ANALYSIS OF NEURODEGENERATIVE DISEASE RELATED NEUROPATHOLOGY IN RETINA AND OLFACTORY BULB OF RODENT MODELS

Jörg Neddens*, Ainara Lopez*, Vera Niederkofler, Stefanie Flunkert and Birgit Hutter-Paier

*contributed equally

QPS Neuropharmacology, Parkring 12, 8074 Grambach, Austria

QPS helps you navigate

BACKGROUND

Alzheimer´s disease (AD) is the most common form of neurodegenerative dementia. Major hallmarks of the disease are: (1) extracellular plaque deposits of the β-amyloid peptide (Aβ) and (2) intracellular neurofibrillary tangles of hyperphosphorylated tau. Published research suggests an association between AD and functional impairments of sensory systems. In fact, the occurrence of Tau-mediated glaucoma has been recently reported, as well as AD protein-associated neuropathology in sensory systems (M. Chiasseu et al., 2016). The current study is designed to analyze the neuropathological changes occurring both in brain and retina of different AD animal models and address suitable biomarkers for early screening tests for AD.

MATERIALS AND METHODS

Eyes and brains from the rodent AD models TAU mice (Tau 441/V337M; R406W-Thy1), APP mice (human APP_{SL}-Thy1), TAU rats (Tau 441/P301L-Thy1) and APP rats (APP_{SL}-Thy1), aged 6 and 12 months, were collected. Tissue was cryosectioned and histologically investigated by immunofluorescent microscopy (Zeiss AxioImager Z1 microscope) with the focus on: 1) visual system (visual cortex and retina) and 2) olfactory system (olfactory bulb and piriform cortex). Different neuronal and neuropathological markers were evaluated utilizing the following antibodies: APP (6E10, BioLegend), Tau (PHF-6, Covance), GFAP (Abcam), IBA1 (Wako), ChAT (Millipore), GAD67 (1G102, Millipore) and TH (Novusbio) among others.

