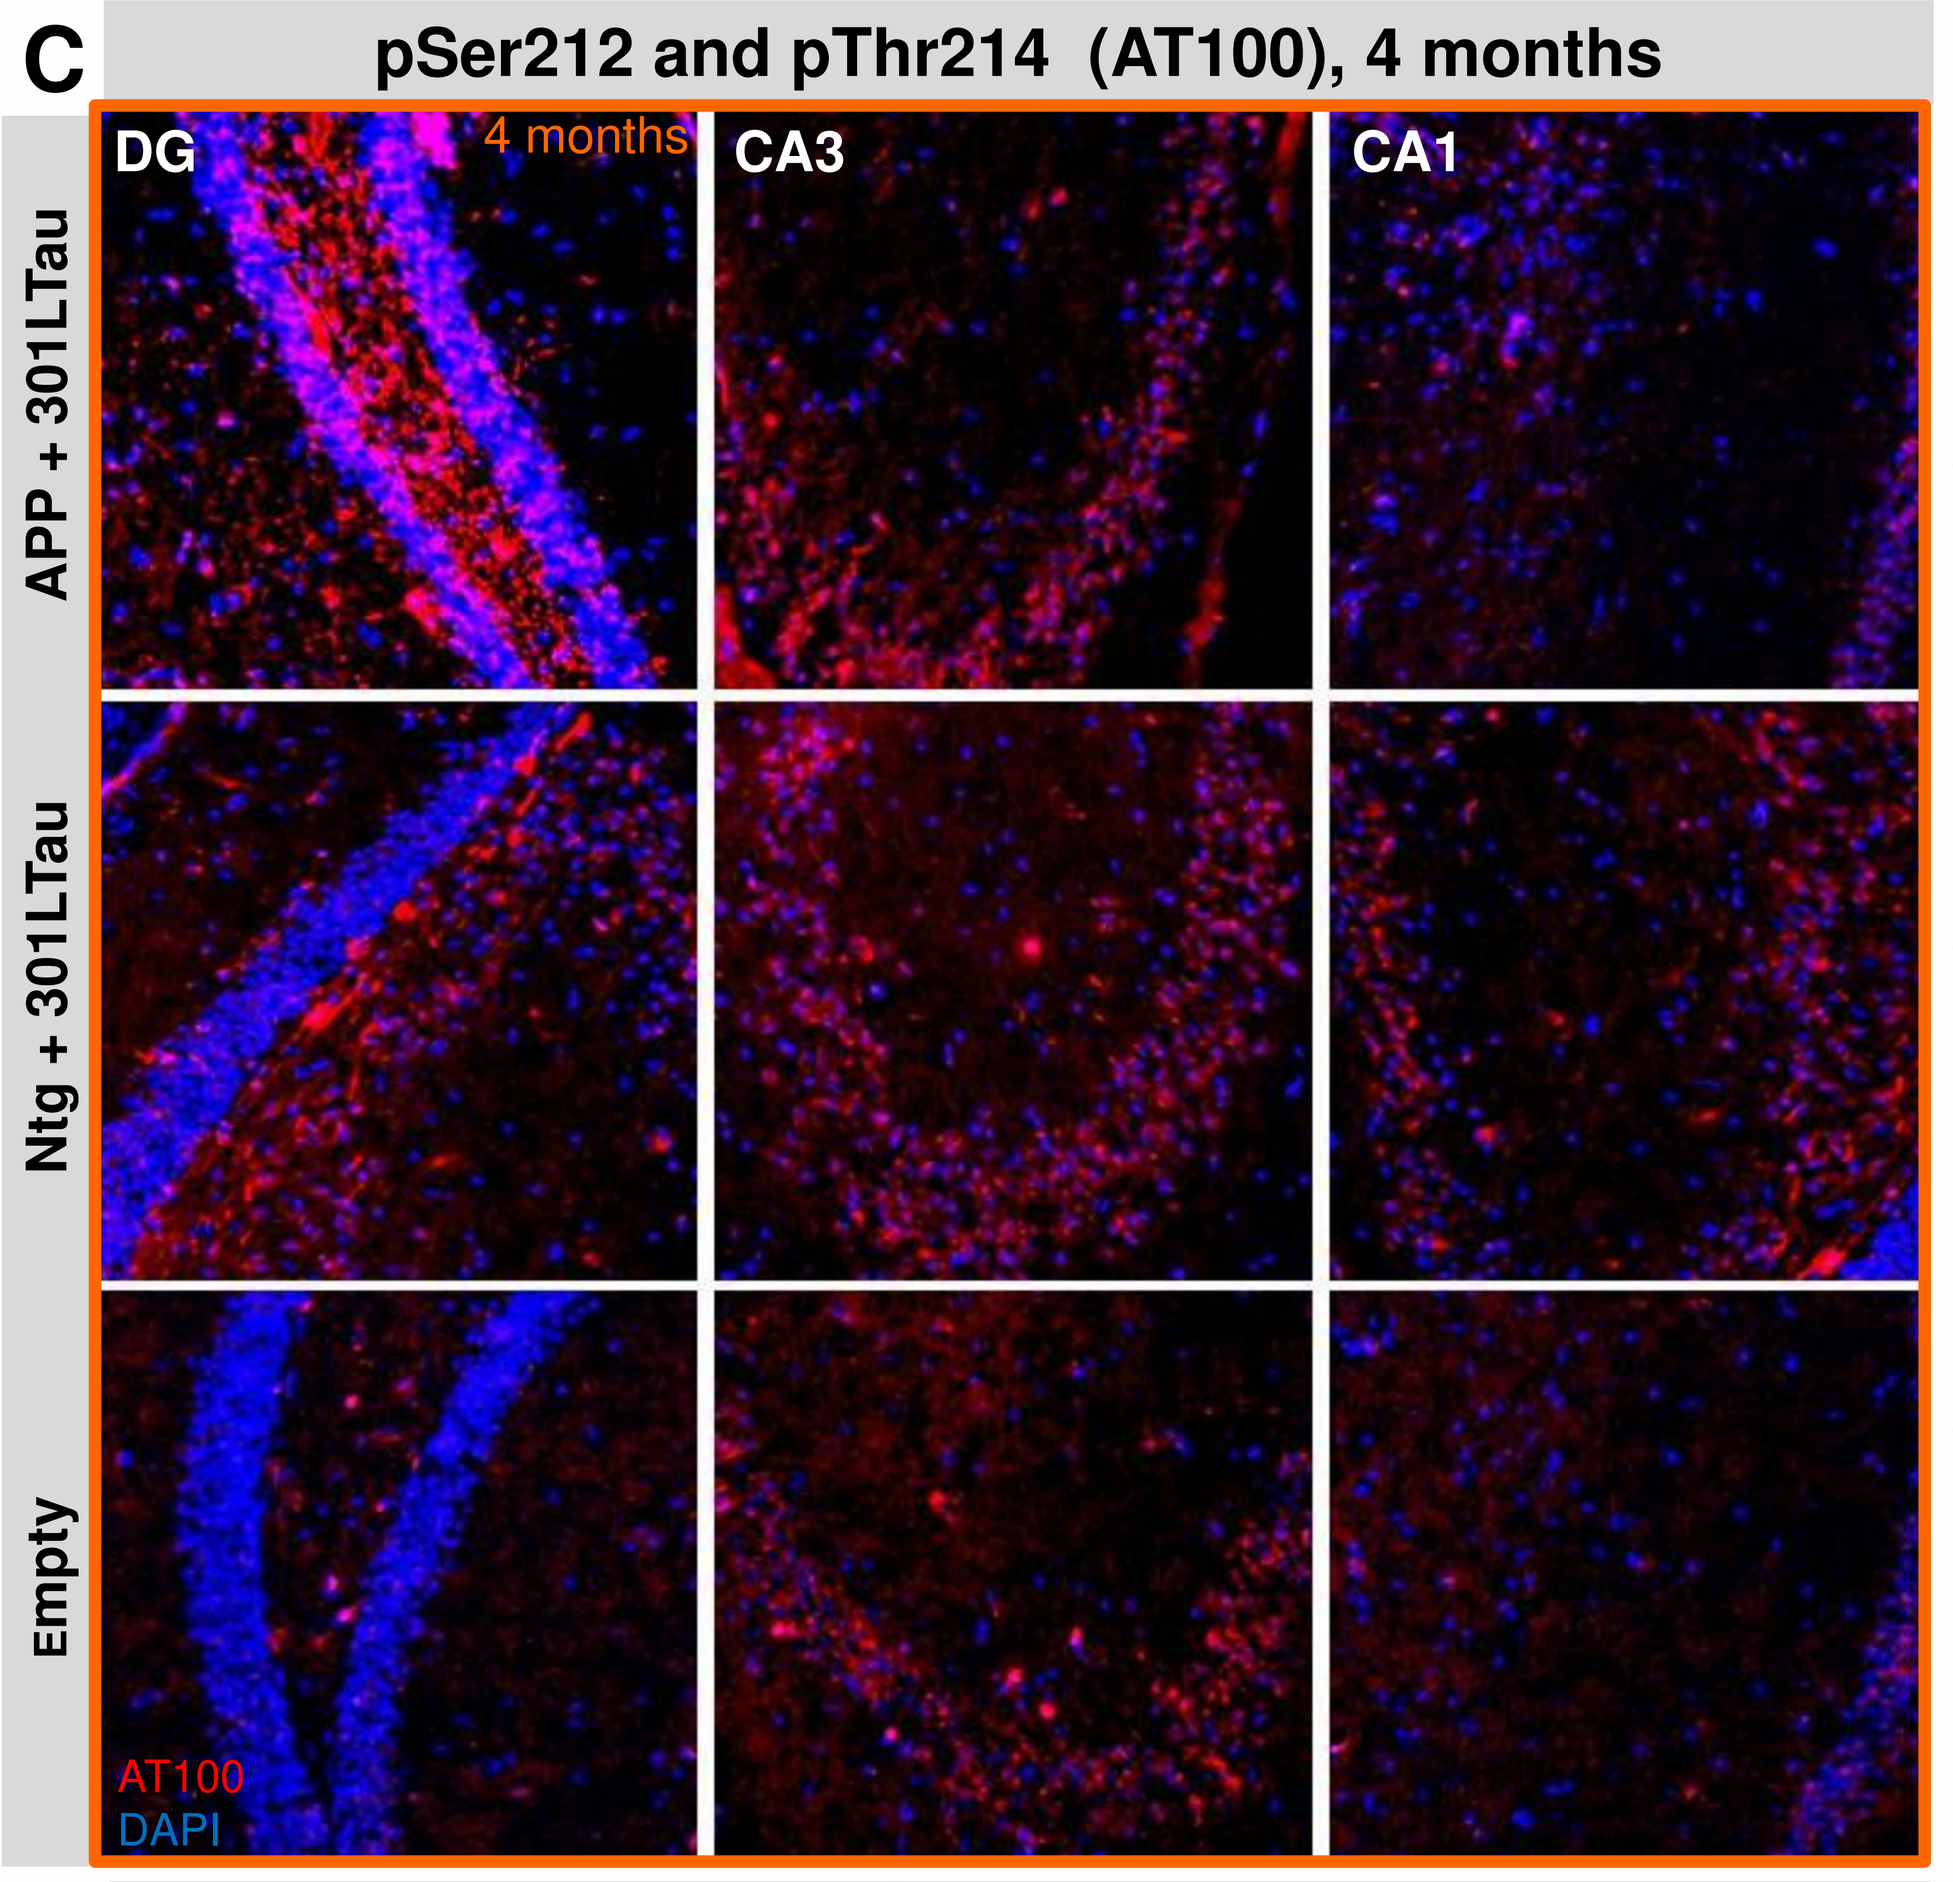
C
pSer212 and pThr214 (AT100), 4 months
DG
4 months
CA3
CA1
APP + 301LTau
Ntg + 301LTau
Empty
AT100
DAPI

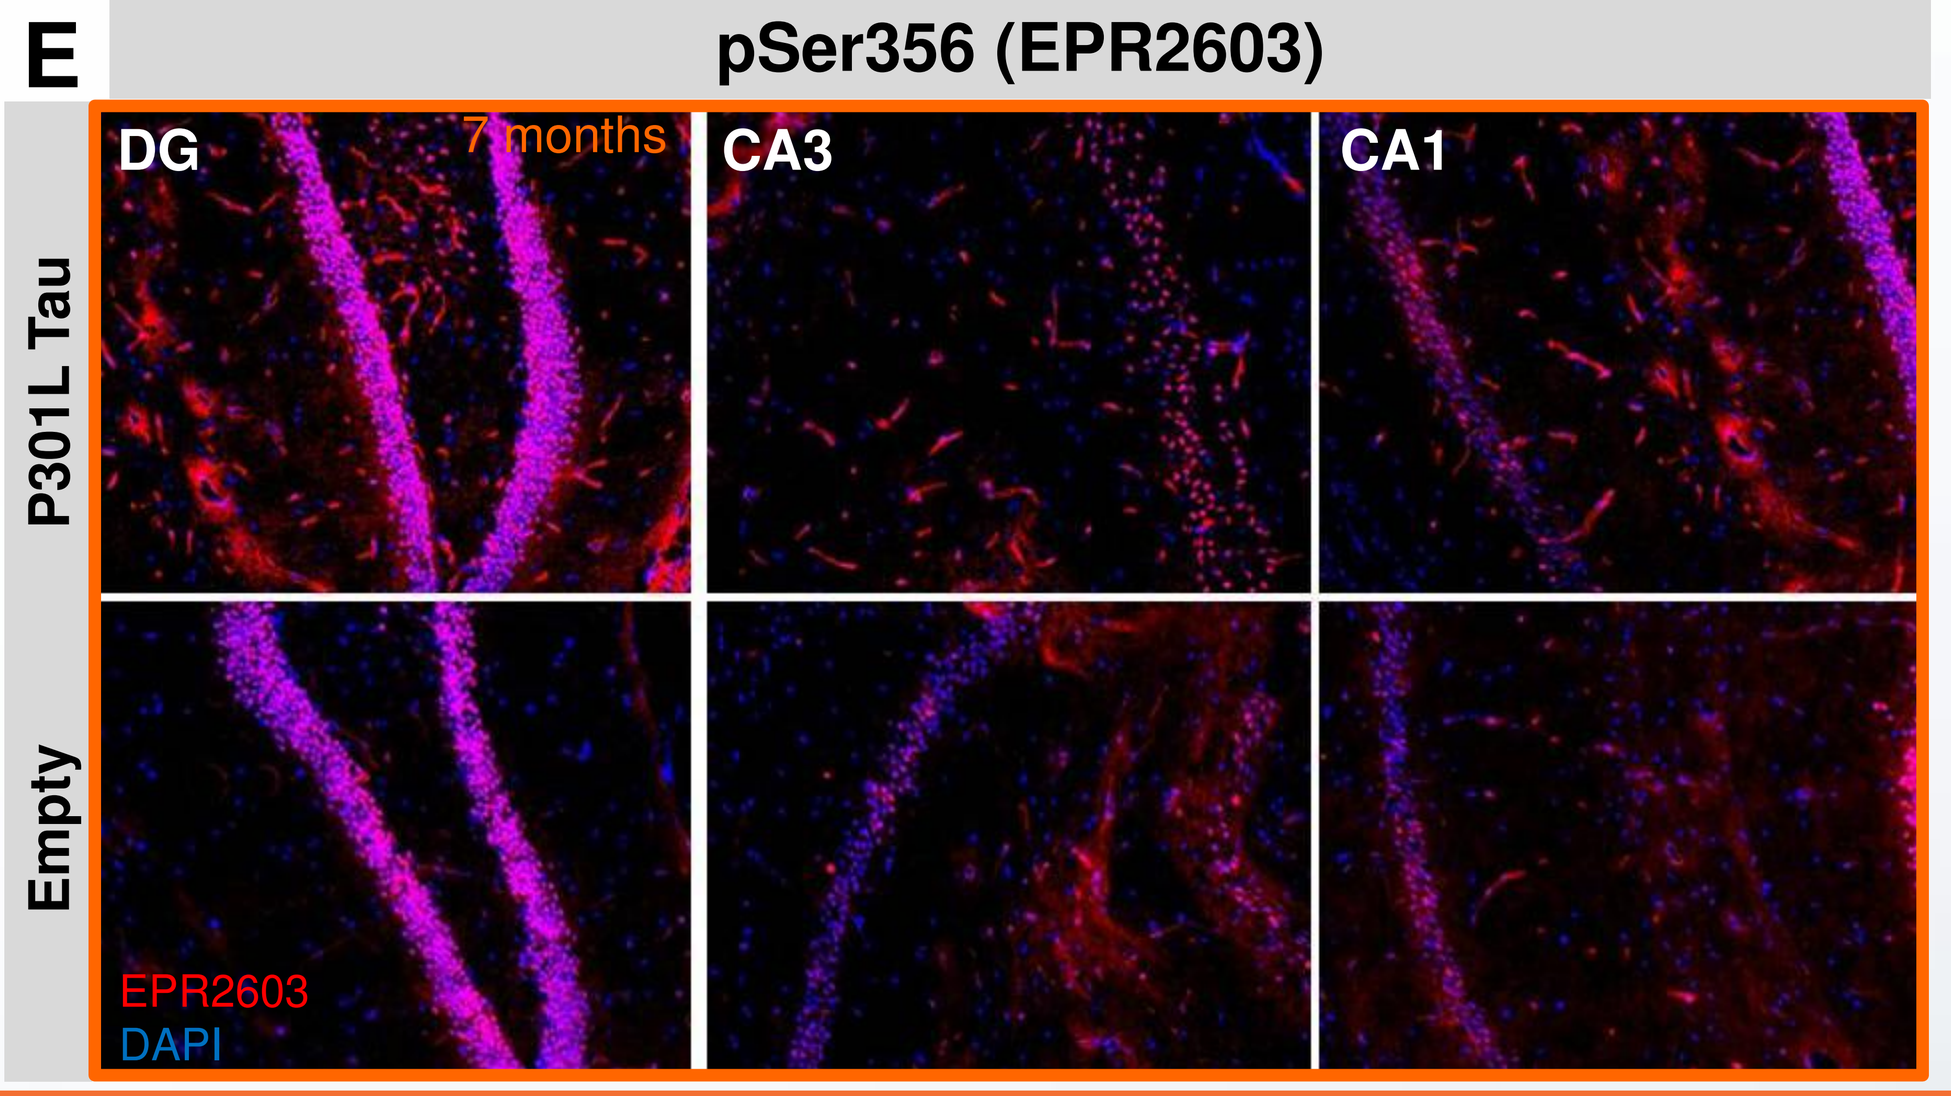
E
pSer356 (EPR2603)
DG
7 months
CA3
CA1
P301L Tau
Empty
EPR2603
DAPI

Figure 3. Virally expressed Tau shows pathological phosphorylation at AD-related residues.
Phosphorylation of several residues was investigated at different time points post injection. Results confirm reports postulating increased phosphorylation nearby determined physiological residues. PHF-13, one of these cases, is presented in A, where hyper-phosphorylation of this residue is well noticed when P301L Tau is induced.
Epitopes detected by antibodies pSer262, AT100, AT180 and EPR2603 are phosphorylated motifs frequently found in AD brain but not in healthy individuals *(Wang (2008).Progr. in Neurobiology)*

As shown in B, C, D and E, these anomalous phosphorylated Tau is shown as fibrillary-like structures not present in empty vector controls. The most affected region in all cases was the DG close to the injection site, suggesting that pathological phosphorylation at AD-related residues may be promoted by high P301L Tau concentration.

