Concomitant Expression of Induced AAV-P301L Tau and Transgenic β-Amyloid in a New Mouse Model Closer Mimicking Human Alzheimer's Disease

Meritxell Aguiló[1,2], Balázs Dobrovich[1], Joerg Neddens[1], Stefanie Flunkert[1], Nicole Taub[1], Robert Zimmermann[2], Birgit Hutter-Paier[1]

[1]QPS Austria GmbH, Parkring 12, 8074 Grambach, Austria [2] Institute of Molecular Biosciences, University of Graz, Graz, Austria

QPS® helps you navigate

BACKGROUND

AD brains are diagnosed under the co-existence of two main pathological hallmarks: (1) Neurofibrillary tangles (NFT) composed of hyperphosphorylated Tau aggregates, and (2) senile plaques comprising of insoluble β-amyloid (Aβ). Mentioned pathological events do not develop in the same brain region; while NFT production starts in the entorhinal (ERC) and transentorhinal cortex, Aβ plaque loading begins in the neocortex. Moreover, the onset of both pathologies is not triggered at the same time. Hence, we developed an inducible mouse model reproducing both spatio-temporal conditions aiming to obtain trustworthy results in AD preclinical trials.

MATERIALS AND METHODS

Recombinant Adeno-Associated Virus serotype 9 (AAV9) with human P301L Tau gene or empty vector was intracerebrally inoculated into the ERC of 3 months old male APP_{SL} mice and non-transgenic (Ntg) littermates.
(Surgery coordinates: AP: -1.8; ML: +-1.4; DV: -1.4).

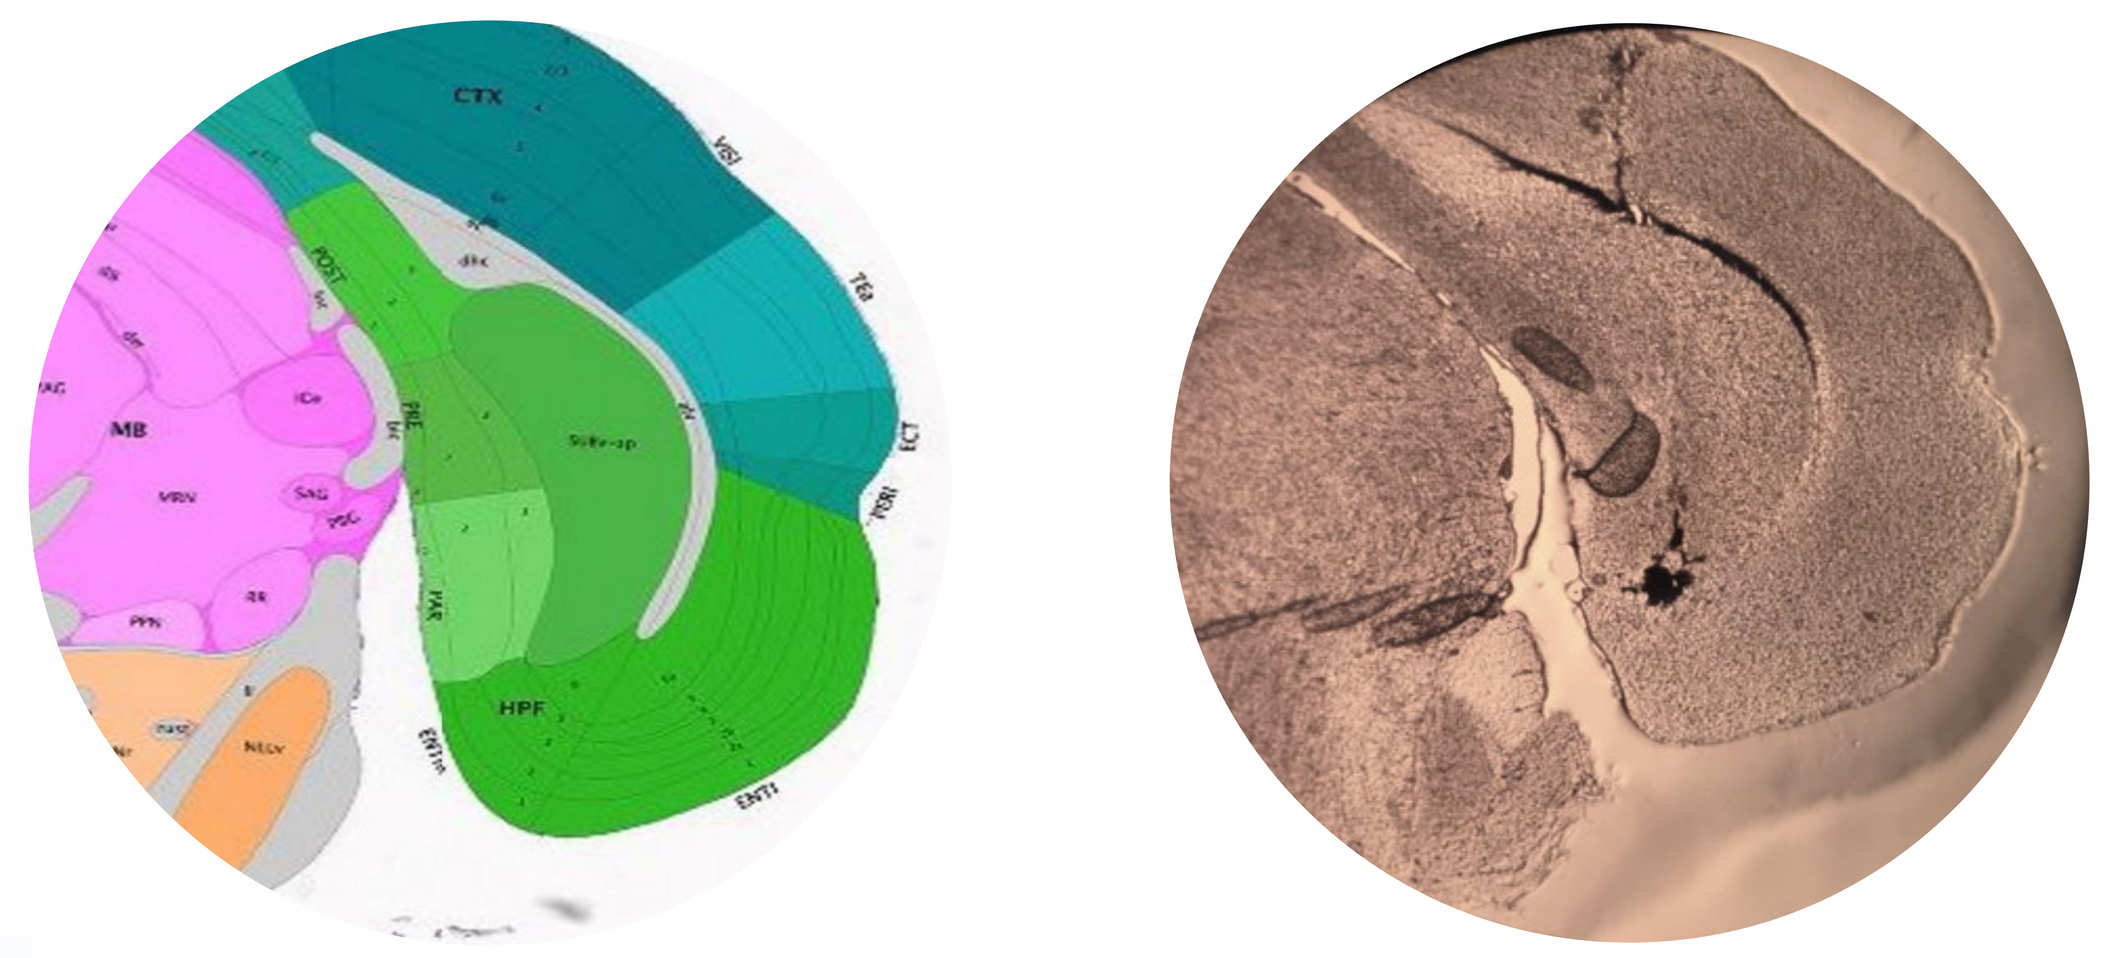

Immunohistological analyses were performed using:

- ❖ Human Tau (Tau 13, Enzo Life Sciences)
- ❖ pThr231Tau (AT180, Thermo Scientific)
- ❖ Aggregated beta amyloid (LOC, abcam)