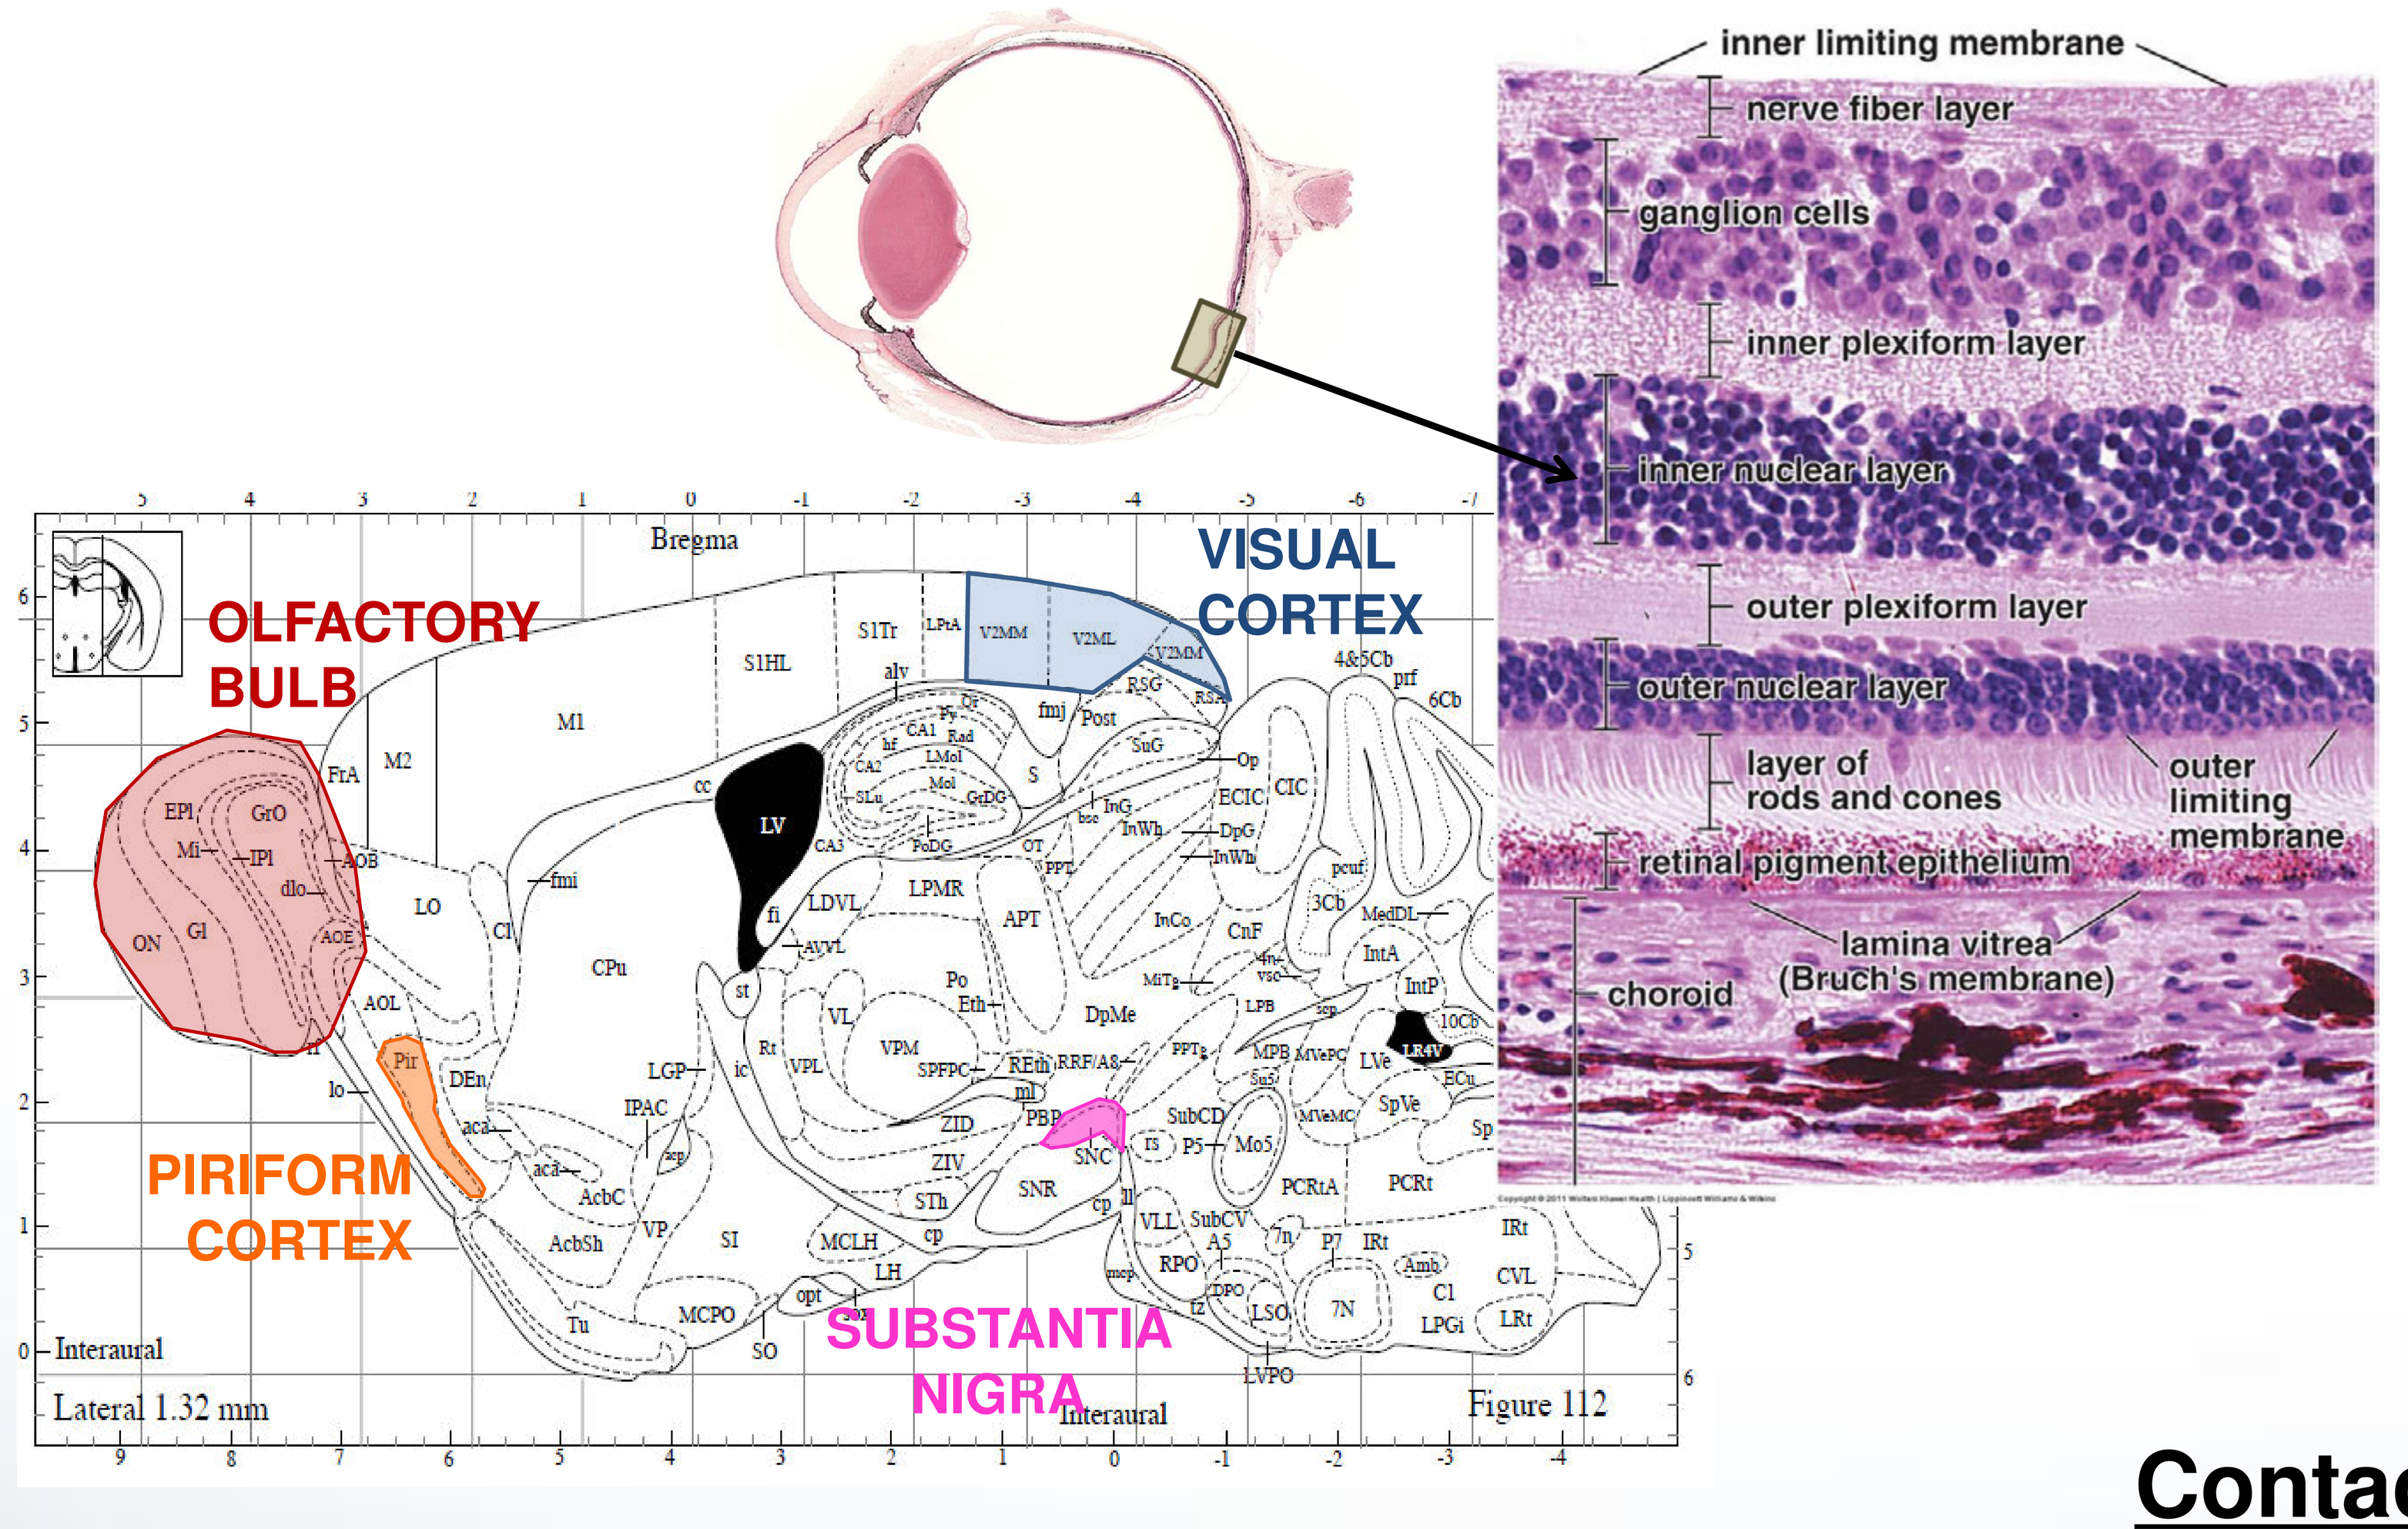

inner limiting membrane
nerve fiber layer
ganglion cells
inner plexiform layer
inner nuclear layer
outer plexiform layer
outer nuclear layer
layer of rods and cones
outer limiting membrane
retinal pigment epithelium
lamina vitrea (Bruch's membrane)
choroid
OLFACTORY BULB
VISUAL CORTEX
PIRIFORM CORTEX
SUBSTANTIA NIGRA
Bregma
Interaural
Lateral 1.32 mm
Figure 112

RESULTS

Microglia and astroglia were detected with antibodies against IBA1 and GFAP, respectively (**Figure 1**). Expression is detectable in all areas of interest and is currently subject to quantitative image analysis.