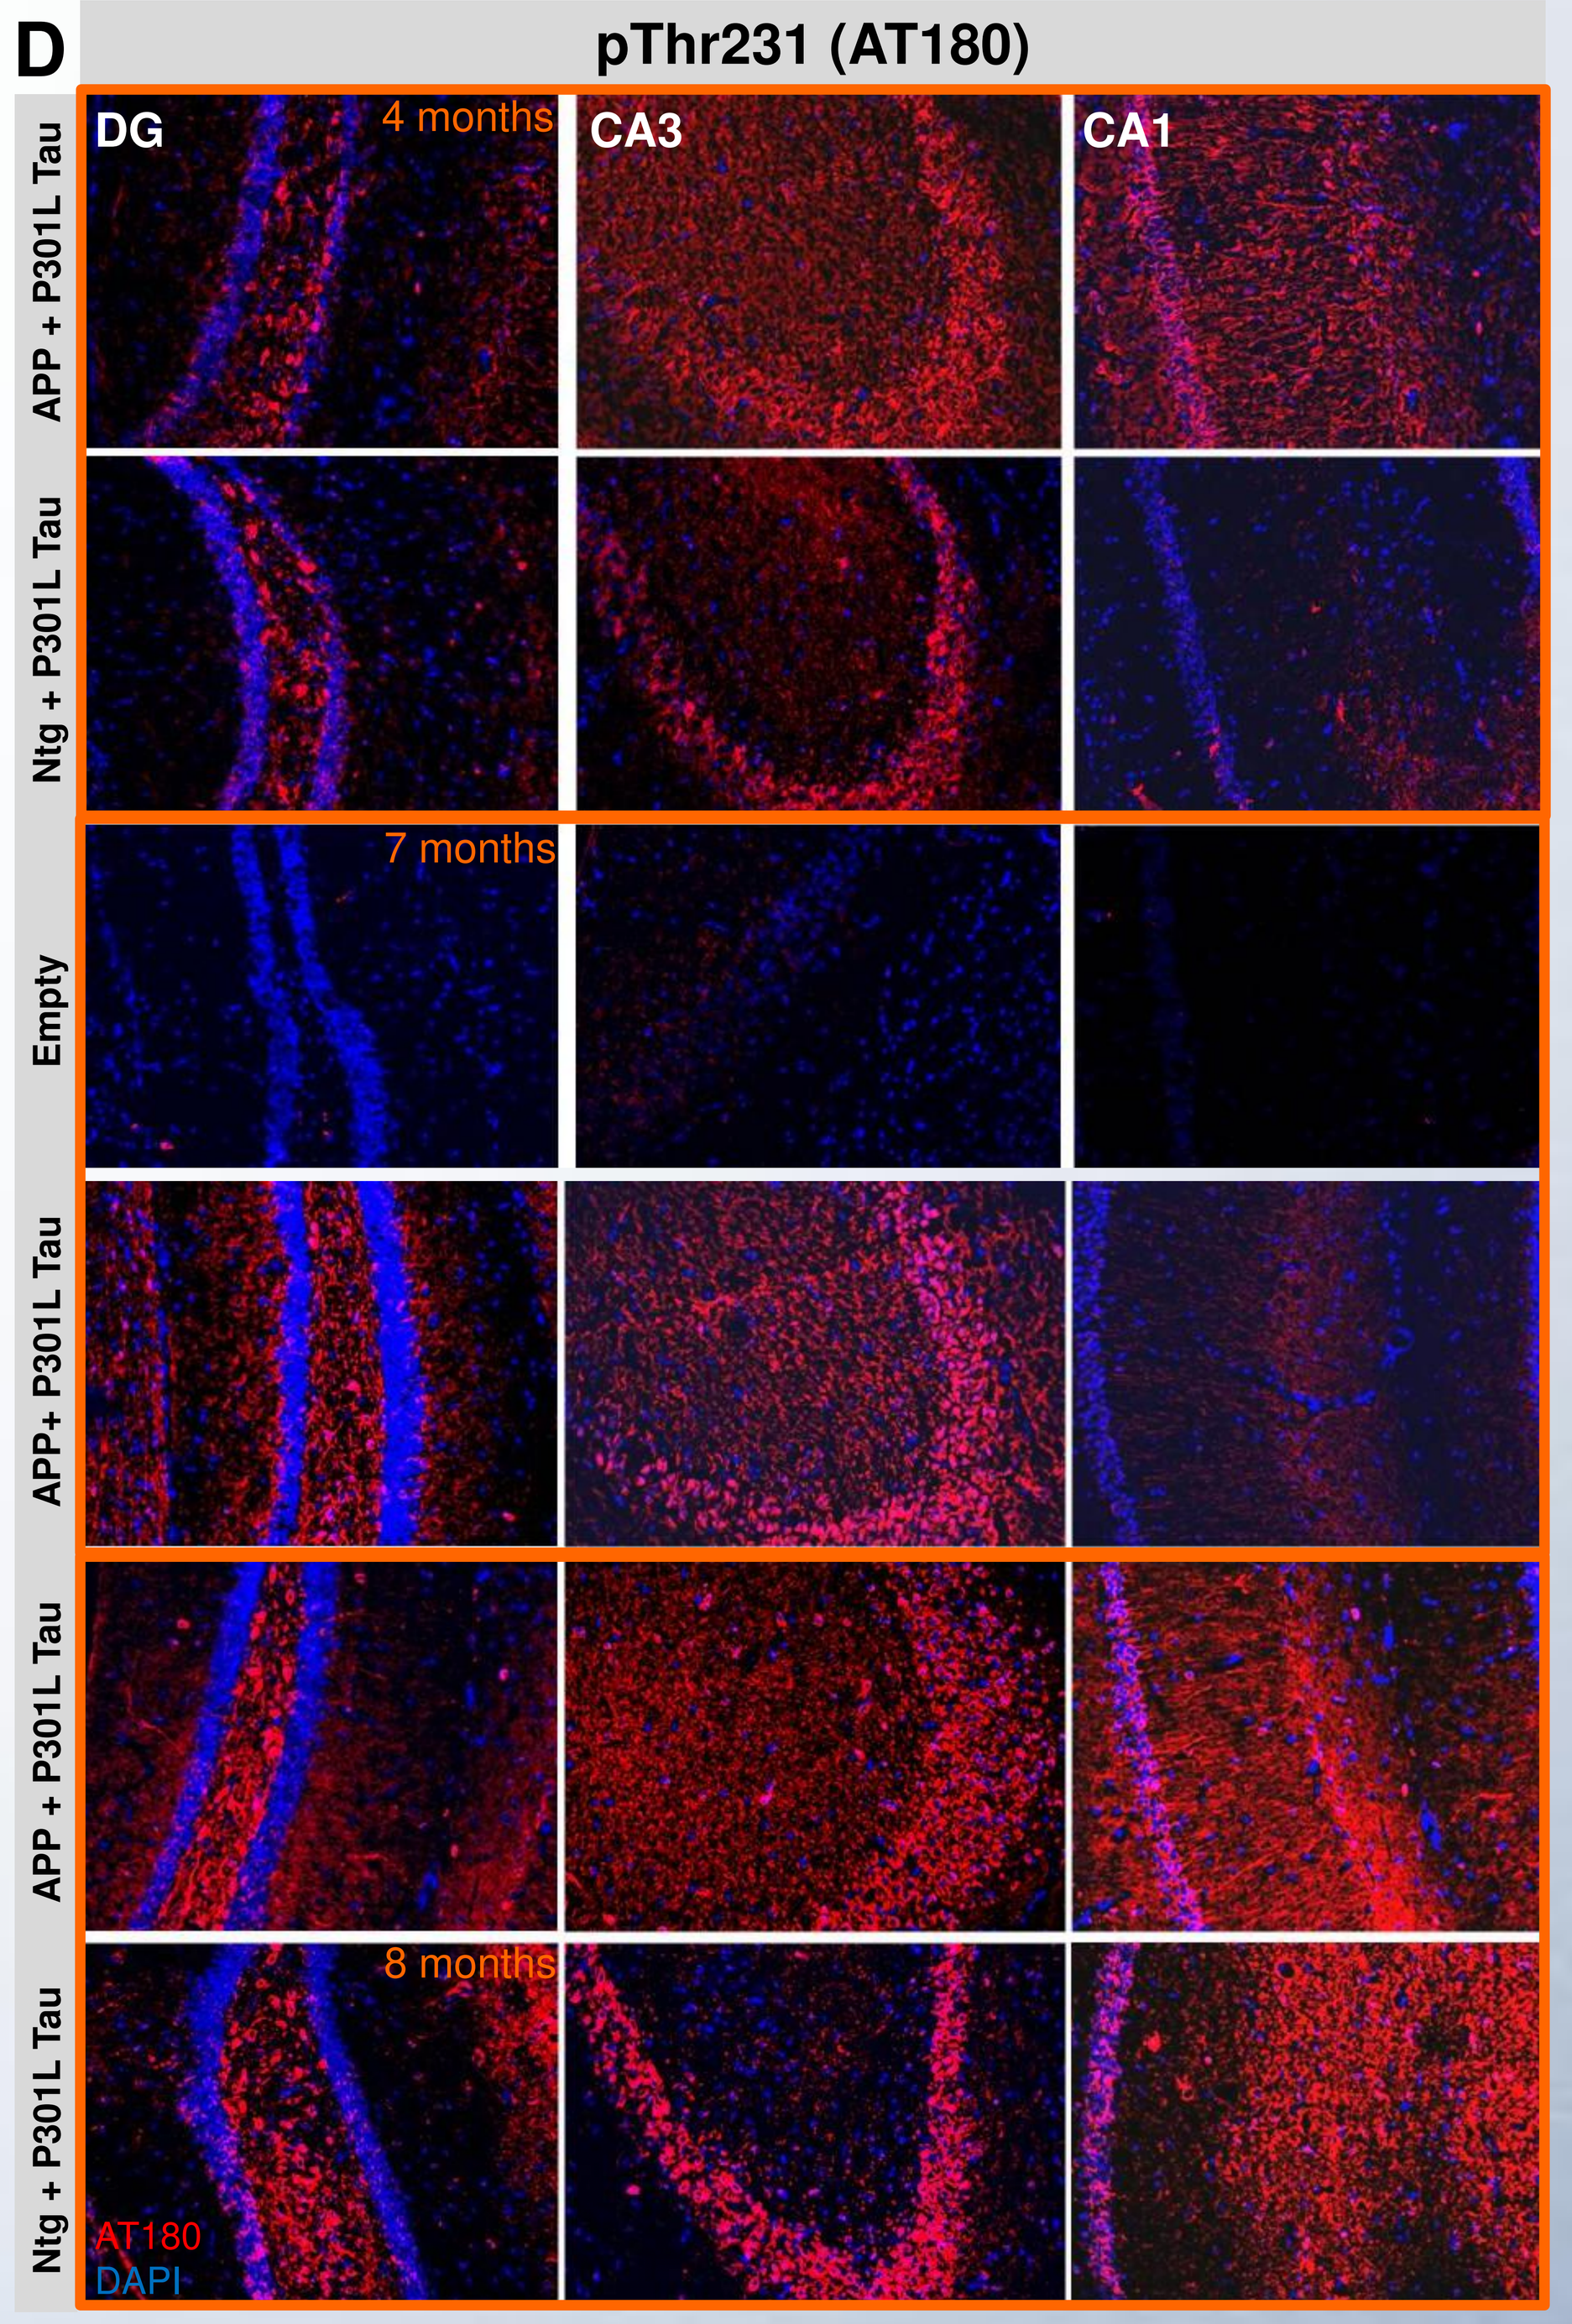
D
pThr231 (AT180)
DG
4 months
CA3
CA1
APP + P301L Tau
Ntg + P301L Tau
7 months
Empty
APP+ P301L Tau
APP + P301L Tau
8 months
Ntg + P301L Tau
AT180
DAPI

CONCLUSION

High levels of virally induced P301L Tau are present throughout the entire hippocampus of APP_{SL} mice. Tau expression is stable for at least 8 months post injection, providing a long period for drug testing and for investigating interaction of Aβ and Tau. Of note, phosphorylation of several Tau residues typical for human AD has been confirmed in this new mouse model. While analyses are ongoing, we believe these animals constitute a promising new model for drug development and compound testing.

Contact: Birgit Hutter-Paier, PhD | Director Neuropharmacology
QPS Austria GmbH | Parkring 12 | 8074 Grambach | Austria
birgit.hutter-paier@qps.com | www.qpsneuro.com

AAIC 2016, P4-13-265; Poster. #10745