Meet QPS at AAIC 2017

Booth #217

RESULTS

Stable Expression of Human P301L Tau in ERC

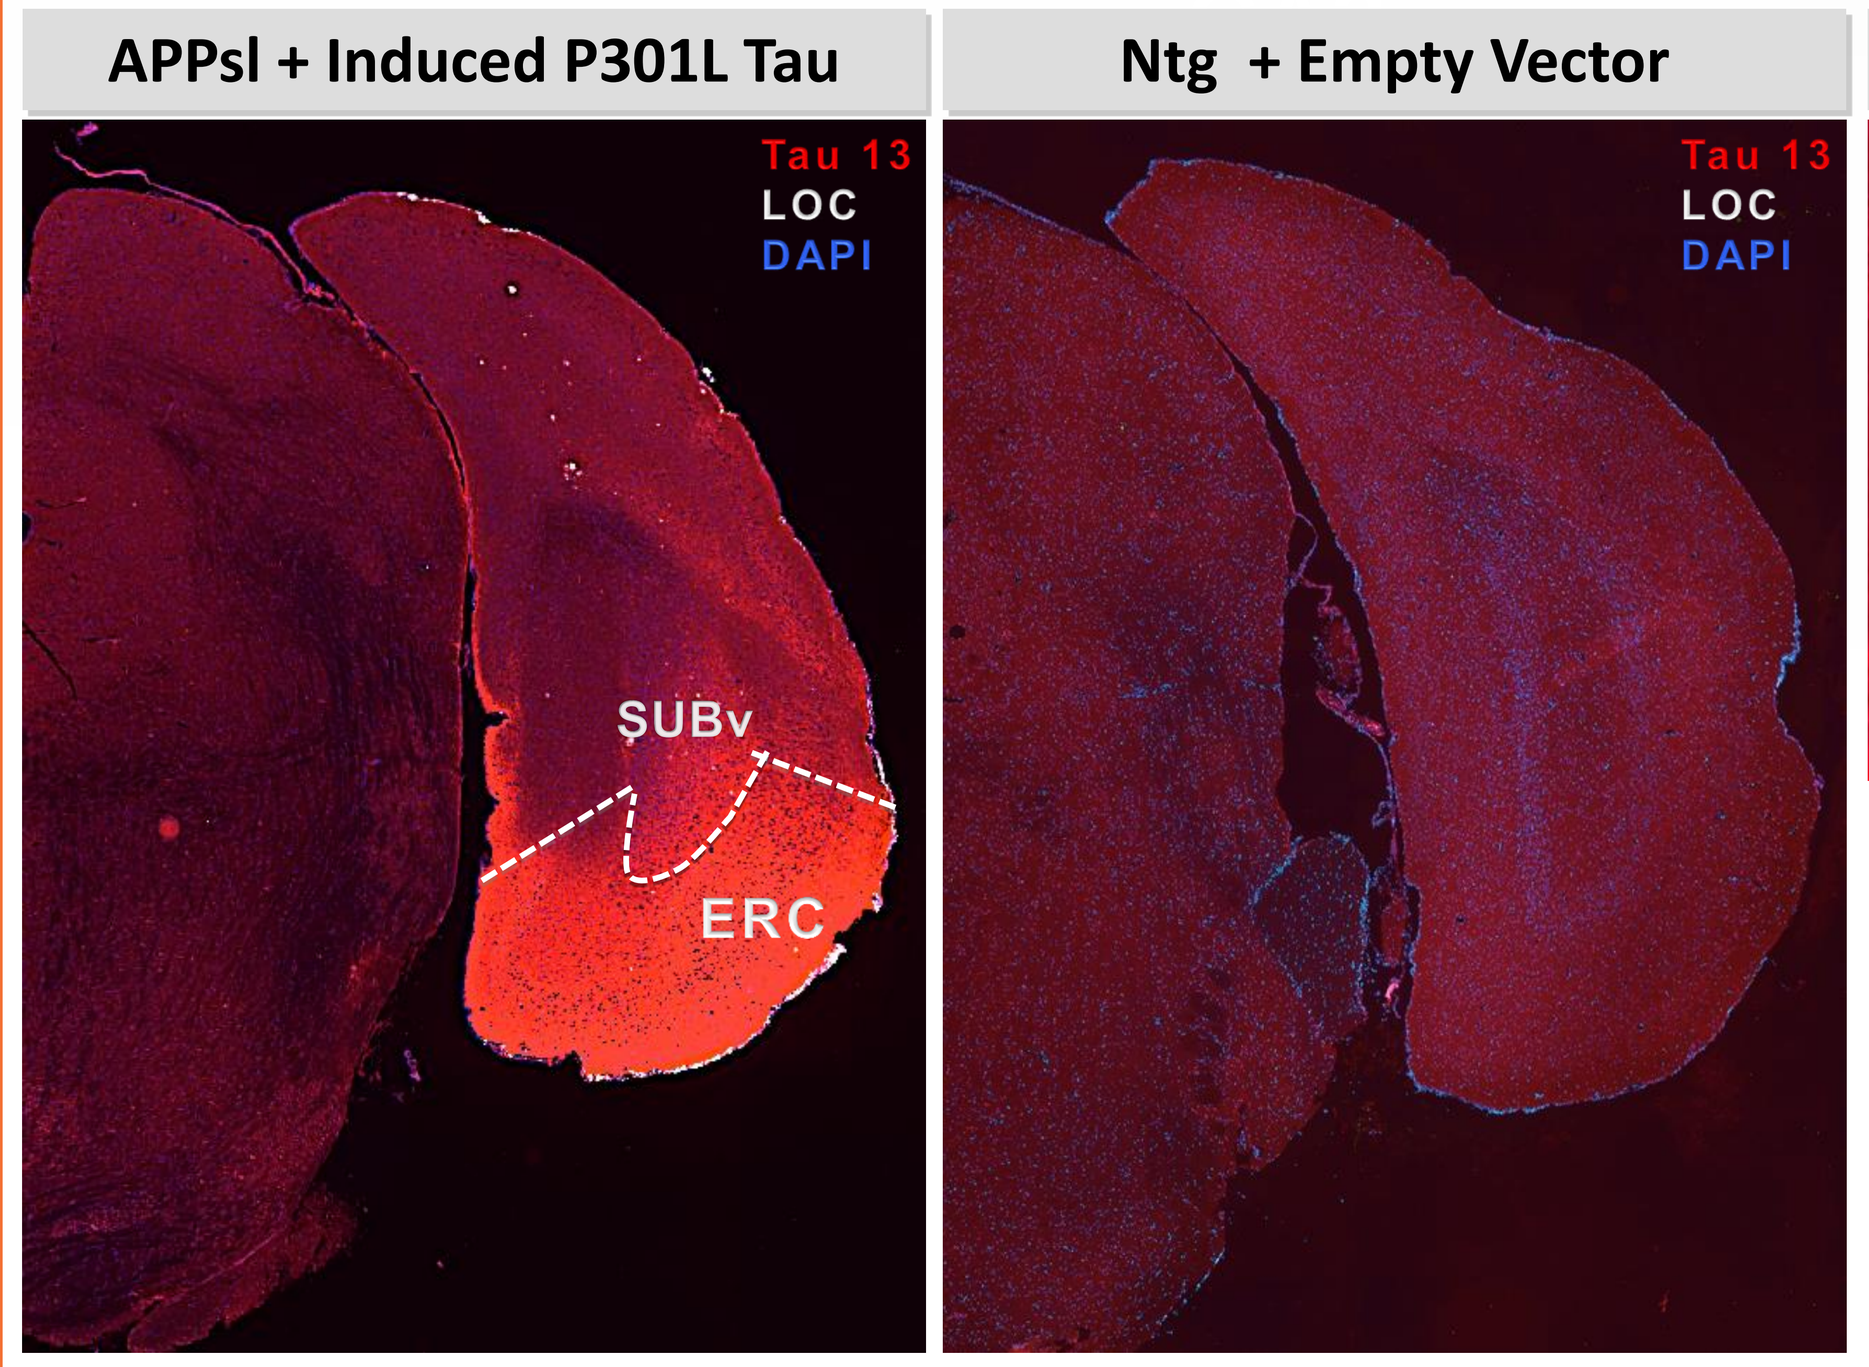
APPsl + Induced P301L Tau
Ntg + Empty Vector
Tau 13
LOC
DAPI
SUBv
ERC