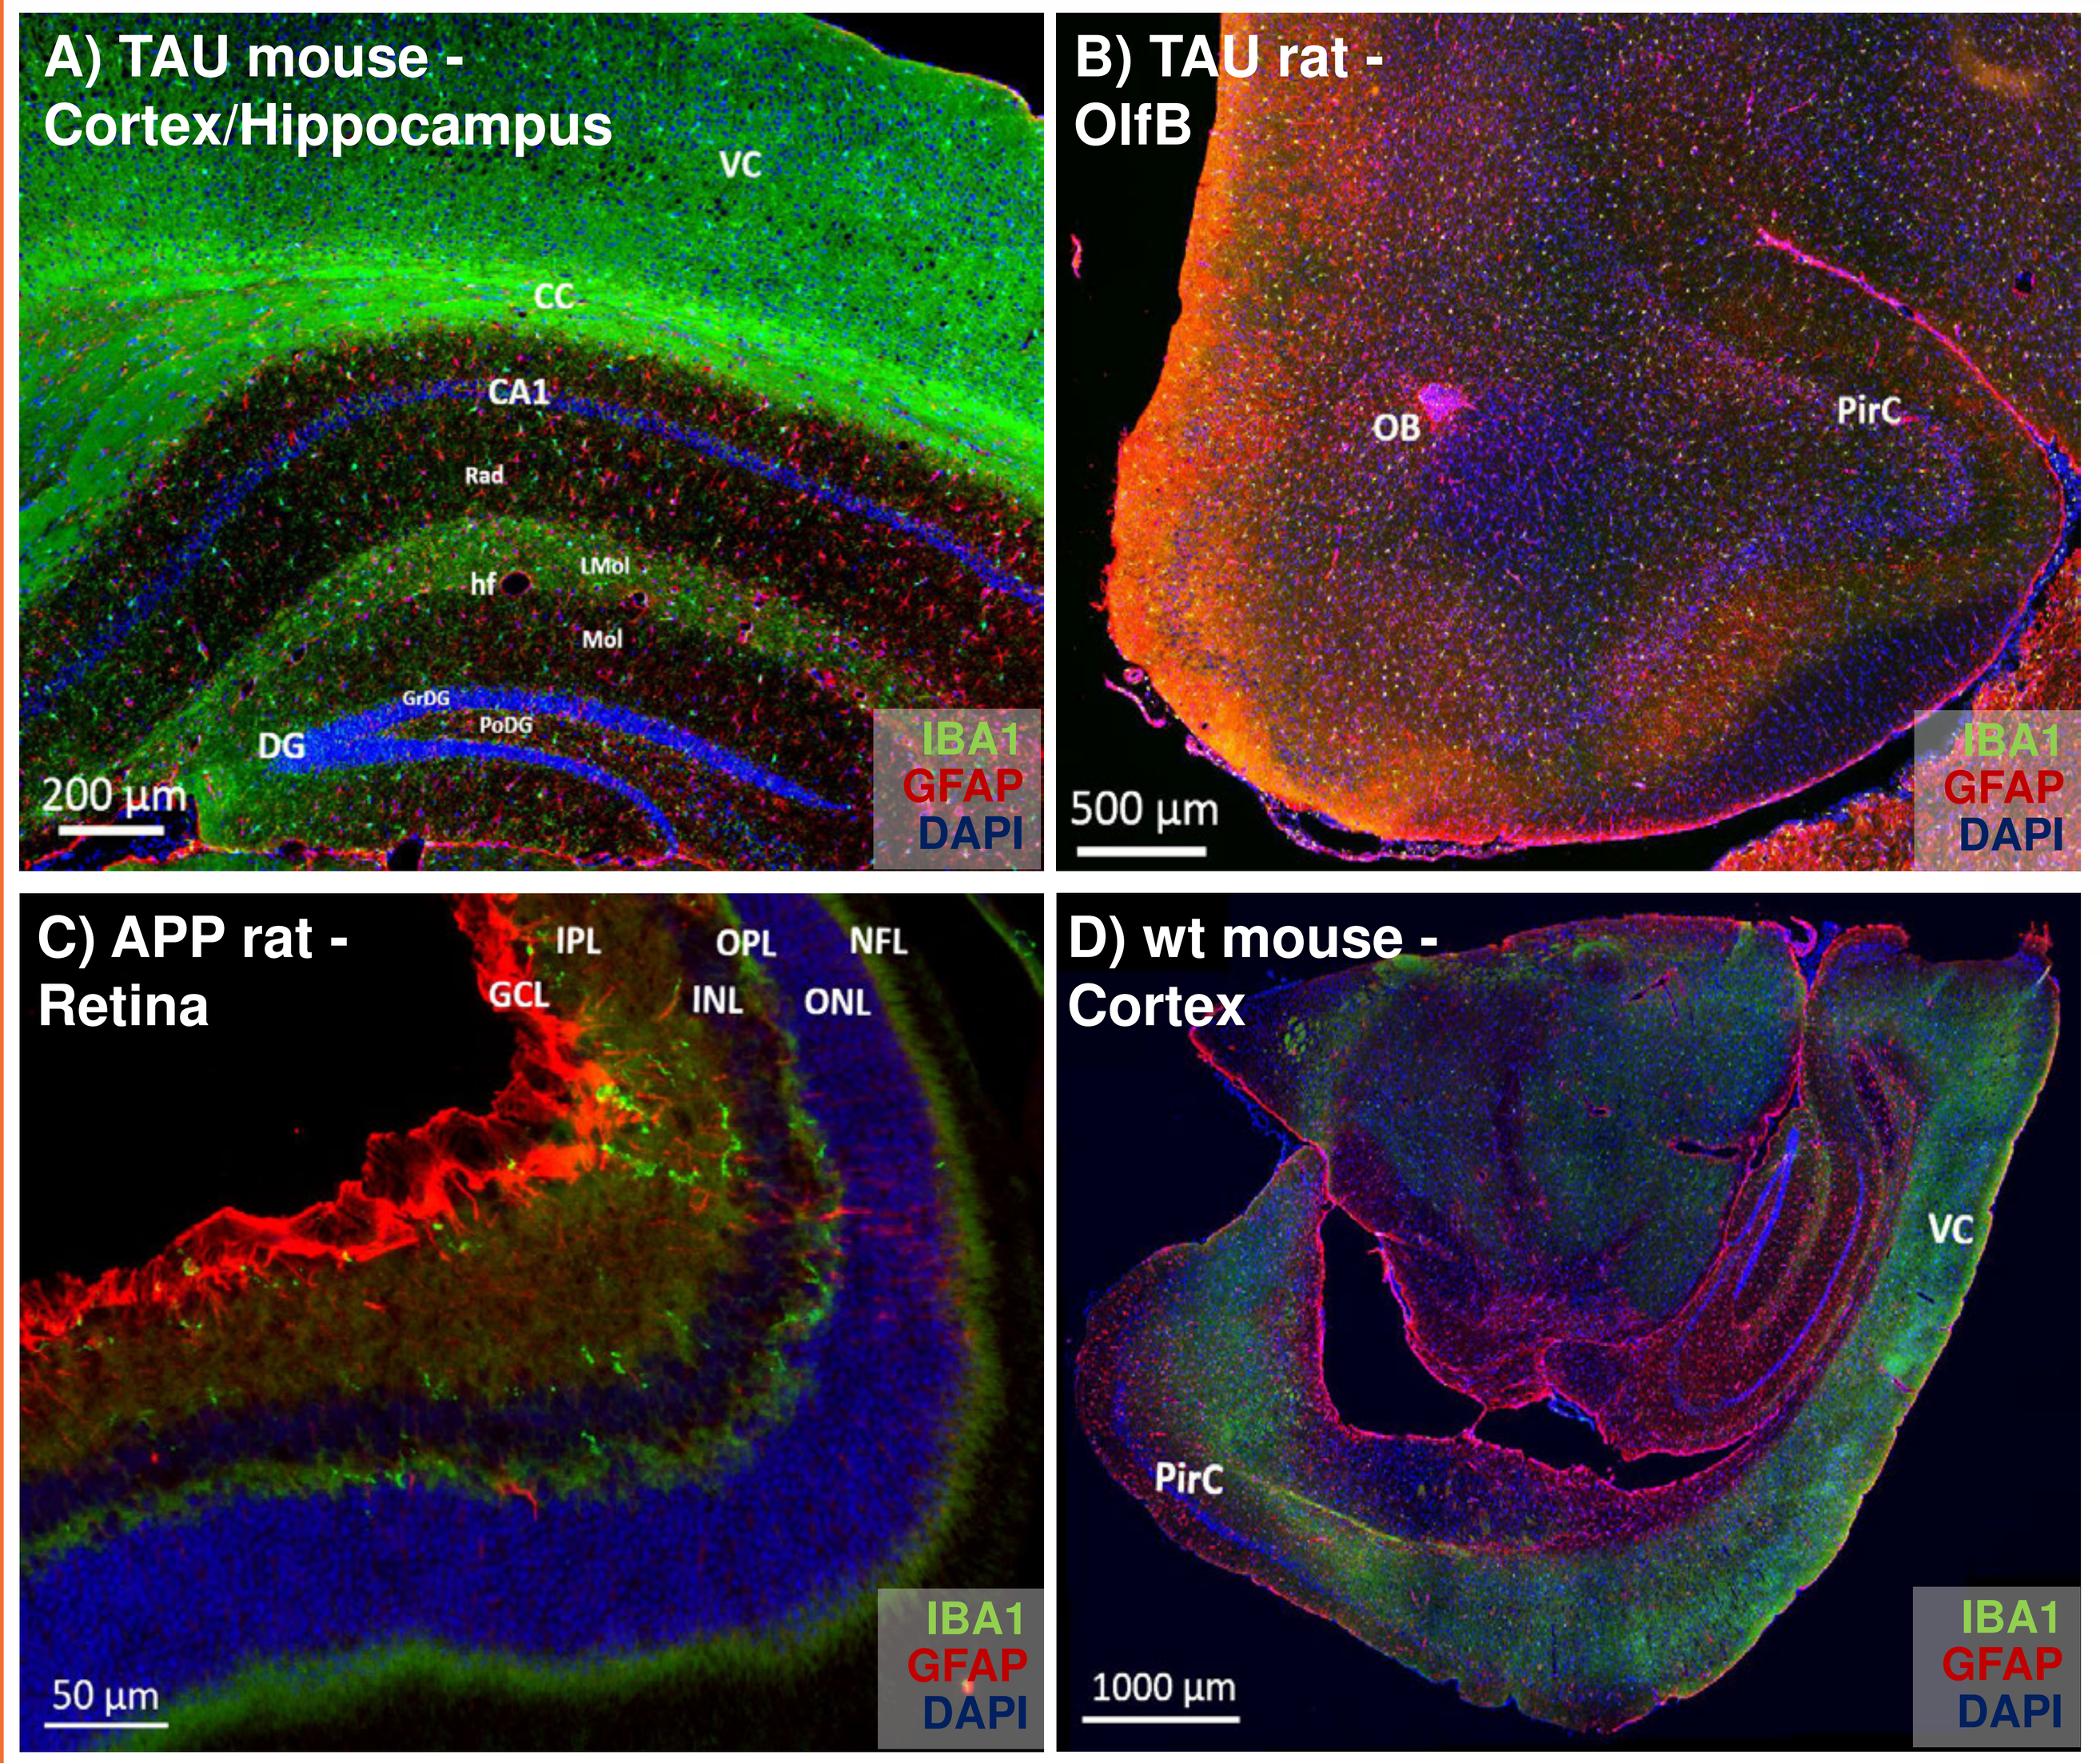

A) TAU mouse - Cortex/Hippocampus
B) TAU rat - OlfB
C) APP rat - Retina
D) wt mouse - Cortex
IBA1
GFAP
DAPI
200 µm
500 µm
50 µm
1000 µm

Figure 1: Gliosis in AD rodent models. Immunofluorescent labelling of IBA1 (green) and GFAP (red) in brain and retina. DAPI (blue) to stain cell nuclei. IBA1+ cells are distributed all over the brain tissue (images **A**, **B** and **D**), although in the retina IBA1 is mainly restricted to nerve fiber layer (NFL), ganglion cell layer (GCL), and inner and outer plexiform layer (IPL and OPL) (**C**). Regions significantly labelled by GFAP are the hippocampus, piriform area and olfactory bulb (OB) in the brain (**A**, **B** and **D**), whereas in the ocular tissue GFAP immunoreactivity is elevated in GCL (**C**).

Immunohistological examination confirms that pTau inclusions can be detected in the brain of TAU rat models (**Figure 2A**), as previously reported in TAU mice. In APP rodents, amyloid deposition is very low since no plaques are detected in the retina (data not shown) while in the brain plaques are rare and occur randomly (**Figures 2E and 2F**).