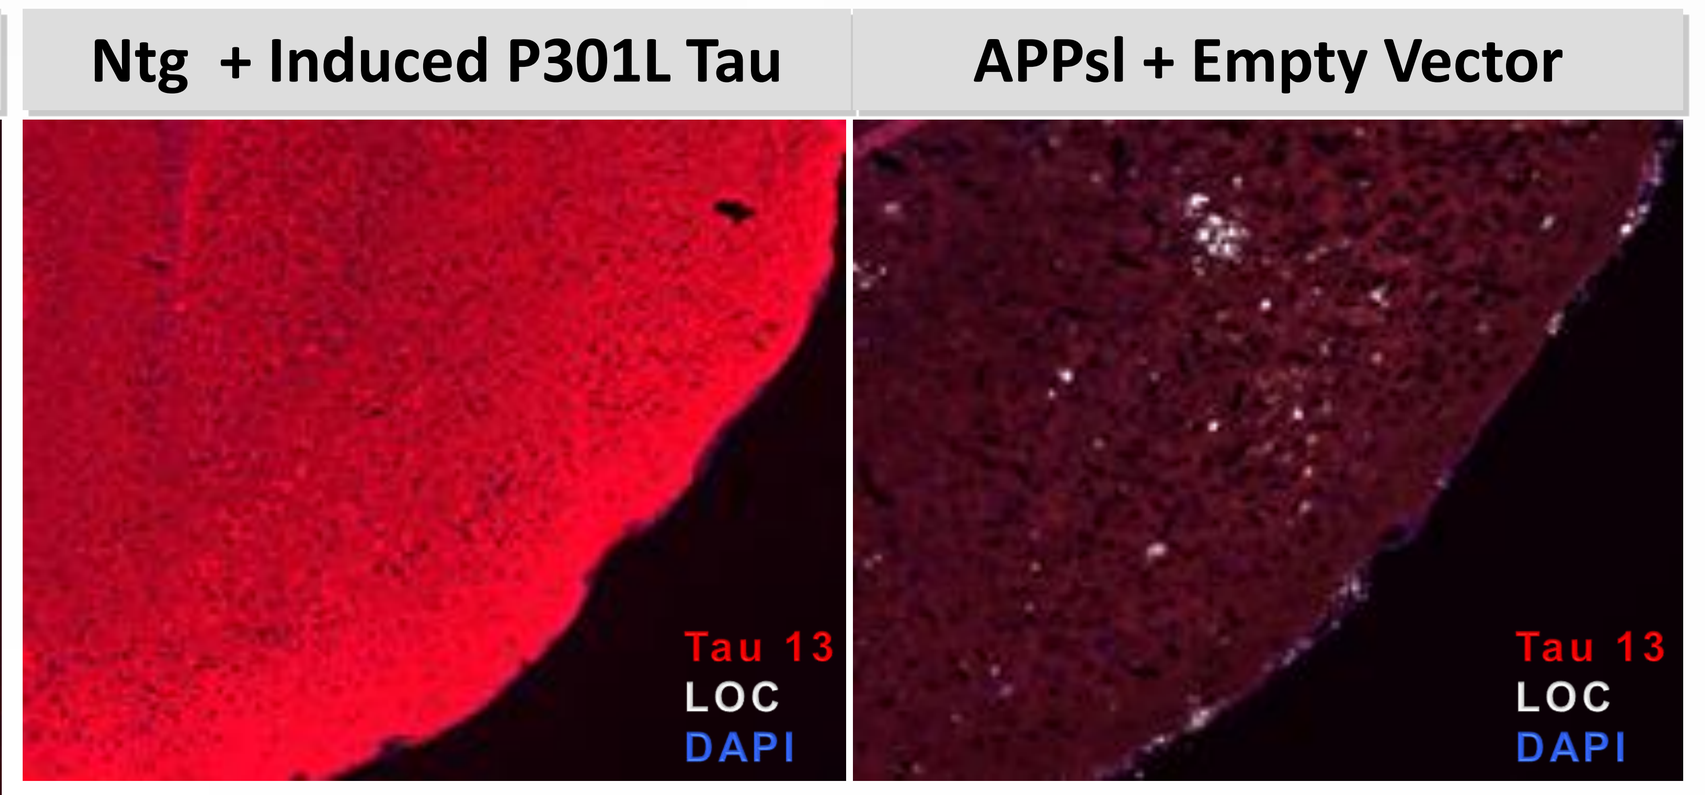
Ntg + Induced P301L Tau
APPsl + Empty Vector
Tau 13
LOC
DAPI

Figure 1. Concomitant occurrence of human P301L Tau and amyloid plaques.
Hemi-brains show intense immunofluorescence of injected human P301L Tau in the ERC **one month after injection**. Expression remained stable at least six months after surgery. In addition to Tau, presence of amyloid plaques is evident in APPsl animals.

Spread of Human P301L Tau to Connected Tissues

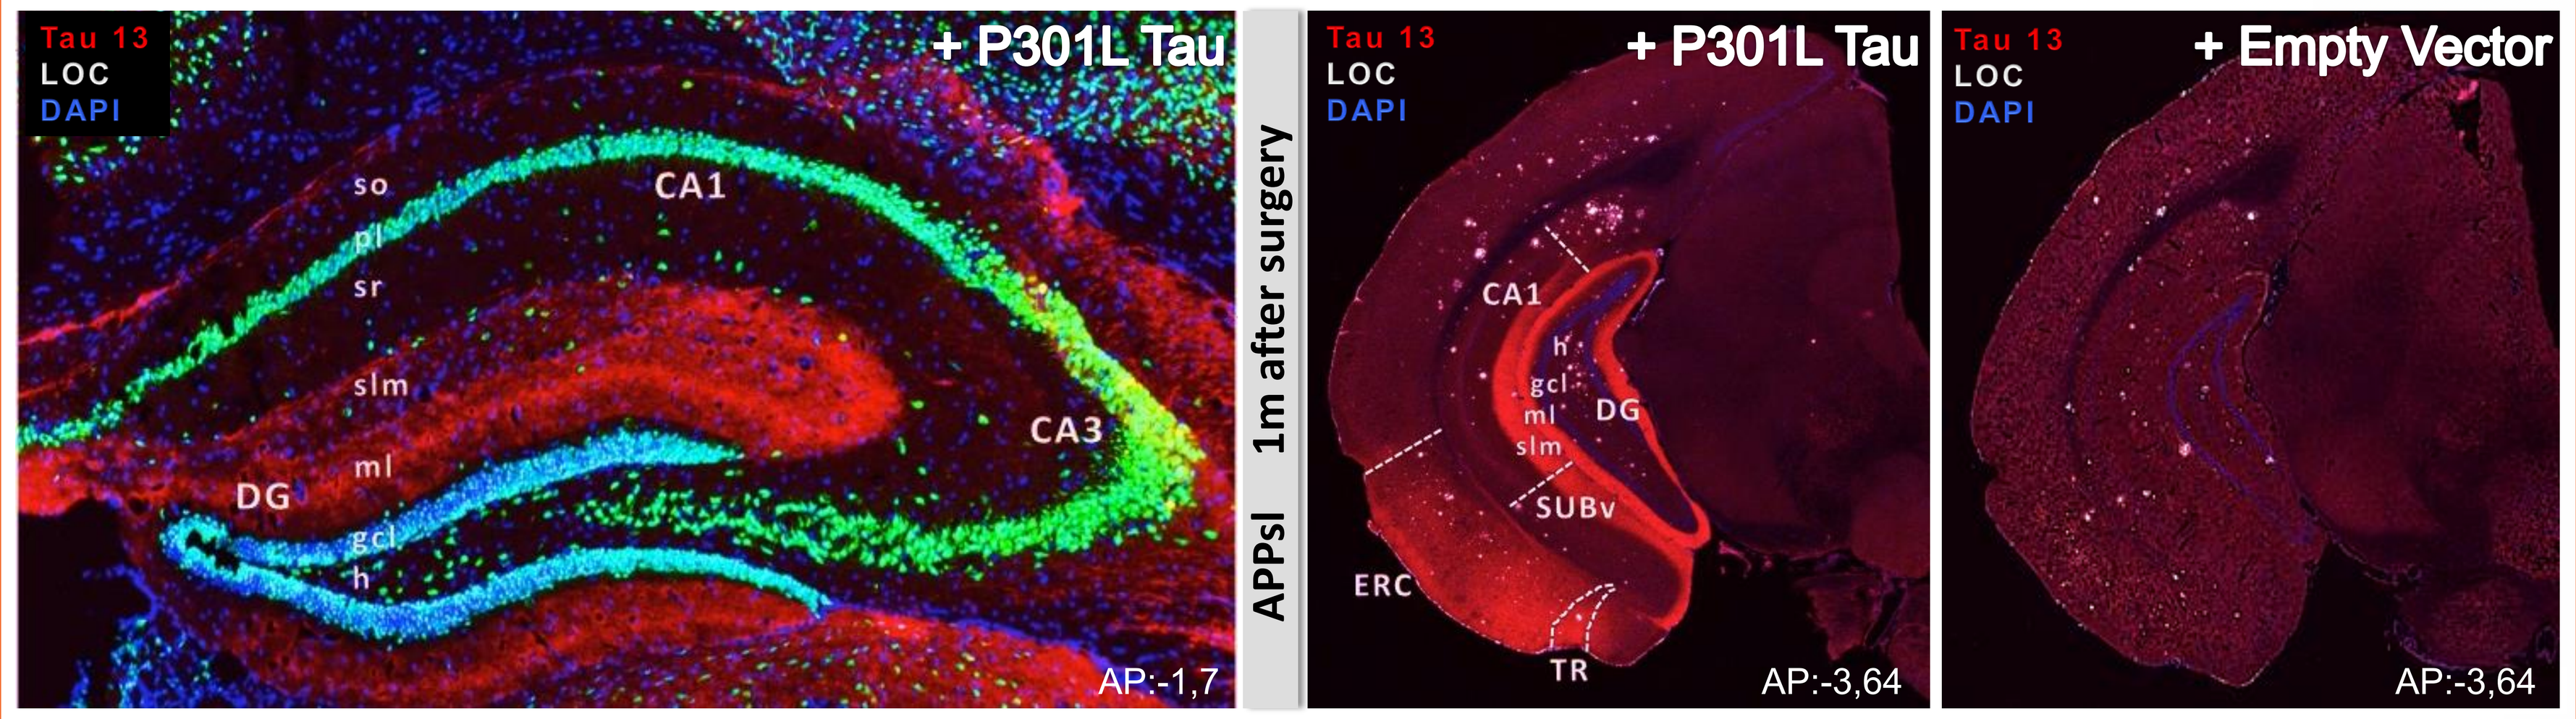
Tau 13
LOC
DAPI
+ P301L Tau
so
pl
sr
slm
ml
gcl
h
CA1
CA3
DG
AP:-1,7
APPsl 1m after surgery
+ P301L Tau
CA1
h
gcl
ml
DG
slm
SUBv
ERC
TR
AP:-3,64
+ Empty Vector
AP:-3,64