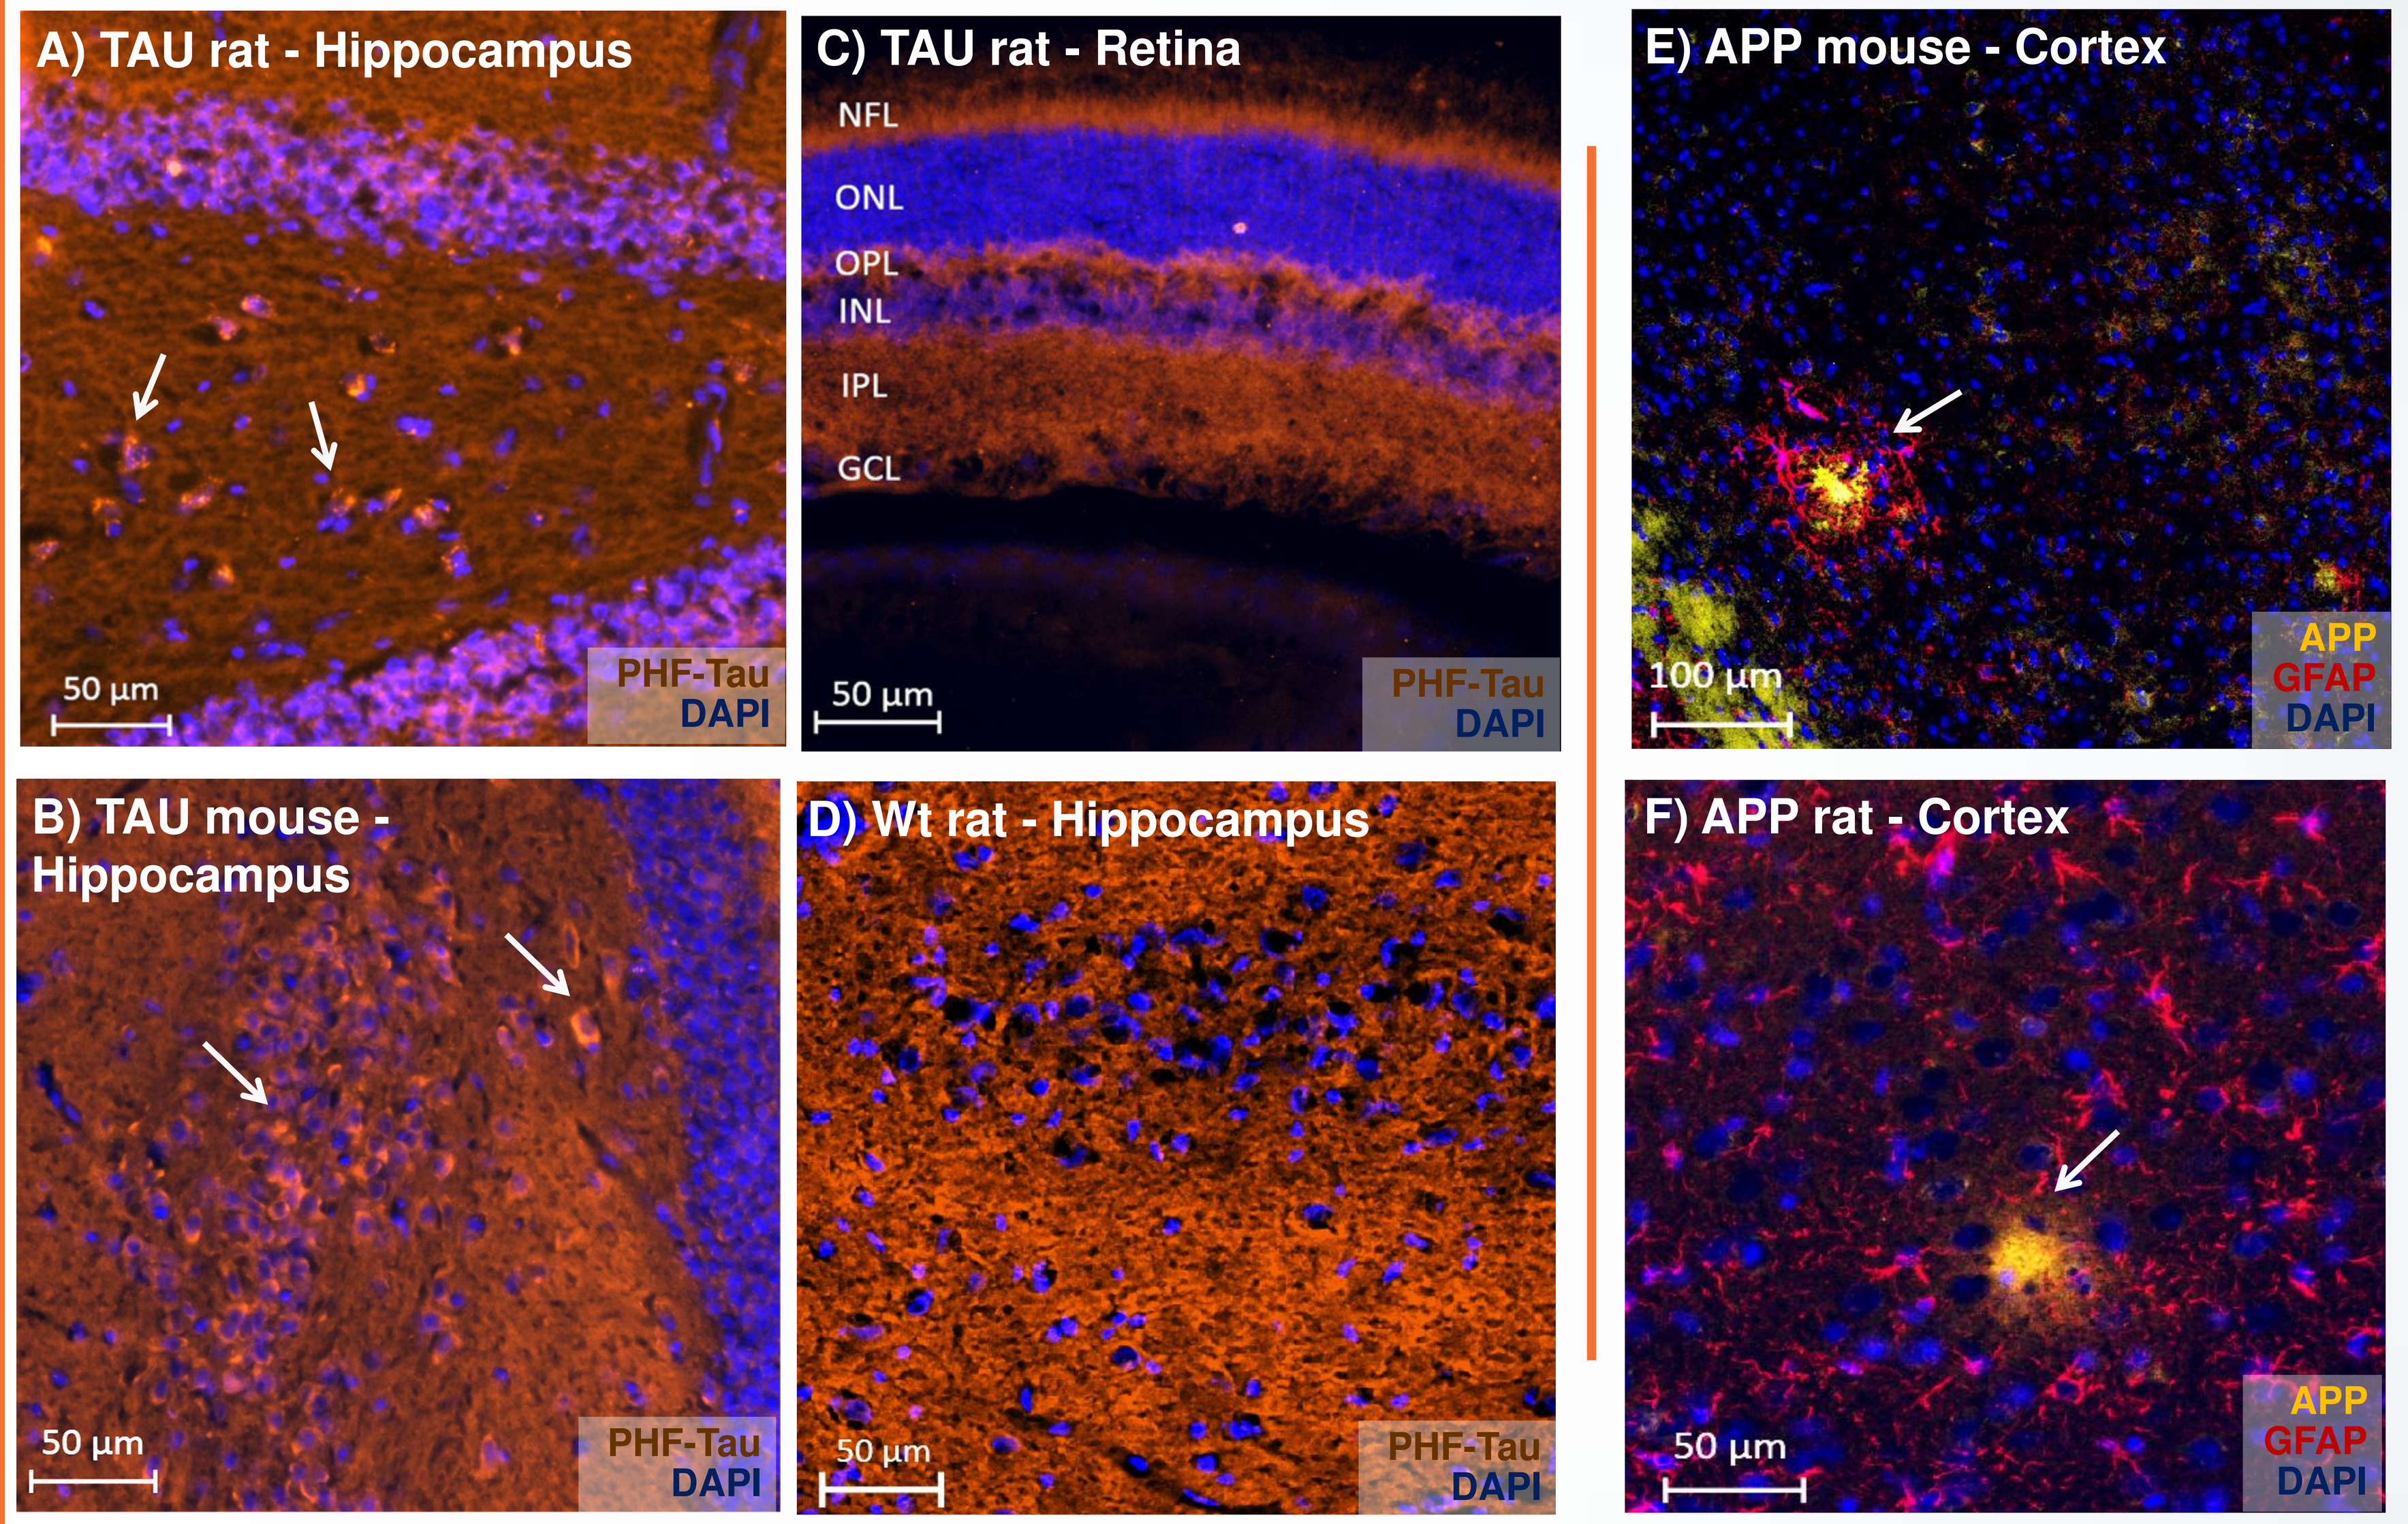

A) TAU rat - Hippocampus
B) TAU mouse - Hippocampus
C) TAU rat - Retina
D) Wt rat - Hippocampus
E) APP mouse - Cortex
F) APP rat - Cortex
NFL
ONL
OPL
INL
IPL
GCL
PHF-Tau
DAPI
APP
GFAP
50 µm
100 µm

Figure 2: Tau and ß-amyloid deposition in AD models. Images **A**, **B** **C** and **D** show PHF-Tau (brown). DAPI (blue) to stain cell nuclei. In the brain (**A**, **B** and **D**) tau is distributed all over the hippocampus, whereas in the retina (**C**) p-tau is principally detected in IPL, OPL and NFL. In TAU rats aggregation of Tau leads to the formation of tangles in the hippocampus (white arrows in **A** and **B**), but not in the ocular tissue (**C**).

Double immunostaining of APP (yellow) and GFAP (red) shows ß-amyloid plaques (white arrows) in the cortex in APP models (**E** and **F**), but not in the retina (data not shown). Colocalization of GFAP and APP indicates that reactive astrocytes cluster around the plaques in AD rodents (**E** and **F**).

AD has a significant effect on the synthesis and secretion of different neurotransmitters. GABAergic, cholinergic and catecholaminergic neurons were labelled (**Figure 3**) and analysis of neurotransmitter systems is currently executed. Additionally, evaluation of other AD markers to study differences between physiological and pathological features of neurons and synapses has been started.