Figure 2. Human P301L Tau spreads from ERC to connected tissues.
Tau P301L focally injected in the ERC was found to spread to connected tissues already one month after injection. Immunofluorescence of human Tau in stratum lacunosum moleculare (slm) of CA1 and in the middle and outer molecular layer (ml) of the dentate gyrus (DG) suggest that Tau is transported via the lateral and medial perforant path, respectively. Time points analyzed: one and six months after injection.

Tau Pathology: Pre-NFTs

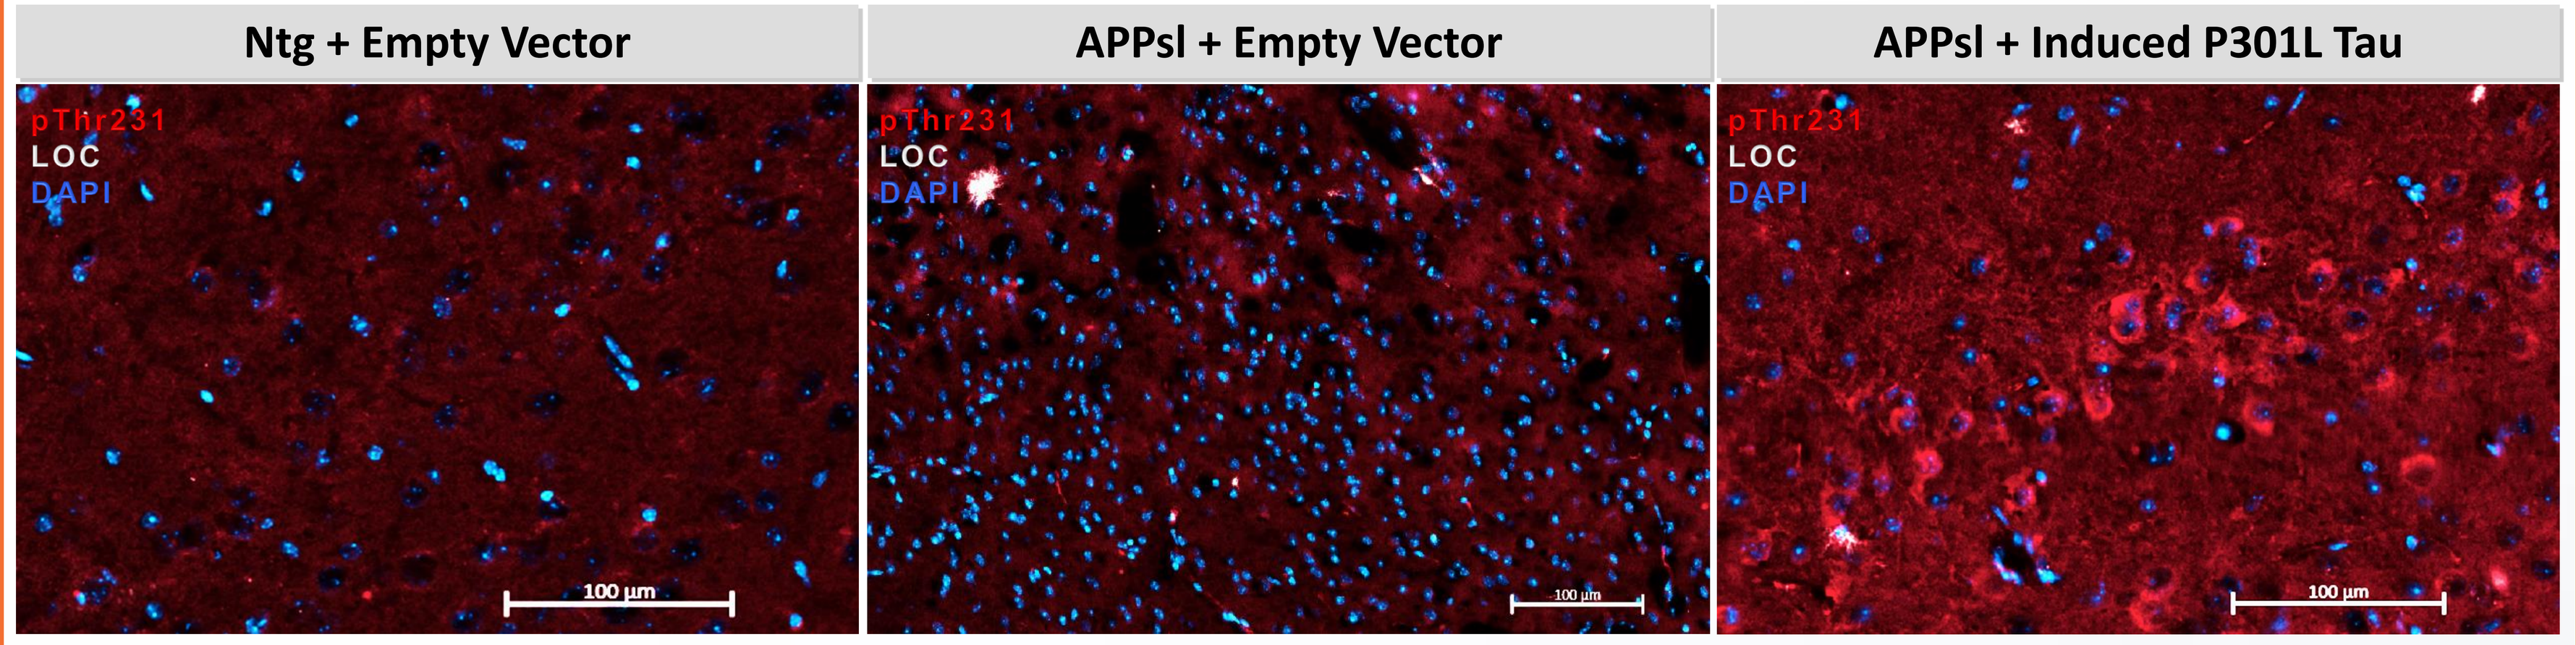
Ntg + Empty Vector
APPsl + Empty Vector
APPsl + Induced P301L Tau
pThr231
LOC
DAPI
100 µm

Figure 2. Pre-NFTs localized in ERC
Tau pathology in this mouse model was investigated with the AT180 antibody. Pathological phosphorylation of this residue was found in neuronal projections and soma, mimicking AD Tau mislocalization from the axon to the cell body. Time points investigated: one and six months after injection.

Hints of Cognitive Impairment Worsen in APPsl mice Inoculated with Human P301L Tau

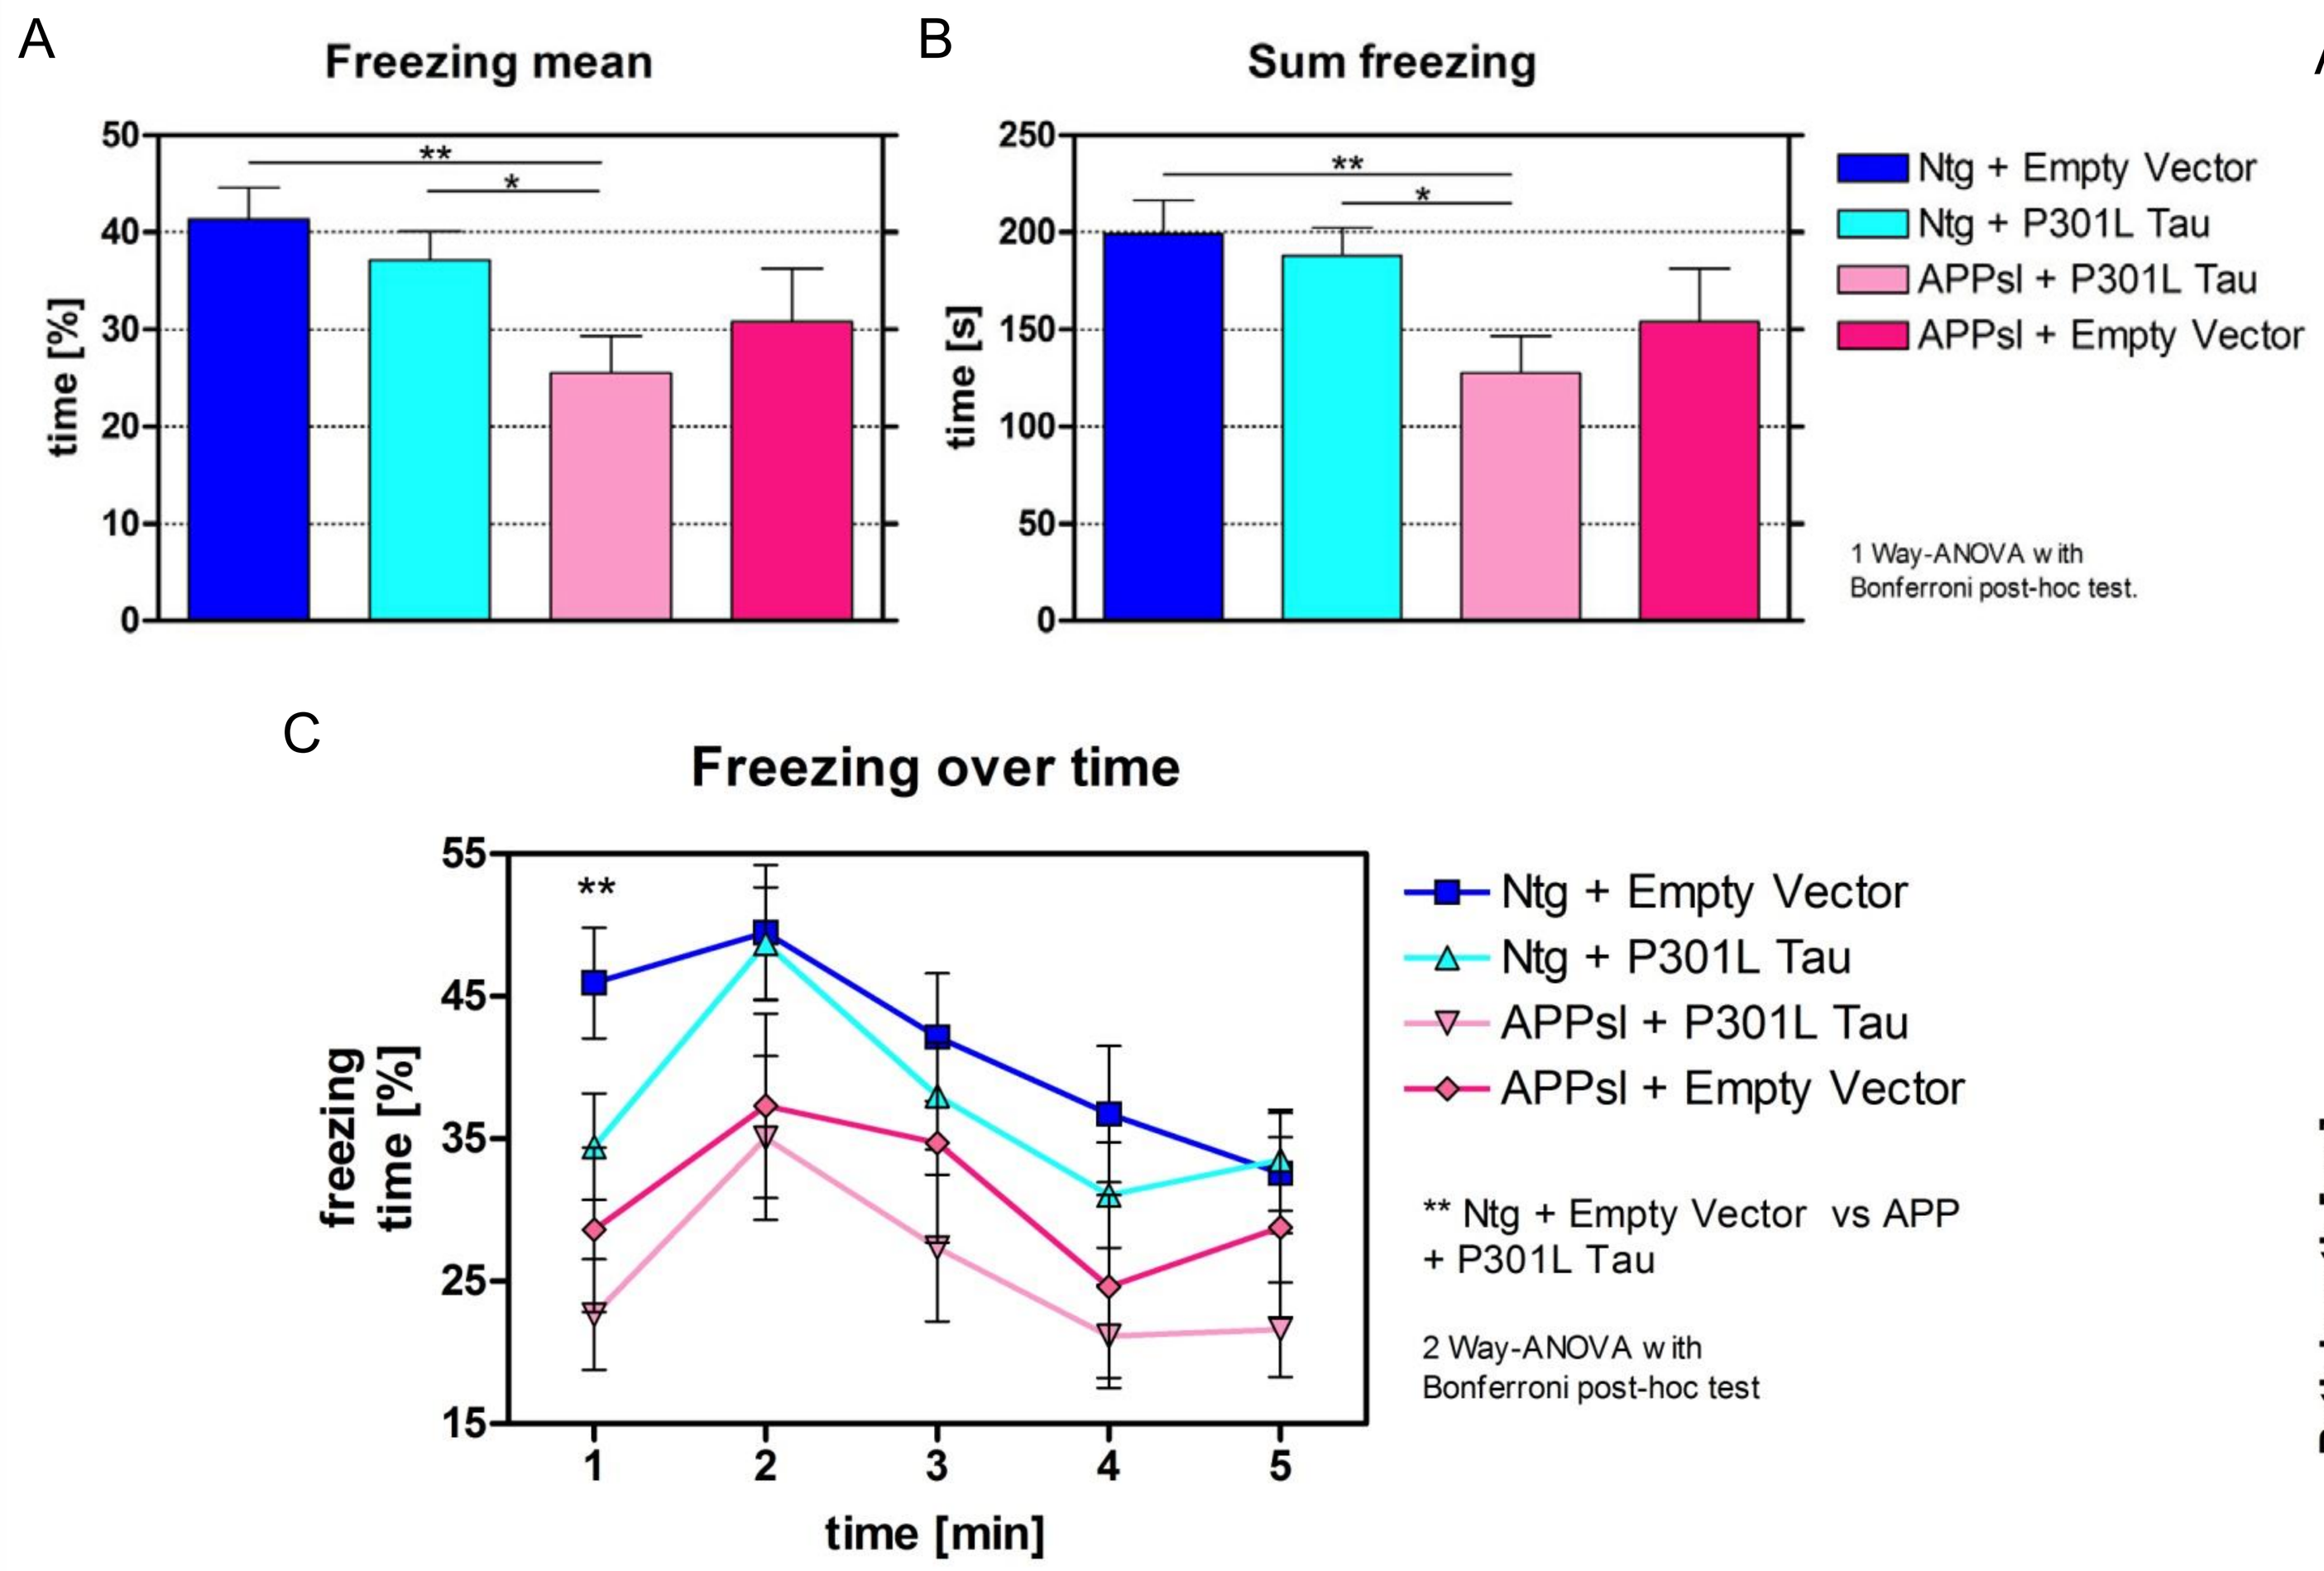
A
Freezing mean
time [%]
B
Sum freezing
time [s]
Ntg + Empty Vector
Ntg + P301L Tau
APPsl + P301L Tau
APPsl + Empty Vector
1 Way-ANOVA with Bonferroni post-hoc test.
C
Freezing over time
freezing time [%]
time [min]
** Ntg + Empty Vector vs APP + P301L Tau
2 Way-ANOVA with Bonferroni post-hoc test