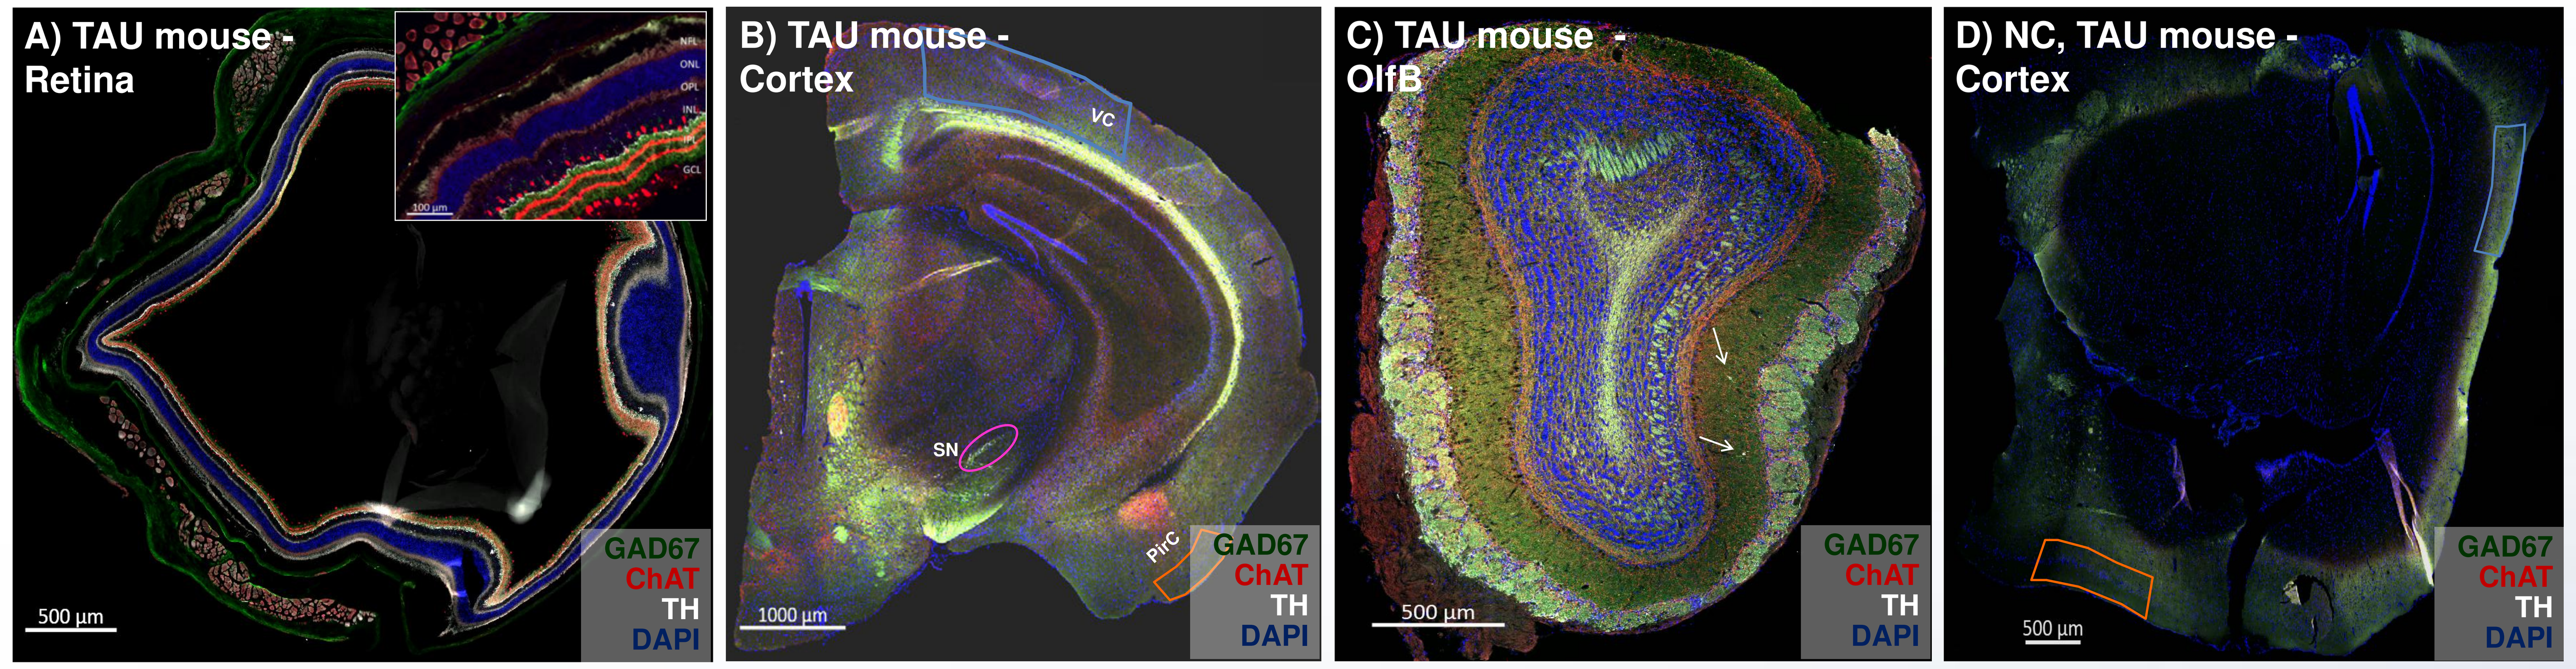

A) TAU mouse - Retina
B) TAU mouse - Cortex
C) TAU mouse - OlfB
D) NC, TAU mouse - Cortex
VC
SN
PirC
GAD67
ChAT
TH
DAPI
500 µm
1000 µm

Figure 3: Detection of neurotransmitters in the brain and retina of TAU mice. Co-labeling of GAD67 (green), ChAT (red) and TH (white) in TAU mice: retina (**A**), visual cortex (**B**), piriform cortex (**B**), substantia nigra (**B**) and olfactory bulb (**C**). DAPI (blue) to stain cell nuclei. In cerebral tissue, ChAT evidences a high density of cholinergic cell fibers and axon varicosities, whereas in the retina it labels two cell populations, one in IPL and one in GCL whose dentrites form a double banded staining pattern in IPL. GAD67 identified GABAergic neuronal cell bodies in brain tissue (white arrows in image **C**), and immunoreactivity in the retina was restricted to IPL. Peak expression of TH is observed in substantia nigra and inner nuclear layer (INL) and inner plexiform layer (IPL) of the eye.

SUMMARY

AD models analysed in this study, together with the specific antibodies tested, provide us with a powerful tool to analyse neuropathology in sensory systems. In fact, by means of triple immunofluorescent labellings performed in our target tissues, it is possible 1) to detect whether gliosis occurs in AD-affected tissues, 2) detect tau accumulation, 3) identify ß-amyloid plaques, and 4) evaluate if there is a variation in the level of neurotransmitters. Ongoing research will now focus on the quantitative analysis of the images obtained. Lastly, further biochemical investigation of the tissues will be executed to receive a further characterization of neuropathology in the sensory system.

<u>Contact:</u> Birgit Hutter-Paier, PhD I Director Neuropharmacology

QPS Austria GmbH I Parkring 12 I 8074 Grambach I Austria

birgit.hutter-paier@qps.com I www.qpsneuro.com

Meet QPS at AAIC 2017

Booth #217

AAIC 2017, P2-03-095, Poster: #17070