Figure 4. Human P301L Tau is necessary in the APPsl mice to have a phenotype in the Contextual Fear Conditioning Test (CFC)
CFC outcomes reveal a statistically significant freezing phenotype at six months after injection only appearing when comparing APPsl + P301L Tau and Ntg + Empty Vector littermates. No differences between APPsl + Empty Vector and the Ntg littermates were detected. A) Sum of the 5 minutes in % of freezing. B) Mean of the 5 min in % of freezing. C) Over time representation of the % of freezing per group along the 5 min test. Mean + SEM (n=25+/-5). Statistical significance is indicated by *<0.05, **<0.01 as determined by One-Way (A,B) or Two-Way (C) ANOVA followed by Bonferroni post-hoc tests.

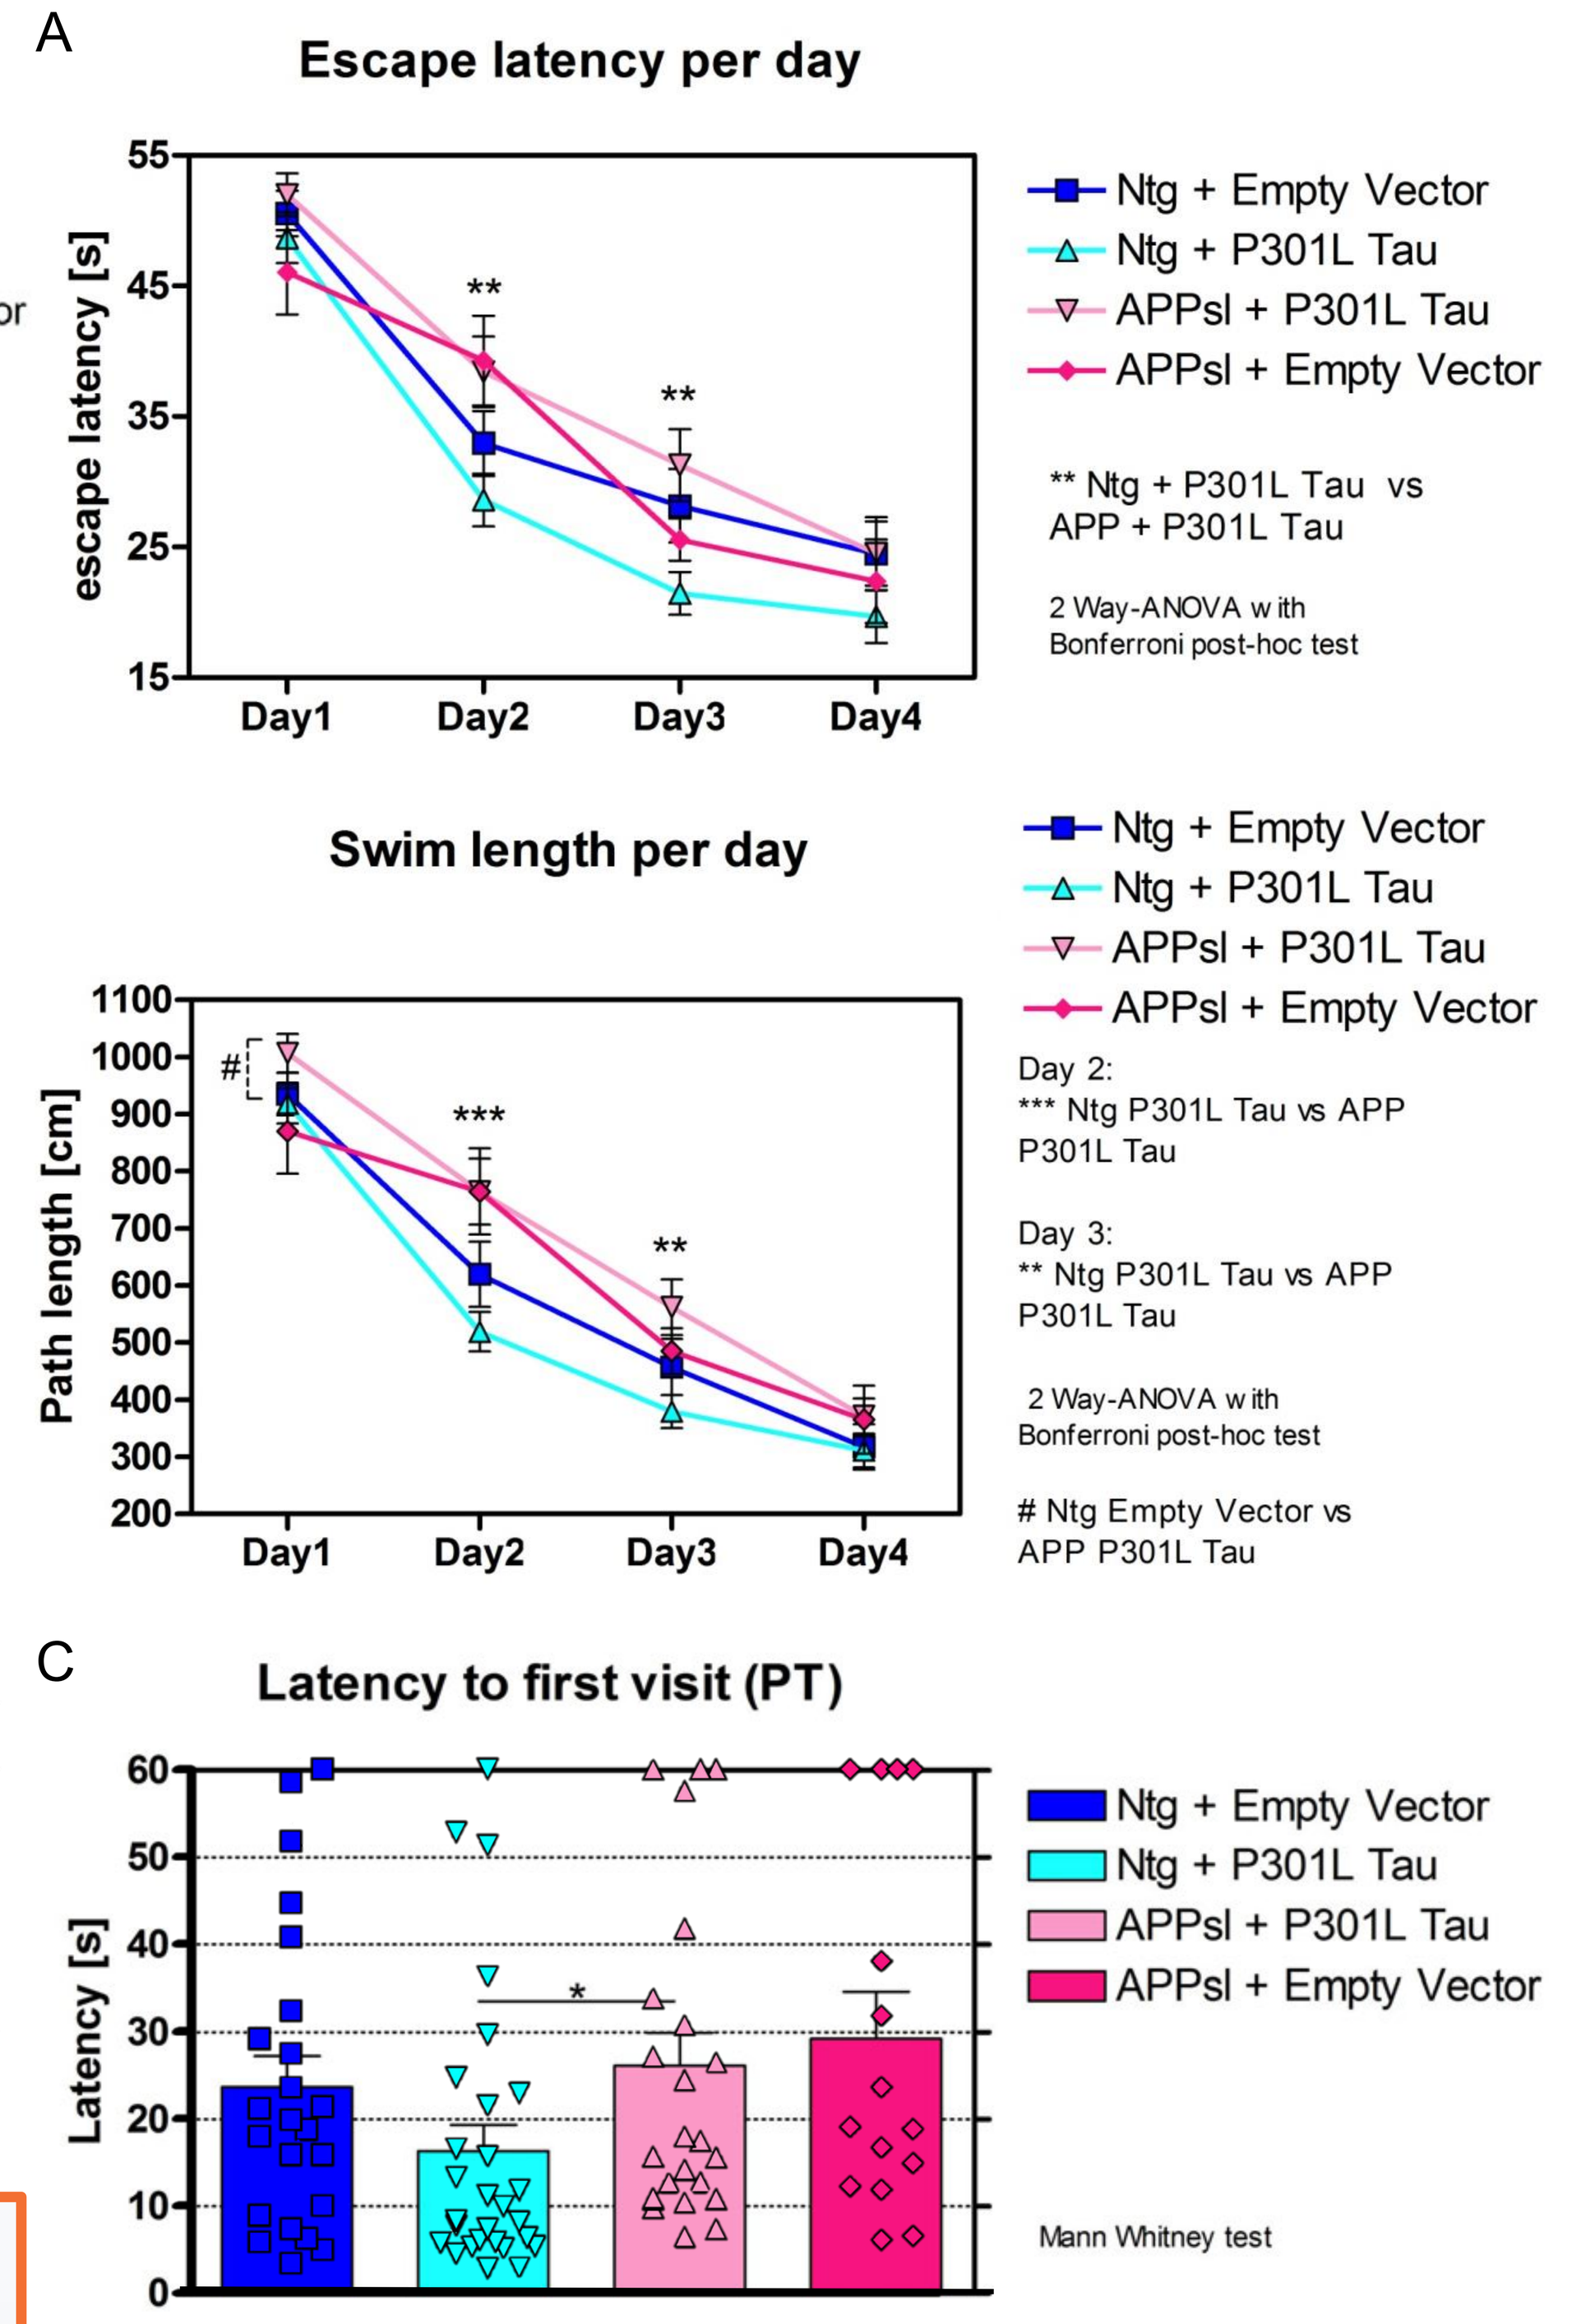
A
Escape latency per day
escape latency [s]
Day1
Day2
Day3
Day4
Ntg + Empty Vector
Ntg + P301L Tau
APPsl + P301L Tau
APPsl + Empty Vector
** Ntg + P301L Tau vs APP + P301L Tau
2 Way-ANOVA with Bonferroni post-hoc test
Swim length per day
Path length [cm]
Day 2:
*** Ntg P301L Tau vs APP P301L Tau
Day 3:
** Ntg P301L Tau vs APP P301L Tau
2 Way-ANOVA with Bonferroni post-hoc test
Ntg Empty Vector vs APP P301L Tau
C
Latency to first visit (PT)
Latency [s]
Mann Whitney test

Figure 5. Trend of cognitive impairment worsens in Morris Water Maze task (MWM).
Escape latency and swim length per day display statistical significant differences between Ntg + P301L Tau and APPsl + P301L Tau groups. The same results are found in latency to first visit to target zone in the probe trial. Significance between APPsl + P301L Tau and Ntg + Empty vector is also arising when looking at path length independently of time. No statistical significances were found between the other groups.(A) Cms needed per day to reach the platform. (B) seconds needed per day to reach the platform. (C) Probe trial, Seconds needed to first cross the target zone. Mean + SEM (n=25+/-5). Statistical significance is indicated by *<0.05, **<0.01, p***<0.001 as determined by Two-Way ANOVA followed by Bonferroni post-hoc Tests (A,B) and Mann Whitney Test (C).

Increased Explorative Behaviour

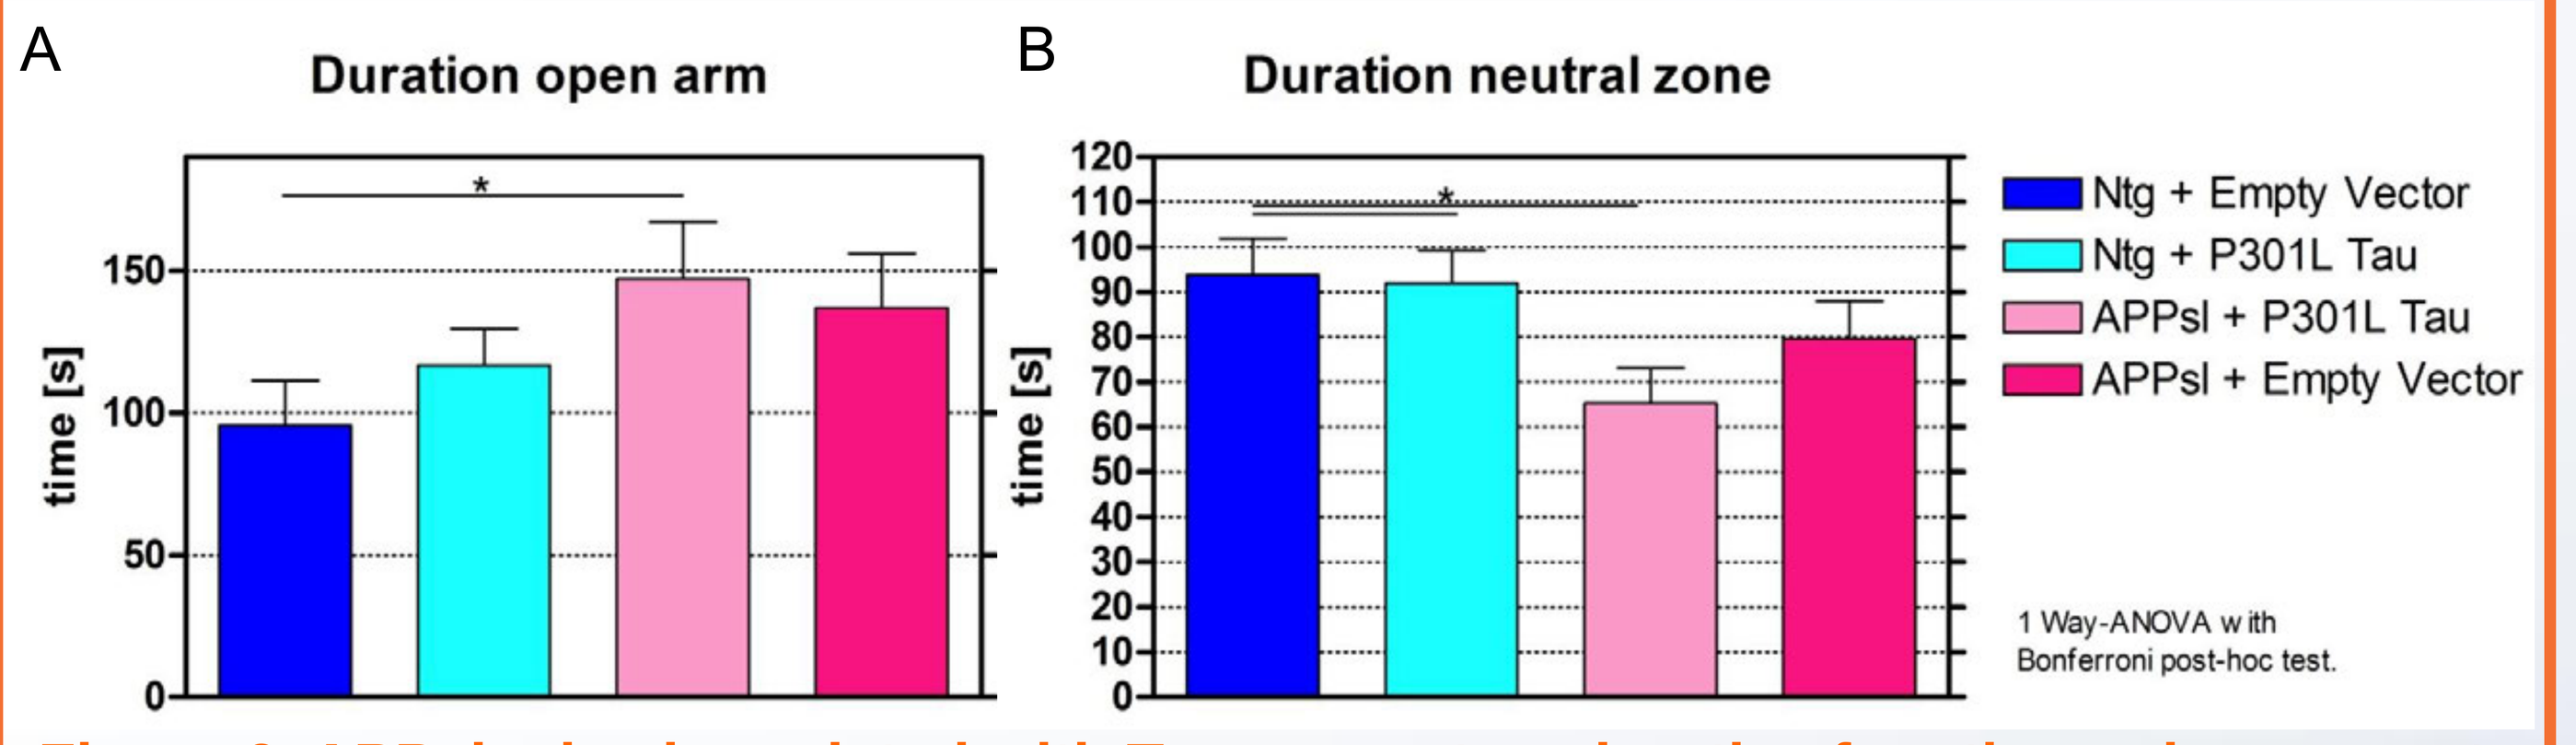
A
Duration open arm
time [s]
B
Duration neutral zone
time [s]
Ntg + Empty Vector
Ntg + P301L Tau
APPsl + P301L Tau
APPsl + Empty Vector
1 Way-ANOVA with Bonferroni post-hoc test.

Figure 6. APPsl mice inoculated with Tau present explorative function enhancement
Elevated Plus Maze results exhibit statistical significant divergences between APPsl + P301L Tau and Ntg littermates regarding the amount of time spent in the neutral and open arm zones, no statistical differences are found in the APPsl + Empty Vector group.
A) Seconds spent in the open arm. B) Seconds spent in the neutral zone. Mean + SEM (n=25+/-5). Statistical significance is indicated by *<0.05 as determined by One-Way ANOVA followed by Bonferroni post-hoc tests.

CONCLUSION

Histological analysis revealed stable expression of human P301L Tau in **entorhinal cortex** and connected brain areas from one month after injection. The mice show a combination of the two main features of AD, a high density of senile plaques, and pre-NFTs Tau. This new inducible mouse model is a promising new tool which mimics spatio-temporal patterns of AD-related pathology and may potentially help decreasing clinical trial failures.

Contact: Birgit Hutter-Paier, PhD | Director Neuropharmacology
QPS Austria GmbH | Parkring 12 | 8074 Grambach | Austria
birgit.hutter-paier@qps.com | www.qpsneuro.com

AAIC 2017, P4-03-048, Poster #17087